FPN: 472P

IMUGENE Developing Cancer Immunotherapies

MEDIZINISCHE UNIVERSITÄT WIEN

Prevention of metastasis formation by combination therapy targeting Her2 and PD-L1 in Her2-expressing tumors based on observed efficacious vaccination against Her2-positive tumors

Joshua Tobias*[1], Sandra Högler[2], Diego Shih-Chieh Lin[3], Yee Chao[3], Erika Garner-Spitzer[1], Nicholas J. Ede[4], Christoph C. Zielinski[5], Sharon Yavrom[4], Michael Kundi[1], Ursula Wiedermann*[1]

[1]Medical University of Vienna, Austria; [2]University of Veterinary Medicine, Vienna, Austria; [3]Taipei Veterans General Hospital, Taipei, Taiwan; [4]Imugene Limited, Sydney, Australia;
[5]Central European Cancer Center, Wiener Privatklinik, and Central European Cooperative Oncology Group (CECOG), Vienna, Austria

Introduction

- HER-2/neu, a member of the epidermal growth factor receptor (EGFR) family, is overexpressed in 6%-30% of breast and gastric cancers.
- Targeting Her-2/neu with trastuzumab, a clinically approved therapeutic monoclonal antibody, is associated with acquired resistance in a significant number of patients.
- HER-Vaxx is a B cell peptide-based anti-HER-2/neu vaccine (IMU-131) comprising trastuzumab's binding site[1,2].
- HER-Vaxx has been shown to be safe and to prolong progression-free survival in patients with HER-2/neu-overexpressing gastric/gastro-esophageal junction cancer (GC) in the phase 1b and 2 HERIZON study (NCT02795988), [2,-4].
- Tumoral upregulation of PD-L1 and downregulation of Her-2/neu expression have both been linked to resistance and subsequent metastasis development following treatment with trastuzumab.

Aim

- We aimed to investigate the upregulation of PD-L1 and downregulation of Her-2/neu expression following Her-2/neu-targeted vaccination in preclinical and clinical settings.

Methods

- Immunohistochemistry (IHC), fluorescence in situ hybridization (FISH), and combined positive score (CPS) assessment were applied to evaluate PD-L1 and Her-2/neu expression levels in:
 1. **Preclinical setting:** using metastasized lungs of mice prophylactically vaccinated with HerVaxx and tail-vein injected with mammary carcinoma cells expressing human Her-2/neu.
 2. **Clinical setting:** Using biopsies from an adult patient from the phase 1b HERIZON trial (NCT02795988), before and after the treatment, who had developed a new metastatic lesion and had lost the expression of Her-2/neu in the primary tumor

Results

Preclinical setting
Her-2/neu targeting vaccination significantly increased the ratio of positive PD-L1 to Her-2/neu metastases

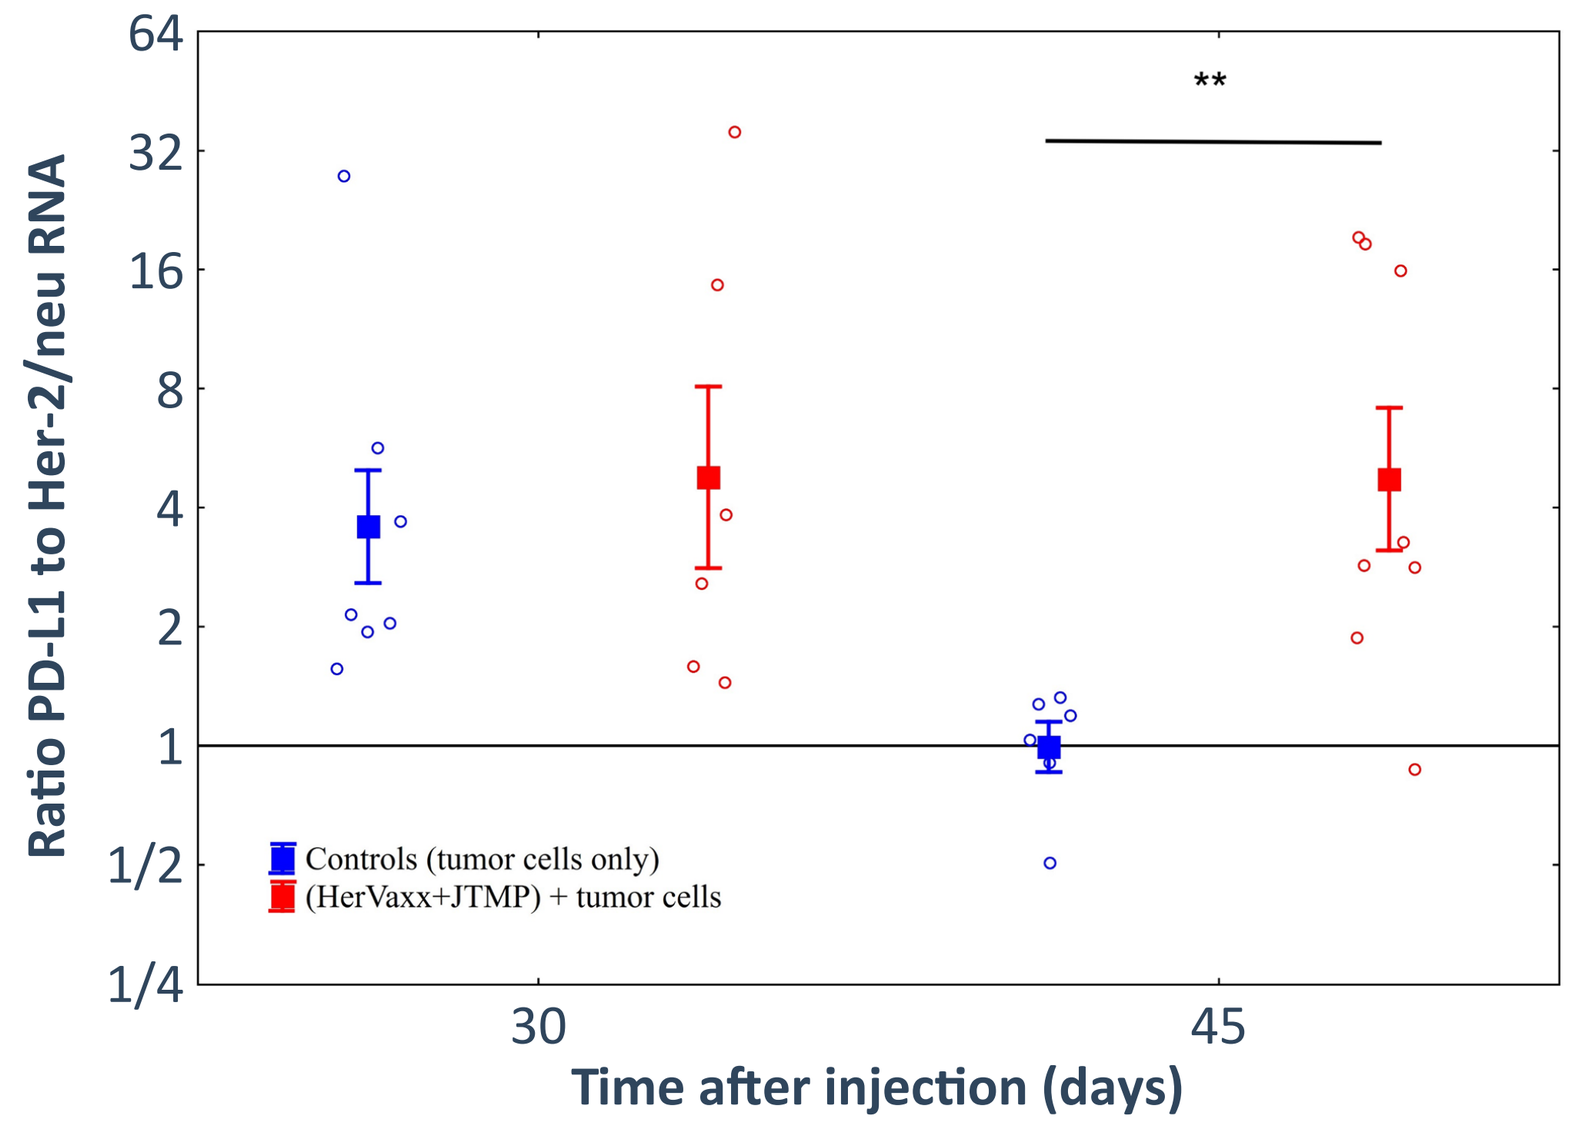

Ratio PD-L1 to Her-2/neu RNA
64
32
16
8
4
2
1
1/2
1/4
**
Controls (tumor cells only)
(HerVaxx+JTMP) + tumor cells
30
45
Time after injection (days)

Preclinical setting
Her-2/neu targeting vaccination increased PD-L1 and decreased Her-2/neu expression in the same tumors

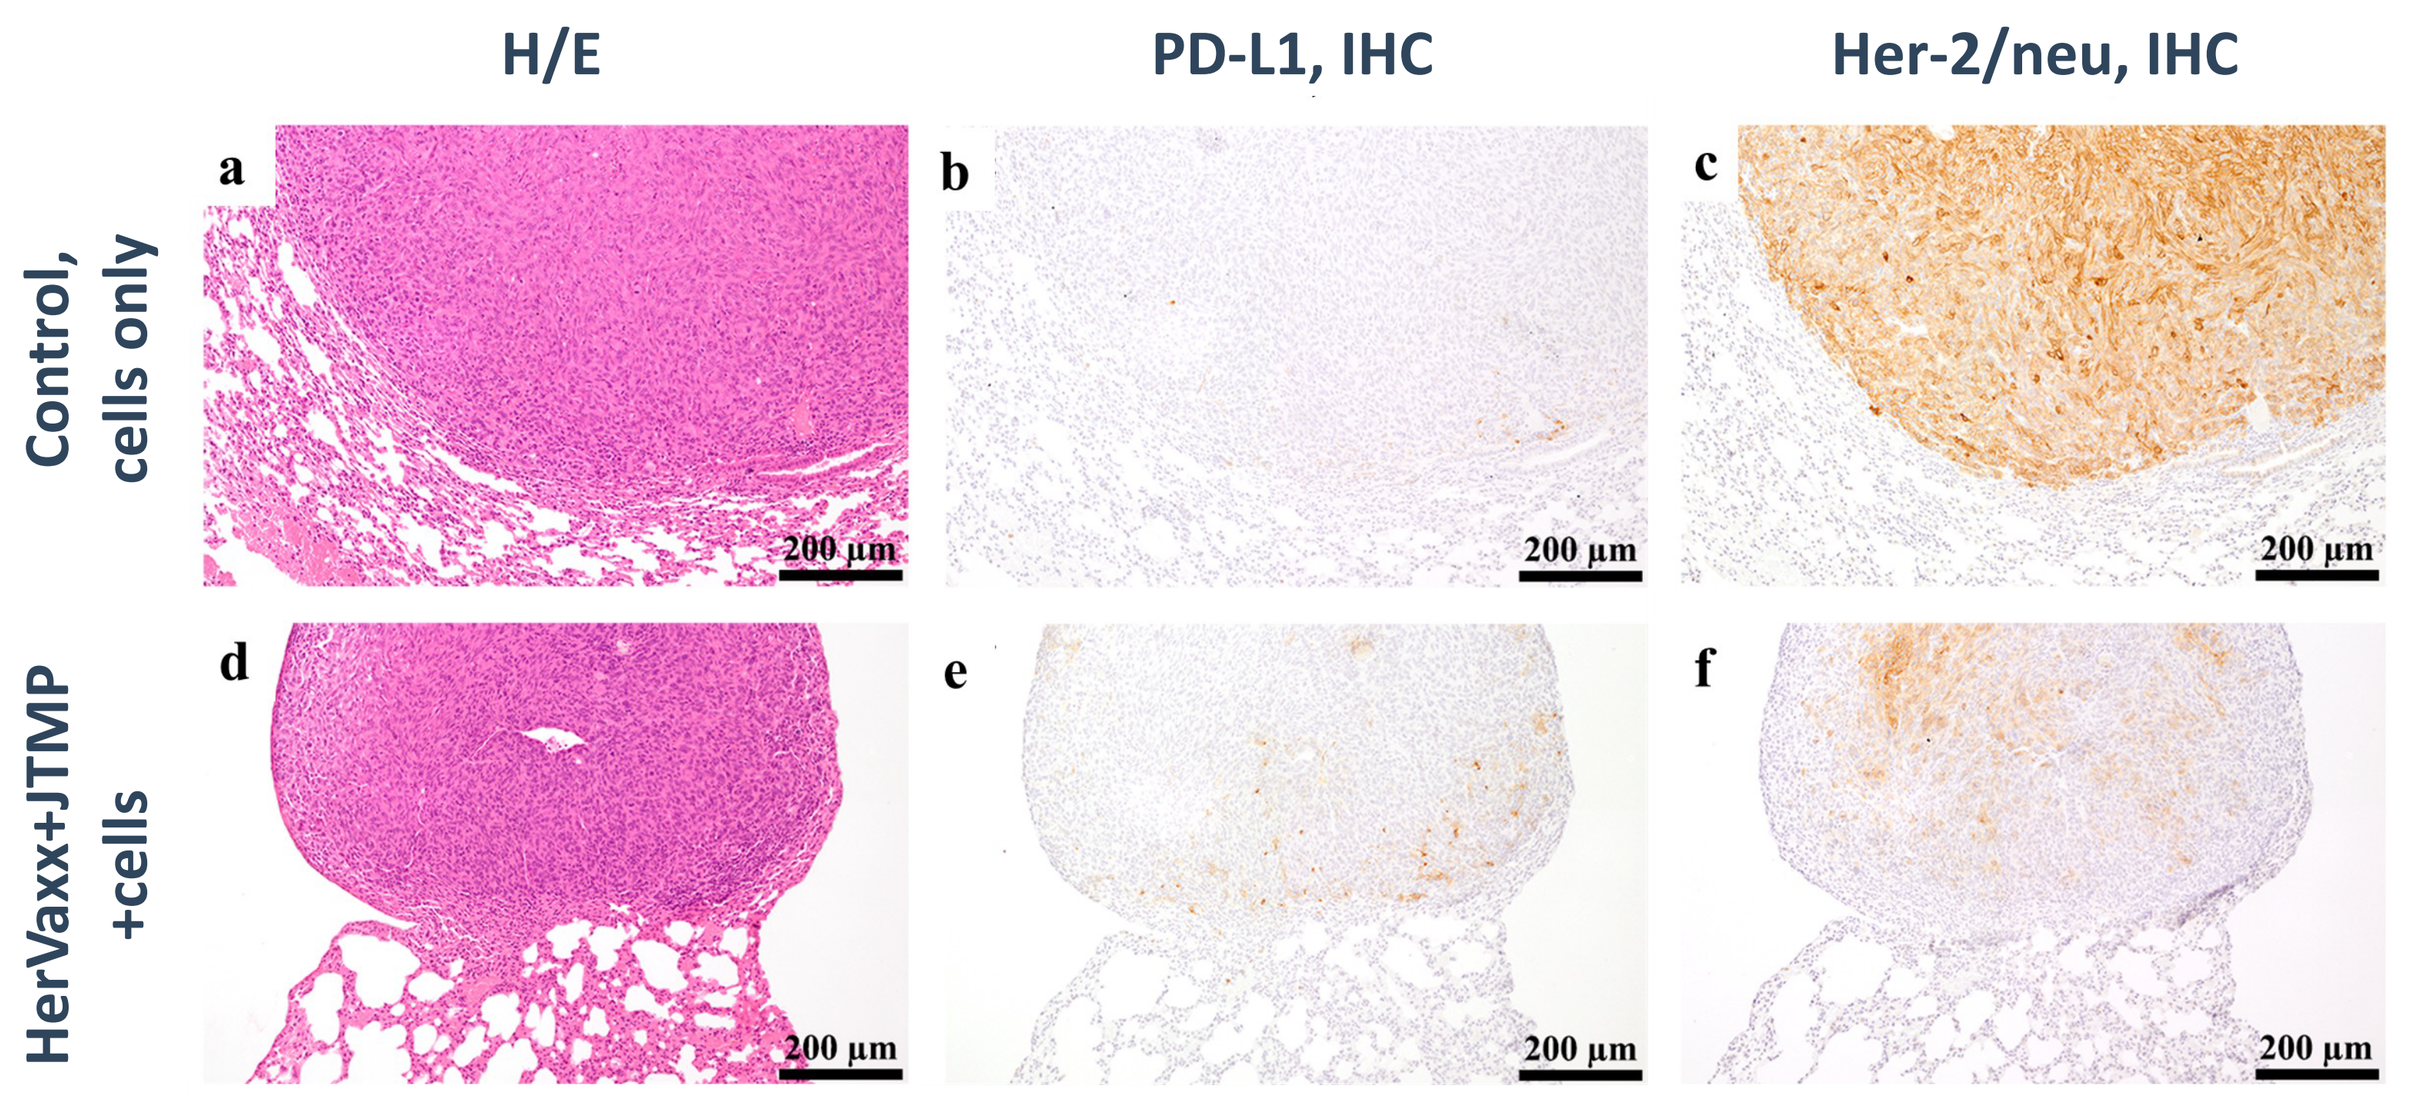

H/E
PD-L1, IHC
Her-2/neu, IHC
Control, cells only
HerVaxx+JTMP +cells
a
b
c
d
e
f
200 µm

Clinical setting
Her-2/neu targeting vaccination increased PD-L1 and decreased Her-2/neu expression in the primary tumor

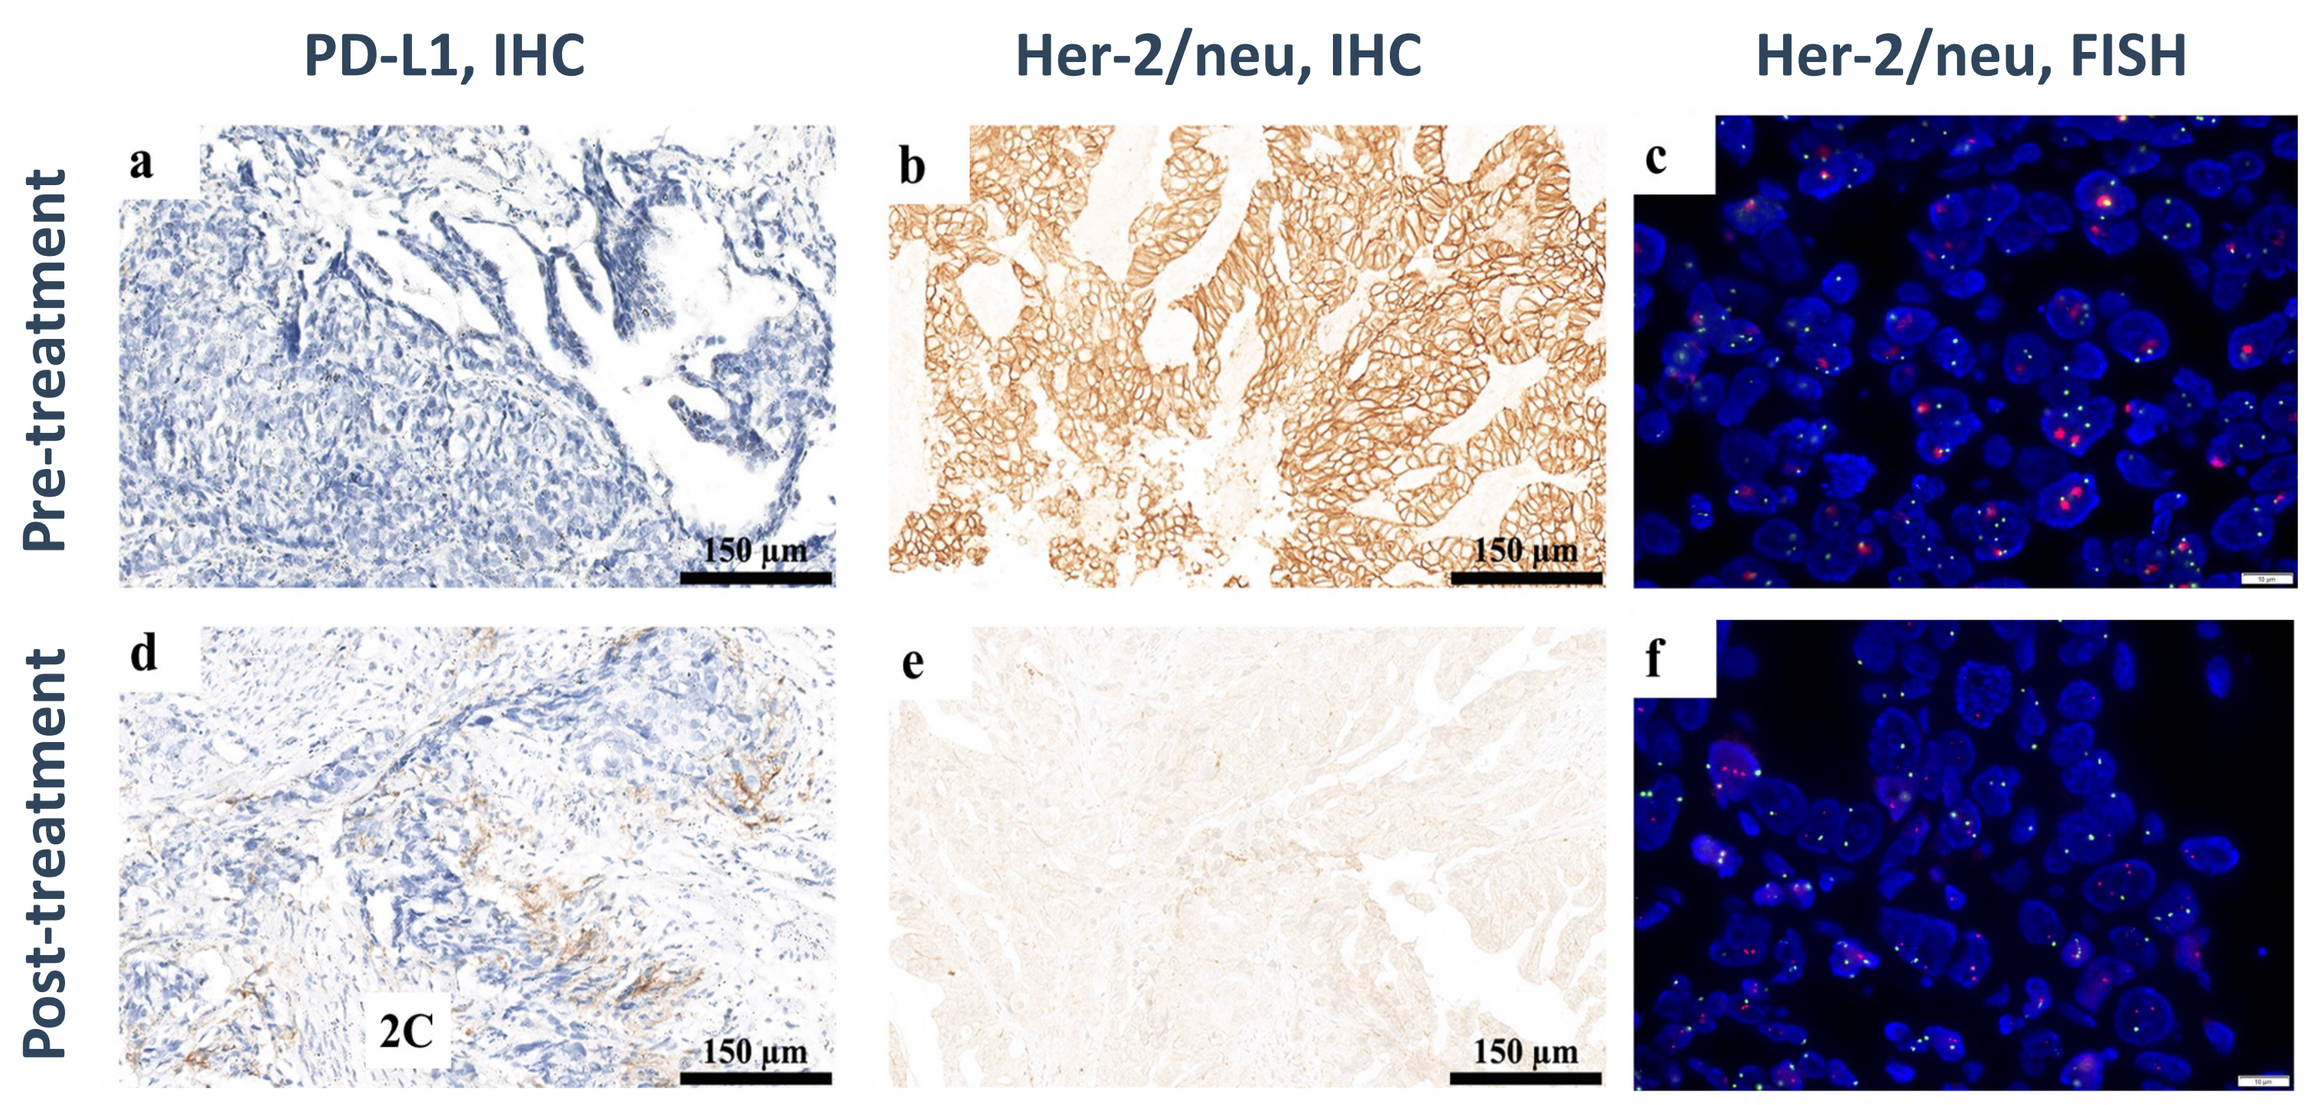

PD-L1, IHC
Her-2/neu, IHC
Her-2/neu, FISH
Pre-treatment
Post-treatment
a
b
c
d
e
f
2C
150 µm

Conclusions

- <u>Preclinical setting:</u> **Targeting Her-2/neu was associated with concomitant PD-L1 upregulation and the receptor's downregulation of expression in the same tumors, resulting in a significantly higher ratio of PD-L1 to Her-2/neu positive metastases.**
- <u>Clinical setting:</u> **Targeting Her-2/neu was associated with the upregulation of PD-L1 and downregulation of Her-2/neu expression in the evaluated patient's primary tumor, conceivably influencing the disease progression observed in the patient.**

These observations suggest that targeting Her-2/neu induces upregulation of PD-L1, and are clinically relevant for the elimination of tumor cells with a PD-L1 positive and Her-2/neu negative status and prevention of new metastasis development and immune evasion

References

1. Tobias, J., et al. Enhanced and long-term immunogenicity of a Her-2/neu multi-epitope vaccine conjugated to the carrier CRM197 in conjunction with the adjuvant Montanide. *BMC Cancer* 2017;17(1), 118.
2. Wiedermann, U., et al. Clinical and Immunologic Responses to a B-Cell Epitope Vaccine in Patients with HER2/neu-Overexpressing Advanced Gastric Cancer-Results from Phase Ib Trial IMU.ACS.001. *Clin Cancer Res*. 2021;27(13), 3649-3660.
3. Maglakelidze, M., et.al. HERIZON: A Phase 2 study of IMU-131, a HER2 targeting peptide vaccine, plus standard of care chemotherapy in patients with HER2 overexpressing metastatic or advanced gastric/GEJ adenocarcinoma *ESMO Asia* Poster. 2022, Singapore.
4. Tobias, J., et al. PD-8 HERIZON: A phase 2 study of HER-Vaxx (IMU-131), a HER2-targeting peptide vaccine plus SOC chemotherapy in patients with HER2+ advanced stomach cancer – correlation of the antibody responses and clinical outcome. *Annals of Oncology*, 2023. 34: p. S4.

Conflicts of Interest

The presenter, Joshua Tobias, declares no potential conflicts of interest.

Contacts

Prof. Dr. Ursula Wiedermann: ursula.wiedermann@meduniwien.ac.at,
Dr. Joshua Tobias: joshua.tobias@meduniwien.ac.at