Abstract 1453: Development of a novel PD-1 vaccine and in combination with two Chimeric HER-2 peptide vaccine provides synergistic inhibition of tumor growth in a syngeneic Balb/c model challenged with CT26/HER-2 carcinoma cell line

Tanios Bekaii-Saab[1], Jay Overholser[2], Yuhong Yang[2], Manuel Penichet[3], Linlin Guo[2], Pravin T.P Kaumaya[2,3]. [1]Mayo Clinic Cancer Center, Phoenix, AZ; [2] The Ohio State University Wexner Medical Center and the [3]James Comprehensive Cancer Center, Columbus, OH; [3]UCLA, Los Angeles, CA

Background

Therapeutic blockade of the signaling axis between PD-1 and PD-L1 with monoclonal antibodies has shown remarkable clinical success in the treatment of cancer and demonstrated impressive activity across a broad set of cancer subtypes. Contrary to treatment with monoclonal antibodies, chimeric B-cell cancer vaccines have the advantage of producing a specific immune response that can potentially induce memory B & T cell responses, while reducing immune evasion and suppression. **KEY-Vaxx** is a B-cell peptide cancer vaccine which aims to induce the body to produce polyclonal antibodies that block PD-1 signalling, and thus produce an anticancer effect similar to Keytruda and Opdivo. KEY-Vaxx has shown great potential in preclinical studies. It outperformed an industry-standard mouse anti-PD-1 antibody in a mouse model of HER2+ colorectal cancer. When used in combination with our HER-2 combination vaccine B-Vaxx (see poster XXX) we obtained synergistic inhibition of tumor growth in a colon carcinoma Balb/c model challenged with CT26/HER-2 cell line. The combined vaccines were safe with no evidence of toxicity or autoimmunity.

PD-1 Peptide epitope selection, design, synthesis & characterization

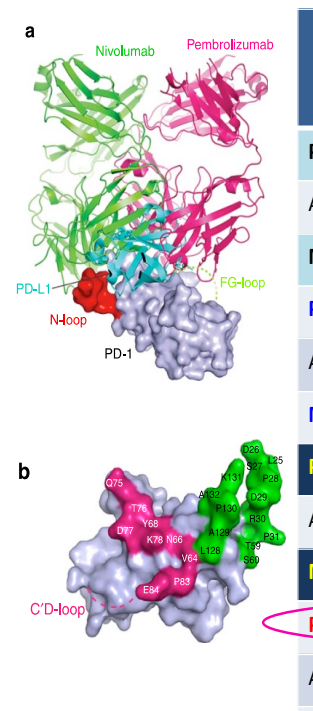
a
Nivolumab
Pembrolizumab
PD-L1
FG-loop
N-loop
PD-1
b
C'D-loop

PEPTIDES	AMINO ACID SEQUENCES OF SYNTHESIZED PD-1 PEPTIDES
PD-1 (32-50)	H_2N-^{32}V-L-N-W-Y-R-M-S-P-S-N-Q-T-D-K-L-A-A-F^{50}-$CONH_2$
AC-PD-1 (32-50)	CH_3CONH-^{32}V-L-N-W-Y-R-M-S-P-S-N-Q-T-D-K-L-A-A-F^{50}-$CONH_2$
MVF-PD-1 (32-50)	*KLLSLIKGVIVHRLEGVE-GPSL-*V-L-N-W-Y-R-M-S-P-S-N-Q-T-D-K-L-A-A-F-CONH2
PD-1 (45-64)	H_2N-^{45}K-L-A-A-F-P-E-D-R-S-Q-P-G-Q-D-C-R-F-R^{64} $CONH_2$
Ac-PD-1 (45-64)	CH_3CONH^{45}K-L-A-A-F-P-E-D-R-S-Q-P-G-Q-D-C-R-F-R^{64} $CONH_2$
MVF-PD-1 (45-64)	*KLLSLIKGVIVHRLEGVE-GPSL*^{45}K-L-A-A-F-P-E-D-R-S-Q-P-G-Q-D-C-R-F-R^{64} $CONH_2$
PD-1 (73-90)	H_2N-^{73}D-F-H-M-S-V-V-R-A-R-R-N-D-S-G-T-Y-L^{90} -$CONH_2$
AC-PD-1 (73-90)	CH_3CONH-^{73}D-F-H-M-S-V-V-R-A-R-R-N-D-S-G-T-Y-L^{90} -$CONH_2$
MVF-PD-1 (73-90)	*KLLSLIKGVIVHRLEGVE-GPSL-*^{73}D-F-H-M-S-V-V-R-A-R-R-N-D-S-G-T-Y-L^{90} -CONH2
PD-1 (92-110)	H_2N-^{92}G-A-I-S-L-A-P-K-A-Q-I-K-E-S-L-R-A-E-L^{110} -$CONH_2$
AC-PD-1 (92-110)	CH_3CONH-^{92}G-A-I-S-L-A-P-K-A-Q-I-K-E-S-L-R-A-E-L^{110} -$CONH_2$
MVF-PD-1 (92-110)	*KLLSLIKGVIVHRLEGVE-GPSL-*^{92}G-A-I-S-L-A-P-K-A-Q-I-K-E-S-L-R-A-E-L^{110}-CONH2

KEY-Vaxx PD-1 epitope

KEY-Vaxx peptide vaccine

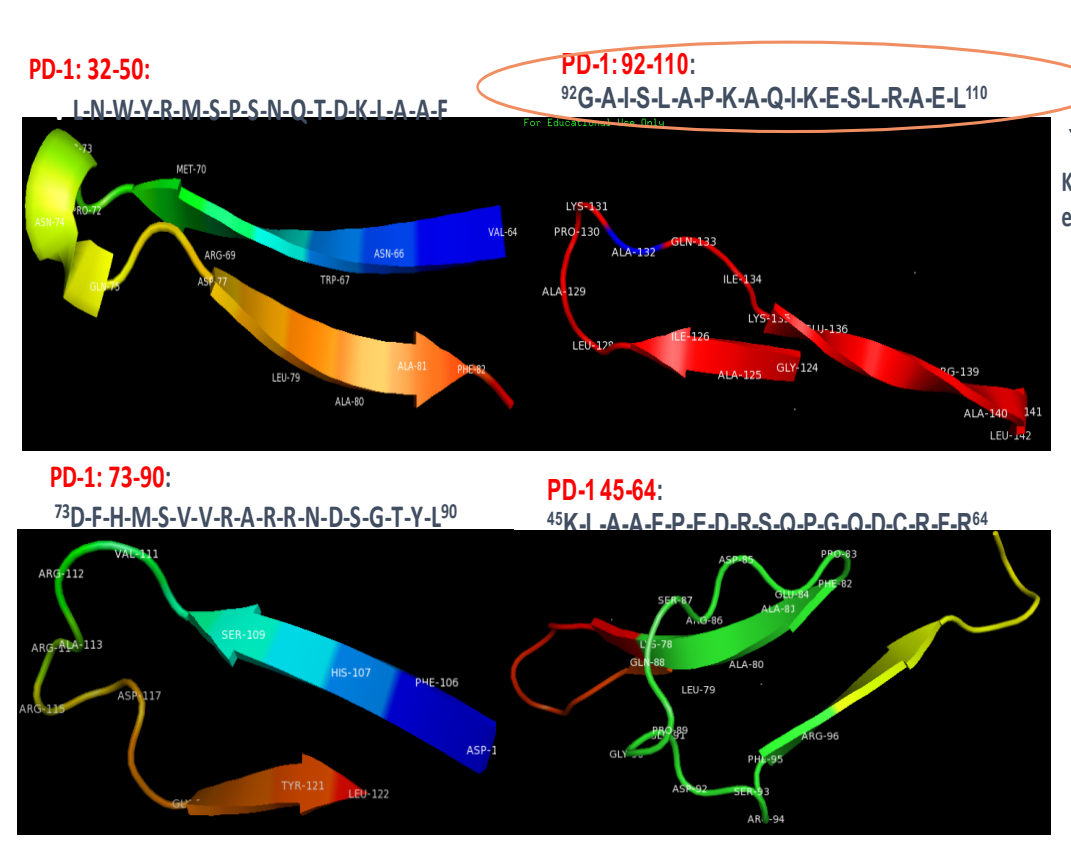
PD-1: 32-50:
L-N-W-Y-R-M-S-P-S-N-Q-T-D-K-L-A-A-F
PD-1: 92-110:
92G-A-I-S-L-A-P-K-A-Q-I-K-E-S-L-R-A-E-L110
KEY-Vaxx PD-1 epitope
PD-1: 73-90:
73D-F-H-M-S-V-V-R-A-R-R-N-D-S-G-T-Y-L90
PD-1 45-64:
45K-L-A-A-F-P-E-D-R-S-Q-P-G-Q-D-C-R-F-R64

Several predictive immunogenicity algorithms were used to prioritize four highest scoring B-cell epitope sequences of human PD-1. Amino acid 32-50, 45-64, 73-90 and 92-110 were chosen for evaluation based on the X-ray tructure of PD-1:PDL1. Peptide were synthesized /characterized.

SPR analyses of PD-1 peptides binding to Nivolumab & rhuPD-L1

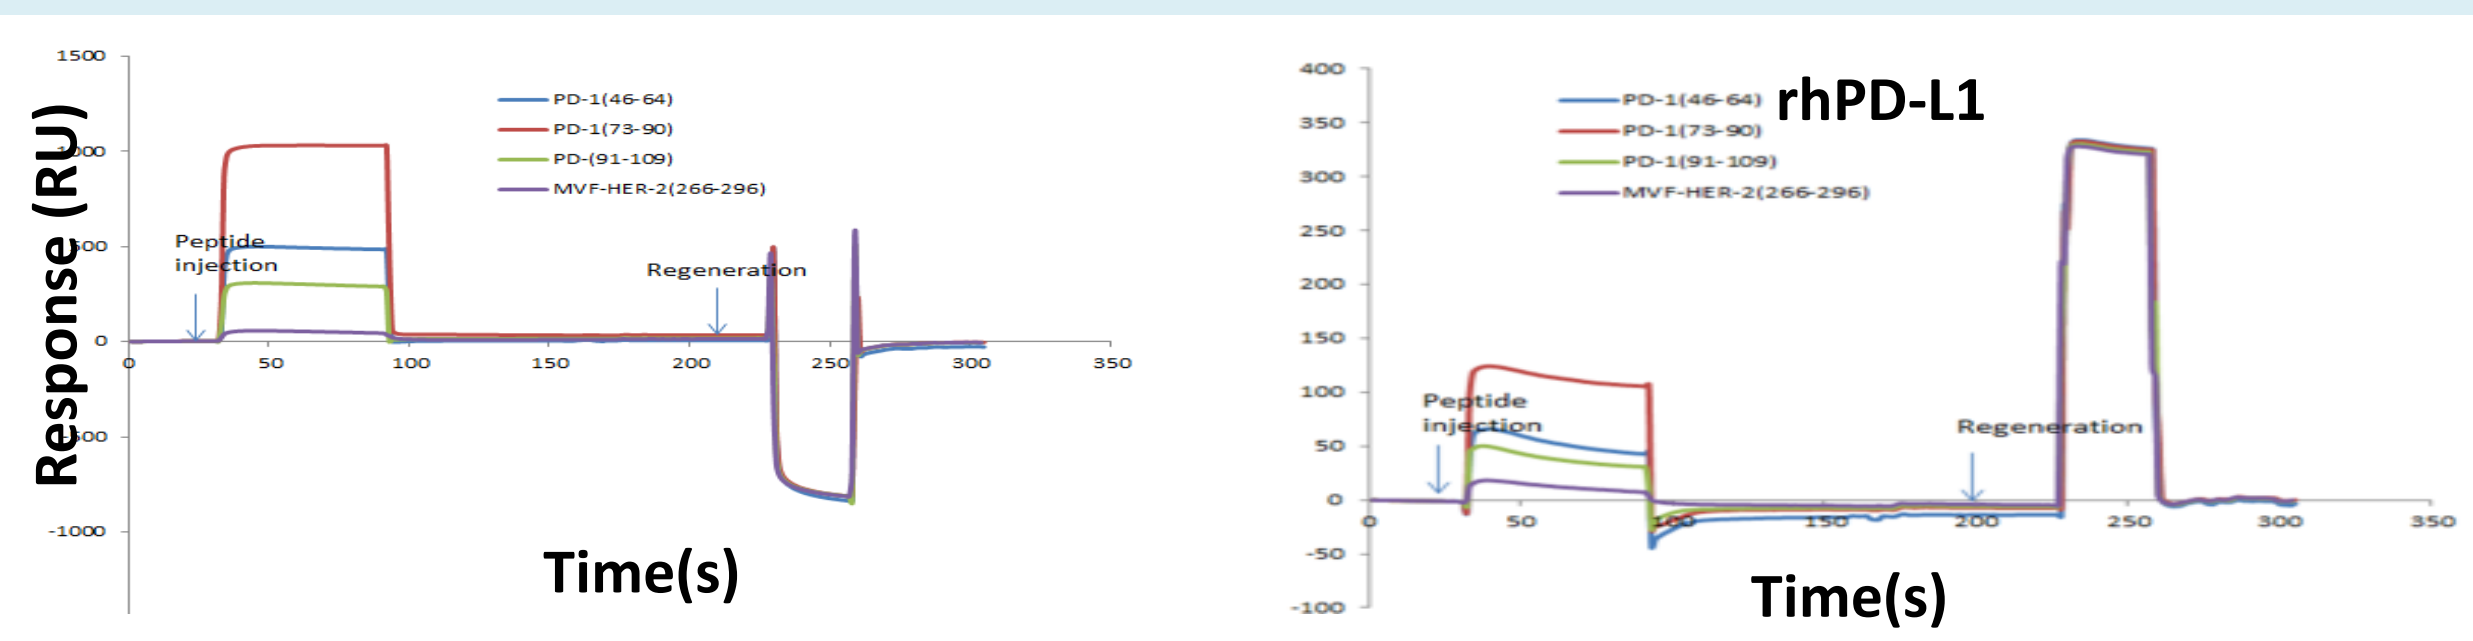
Response (RU)
PD-1(46-64)
PD-1(73-90)
PD-(91-109)
MVF-HER-2(266-296)
Peptide injection
Regeneration
Time(s)
rhPD-L1
PD-1(46-64)
PD-1(73-90)
PD-1(91-109)
MVF-HER-2(266-296)
Peptide injection
Regeneration
Time(s)

The specificity of the selected PD-1 peptides was determined by surface plasmon resonance (SPR) spectroscopy (Biacore T200, at 25°C). The binding affinities to immobilized human PD-L1 (hPD-L1) and Nivolumab on CM5 sensor chip. From these binding studies, **we can conclude that the PD-1 peptides 45-64, 73-90 and 92-110 can recognize the rPD-L1 and Nivolumab and can act as inhibitors of PD-1-PDL1.**

Recognition of mouse PD-1 by αhPD-1 rabbit antibodies

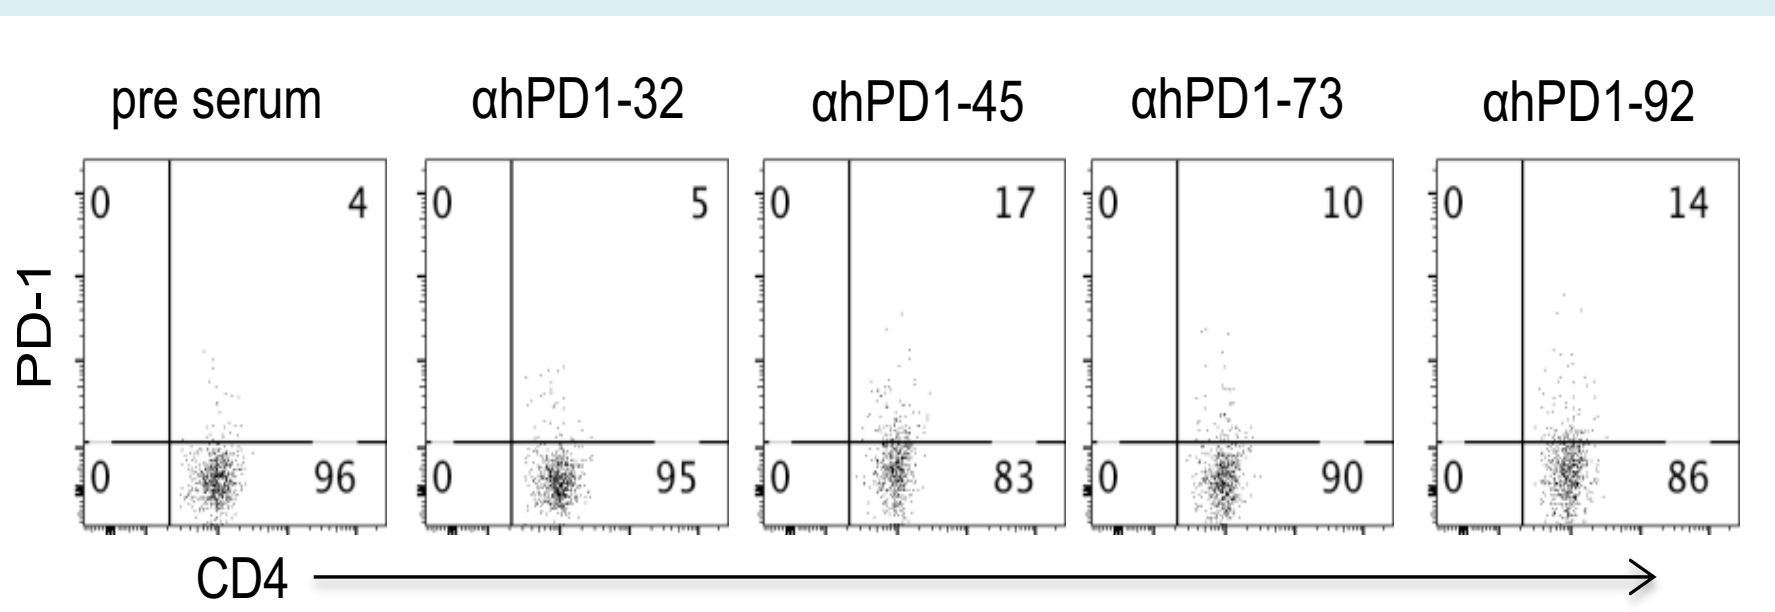
pre serum
αhPD1-32
αhPD1-45
αhPD1-73
αhPD1-92
PD-1
CD4
0 4 0 96
0 5 0 95
0 17 0 83
0 10 0 90
0 14 0 86

Splenocytes from immunized EAE mice were isolated and activated with MOG 35-55. The cells were then incubated with APC conjugated serum from pre-vaccinated mice or mice vaccinated with different antibodies.

Results show αhPD1-45, αhPD1-73 and αhPD1-92 ABS raised in rabbits recognize mouse PD-1 expressed on activated myelin-specific CD4 T cells **confirming the rationale for their use in syngeneic BALB/c studies.**

Peptide antibodies are inhibitory (PD-1 92) or activating (PD-1-45)

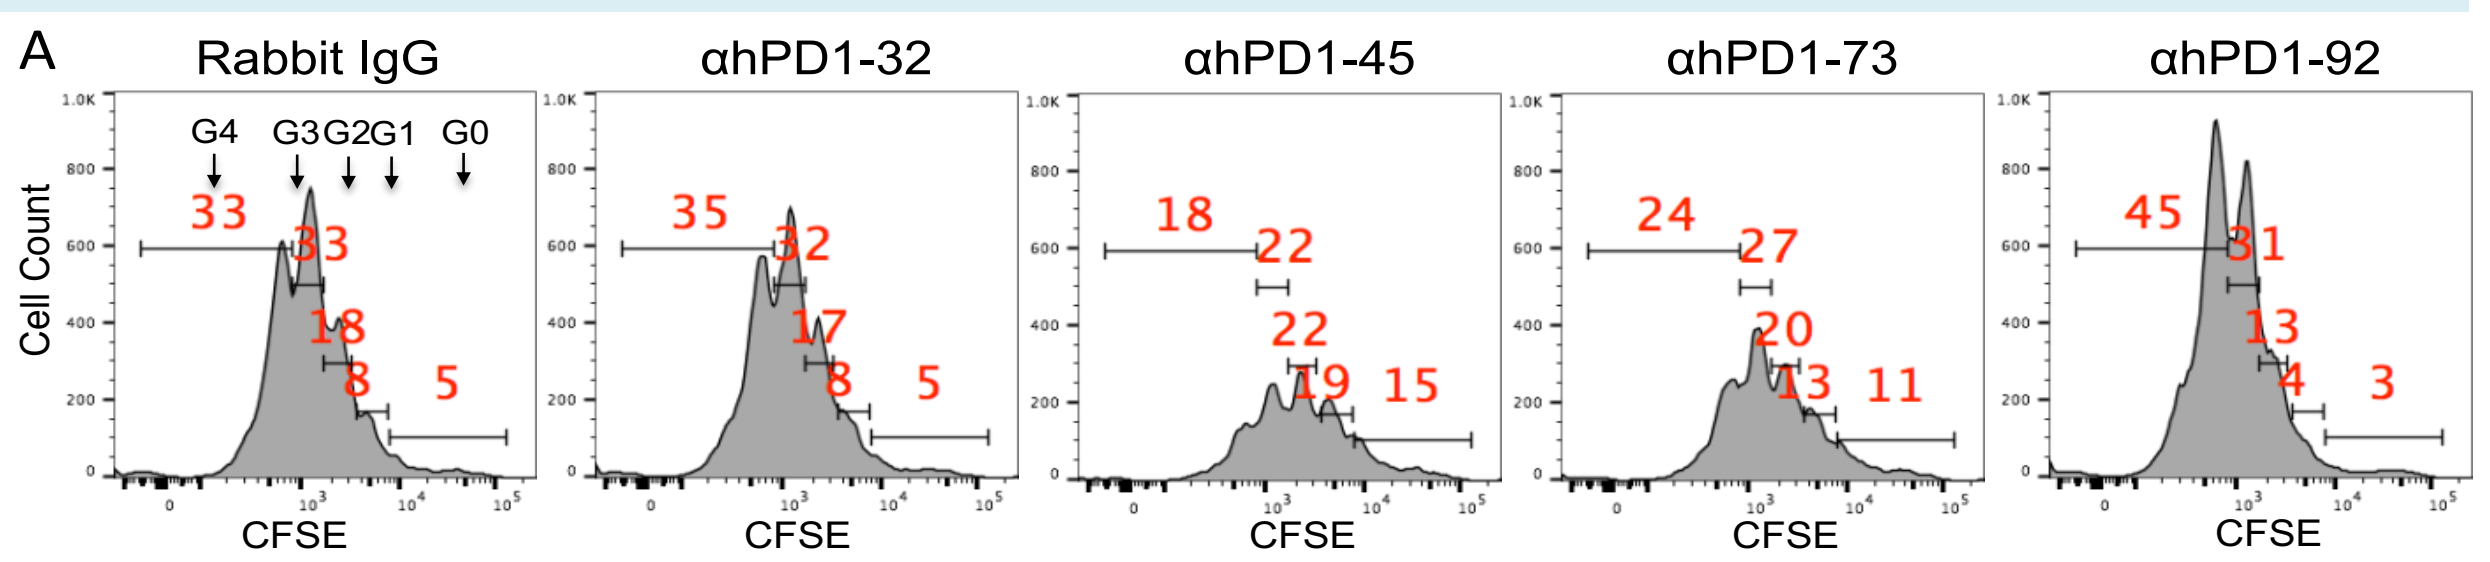
A
Rabbit IgG
αhPD1-32
αhPD1-45
αhPD1-73
αhPD1-92
Cell Count
G4 G3 G2 G1 G0
33 33 18 8 5
35 32 17 8 5
18 22 22 19 15
24 27 20 13 11
45 31 13 4 3
CFSE

CFSE labeled splenocytes from TCR transgenic mice were activated with MBP Ac1-11 for 4 days, in the presence of 50µg/ml of αhPD-1 antibodies. CFSE was analyzed by flow cytometry. Cells were gated on CD4+ cells. Data show that **81% of α-hPD-1-92-110 treated CD4 T cells are highly proliferating vs control IgG** suggesting **α-hPD-1-92-110 blocks PD-1/PD-L1 interaction and leads to enhanced T effector function.** Contrarily, **α-hPD-1(45-64) or α-hPD-1(73-90) treated T cells show decreased proliferation**, suggesting they may activate PD-1 signaling

Immunogenicity of individual MVF-PD-1 peptides in Balb/c mice

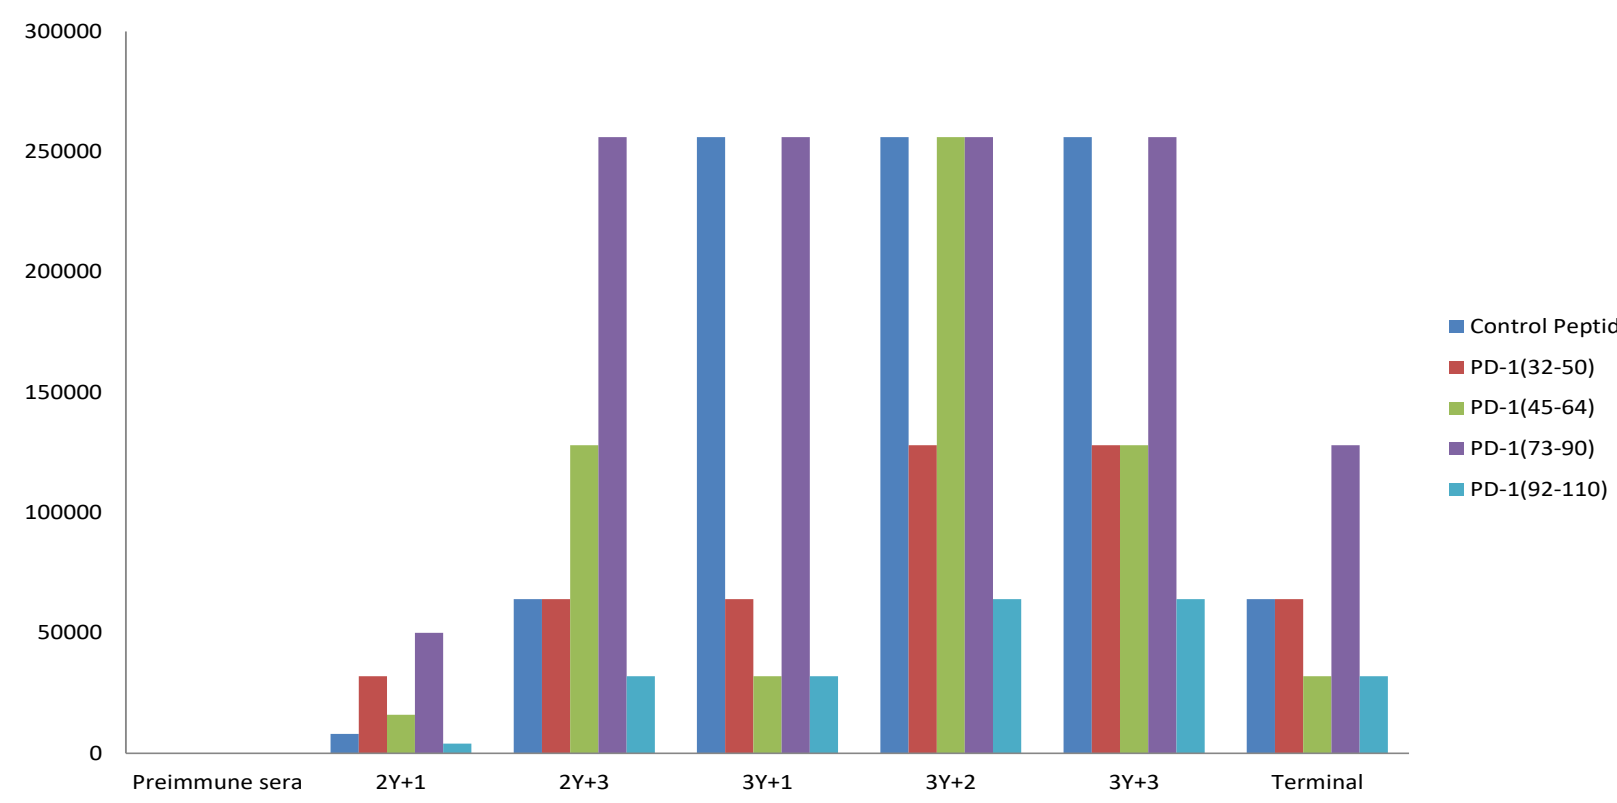
Control Peptide
PD-1(32-50)
PD-1(45-64)
PD-1(73-90)
PD-1(92-110)
Preimmune sera
2Y+1
2Y+3
3Y+1
3Y+2
3Y+3
Terminal

Immunogenicity of PD-1 vaccines The PD-1 peptide vaccines were emulsified in Montanide ISA 720 (1:1) and 100 µg nor-MDP Female Balb/c mice (Charles River Laboratories) at the age of 5 to 6 wk were immunized three times at 3-wk intervals with 100 µg peptide vaccine, and 15 d after the third immunization, the mice were challenged s.c. with **CT-26 tumor cells** (100,000 per mouse Sera pools 2Y+1, 2Y+3, 3Y+1, and 3Y+2 were tested by ELISA**. Each PD-1 epitopes elicited high titers of antipeptide antibodies**

Combination of two HER-2 vaccine: B-Vaxx

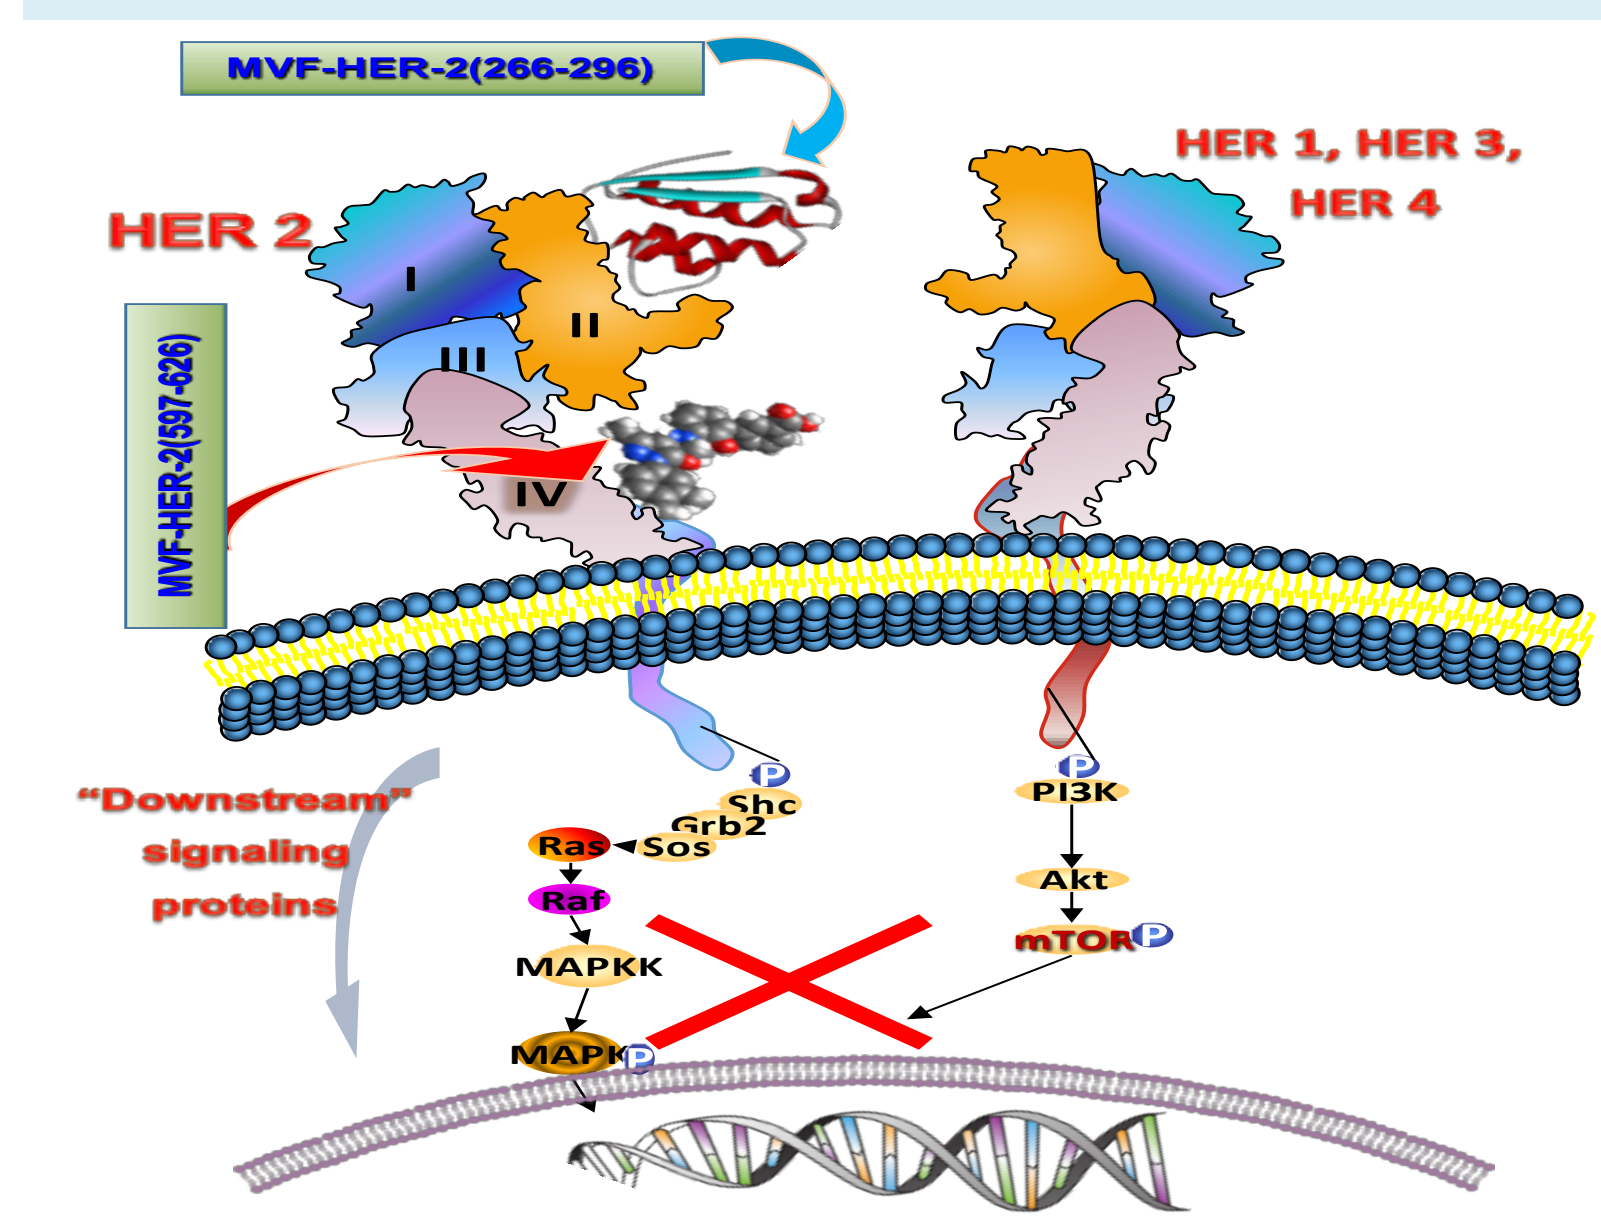
MVF-HER-2(266-296)
HER 2
HER 1, HER 3, HER 4
MVF-HER-2(597-626)
"Downstream" signaling proteins
Shc
Grb2
Ras
Sos
Raf
MAPKK
MAPK
PI3K
Akt
mTOR

Clinical trial MVF-HER-2 peptides: Trastuzumab-binding epitope (597-626) and pertuzumab-binding epitope (266-296) elicit high affinity antipeptide. The anti-HER-2 266-296) antibodies bind domain II whereas anti HER-2 (597-626) bind domain IV consistent with their mechanism of action

1. Allen, S & Kaumaya, PTP *et al.*, (2007) J. Immunol. 178 (1) 472-482; 2, Garrett, J and Kaumaya PTP *et al.*, (2007) J. Immunol. 178 (11),7120-7130 .

Immunogenicity of B-Vaxx + Key-Vaxx

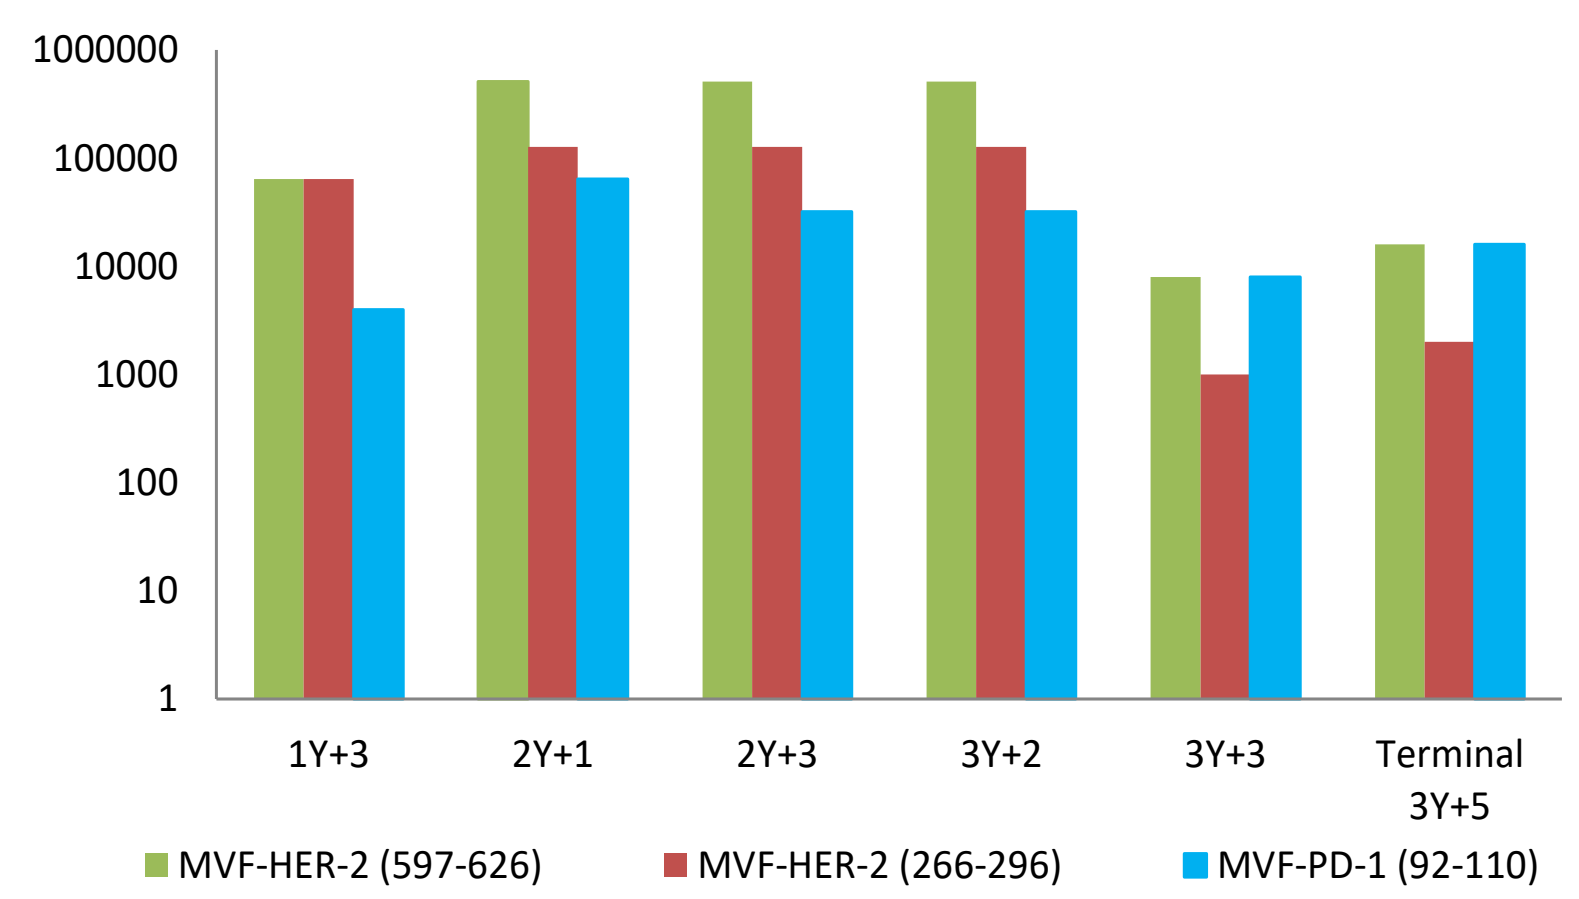
1Y+3
2Y+1
2Y+3
3Y+2
3Y+3
Terminal 3Y+5
MVF-HER-2 (597-626)
MVF-HER-2 (266-296)
MVF-PD-1 (92-110)

Immunogenicity of Triple immunized BALB/c mice monitoring: Sera pools from BALB/c mice immunized with 3 peptides **[HER-2(266-296)**, **HER-2(597-626)**, & **PD-1(92-110)]** were tested by ELISA. Sera samples 1Y+3, 2Y+1, 2Y+3, and 3Y+2 were taken before CT26/Her-2 tumor challenge. Samples 3Y+3 and 3Y+5 were taken at 1 week and 3 weeks post challenge respectively. **Robust HER-2 and PD-1 antibody responses were elicited in all vaccinated mice with predominantly IgG1 antibodies**

Composite Data CT-26 Tumor LWH

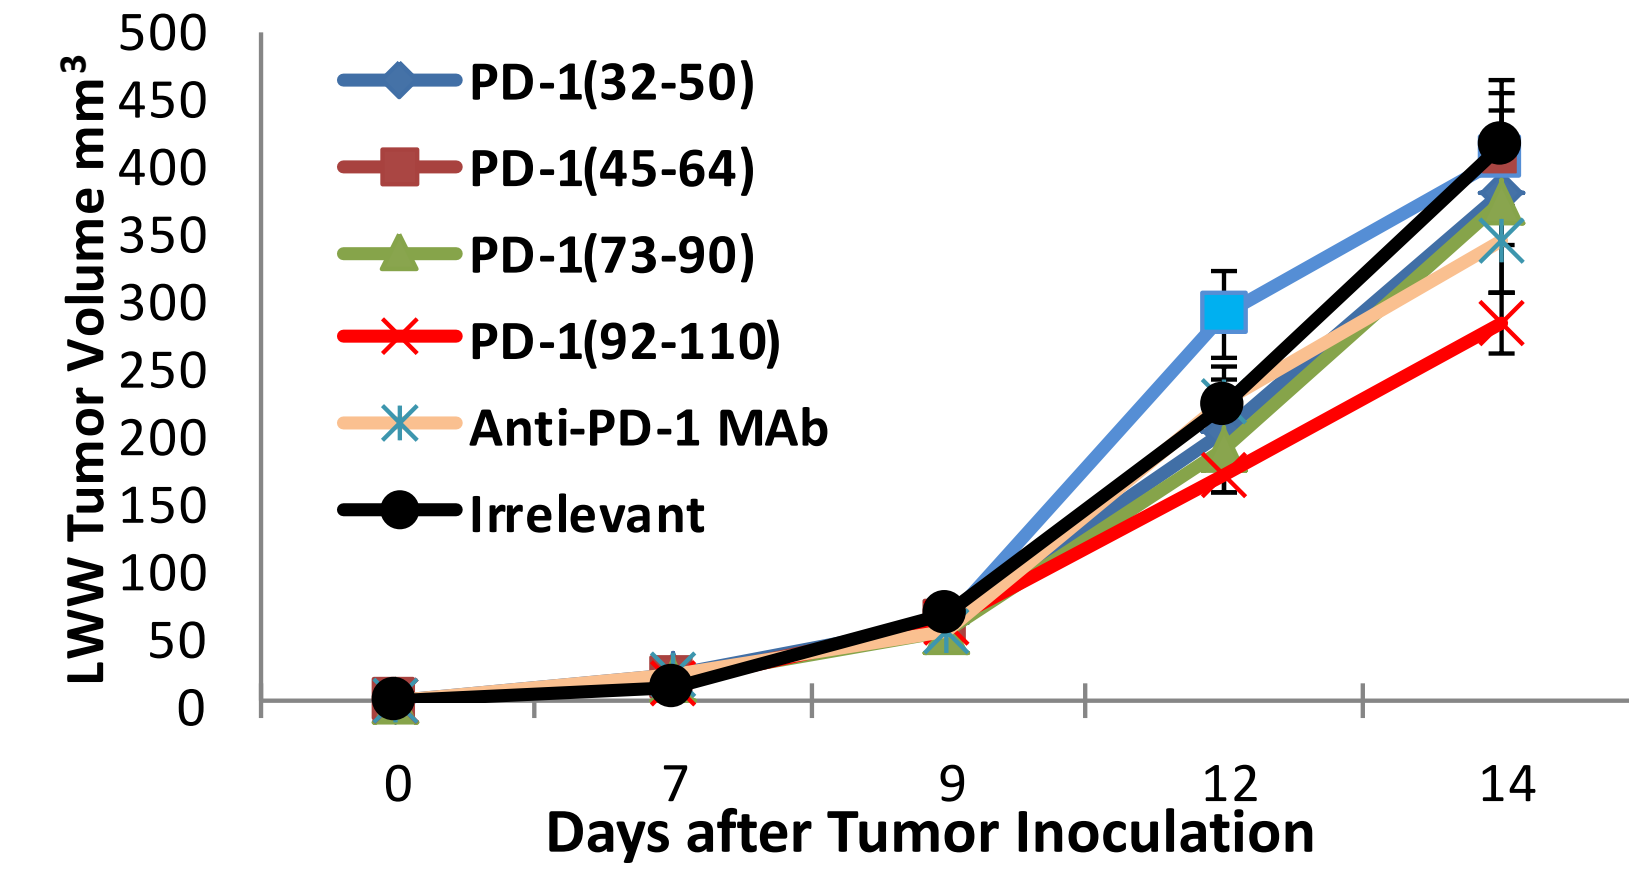
LWW Tumor Volume mm3
PD-1(32-50)
PD-1(45-64)
PD-1(73-90)
PD-1(92-110)
Anti-PD-1 MAb
Irrelevant
Days after Tumor Inoculation

Mean Plots of tumor growth in syngeneic Balb/c mice (5/gps) immunized with four PD-1 MVF-vaccine constructs: (PD-1(32-50), PD-1 (45-64), PD-1 (73-90) PD-1 (92-110), -ve control (irrelevant peptide); +ve control anti-mouse PD-1 MAb (29F.1A12). Mice were challenged 15 days after 3rd vaccination with CT26 carcinoma cells (1x10^5) and tumor growth evaluated. **Mice vaccinated with MVF-PD-1(92-110) showed significant inhibition of tumor growth at Day 14 suggesting that this epitope could be a useful inhibitory vaccine.**

Vaccinations with combo Key-Vaxx and B-Vaxx followed by CT26/HER-2 challenge

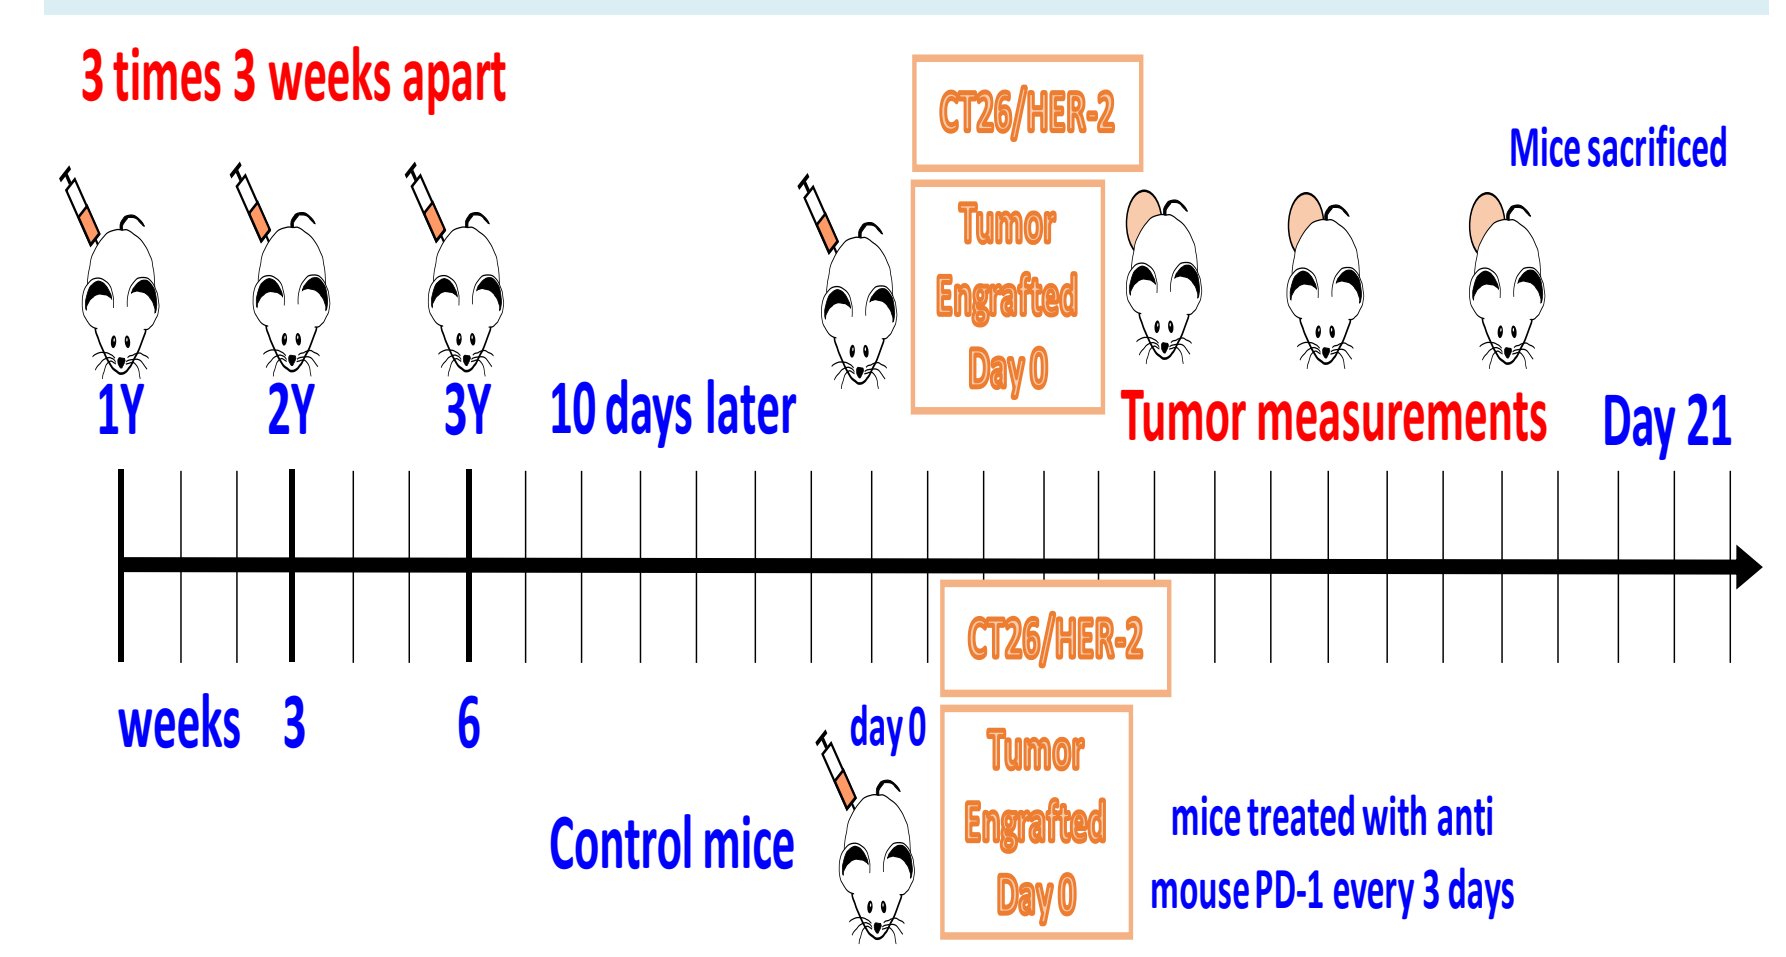
3 times 3 weeks apart
CT26/HER-2
Tumor Engrafted Day 0
Mice sacrificed
1Y
2Y
3Y
10 days later
Tumor measurements
Day 21
weeks 3 6
day 0
Control mice
mice treated with anti mouse PD-1 every 3 days

Vaccination and Challenge Scheme. Balb/c mice (**10 mice/gp**) were immunized with combination of MVF-PD-1 (92-110), MVF-HER-2 (266-296)+MVF-HER-2 (597-626) peptide vaccine constructs emulsified with nor-MDP and ISA 720. Animals were boosted twice at 3 weeks interval. Antibody titers were determined by ELISA. 2 weeks after the final boost 1x10^5 tumor cells from CT26/HER-2 tumor lines were transplanted s.c. Control mice either were challenged with 1x10^5 tumor cells and treated with anti-PD-1 antibody (29F.1A12) twice a week for the duration of the experiment.

Synergistic growth inhibition with triple vaccine in Balb/c-CT26/HER-2 model

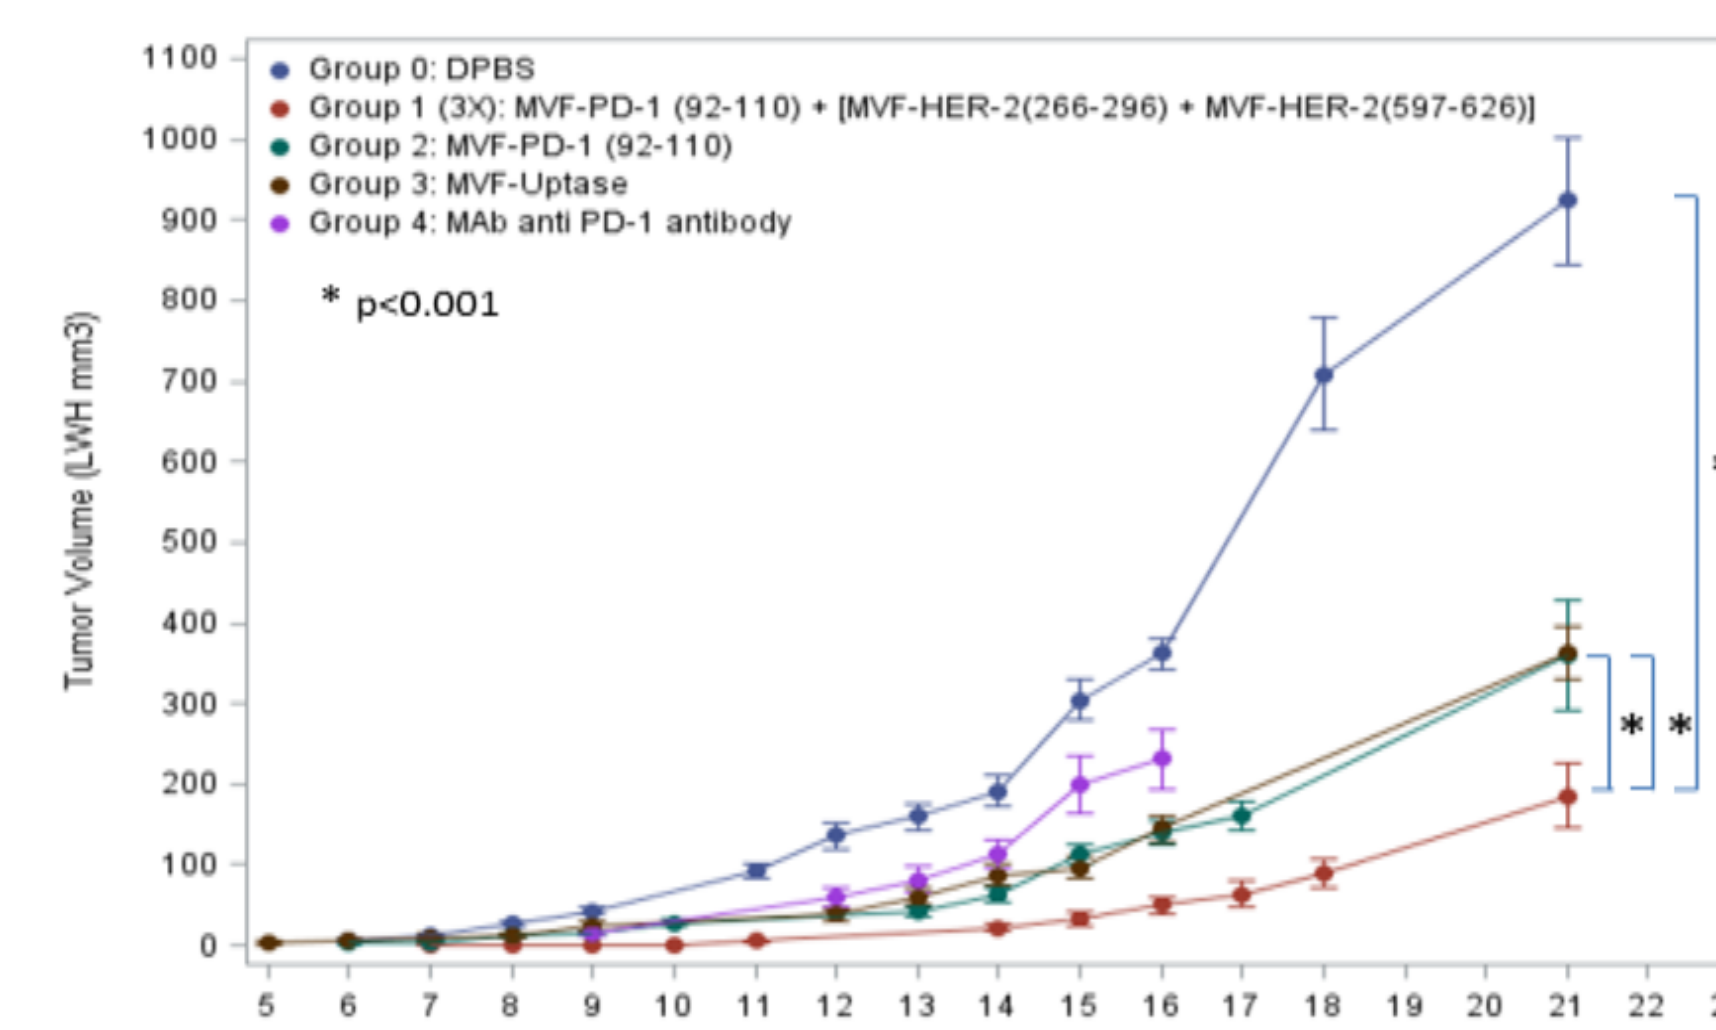
Group 0: DPBS
Group 1 (3X): MVF-PD-1 (92-110) + [MVF-HER-2(266-296) + MVF-HER-2(597-626)]
Group 2: MVF-PD-1 (92-110)
Group 3: MVF-Uptase
Group 4: MAb anti PD-1 antibody
* p<0.001
Tumor Volume (LWH mm3)
Days After Challenge

Results demonstrate that the Triple combo peptide vaccine HER-2 (266) + HER-2 (597) + PD-1 (92) causes significant (*p* <0.001) inhibition of tumor growth in BALB/c challenged with colon carcinoma cell line CT26/HER-2 compared to +ve control α-mouse PD-1 mAb (29F.1A12). MVF-PD-1 (92-110). –ve control PBS or irrelevant peptide.

B-Vaxx and KEY-Vaxx proposed mechanism of action

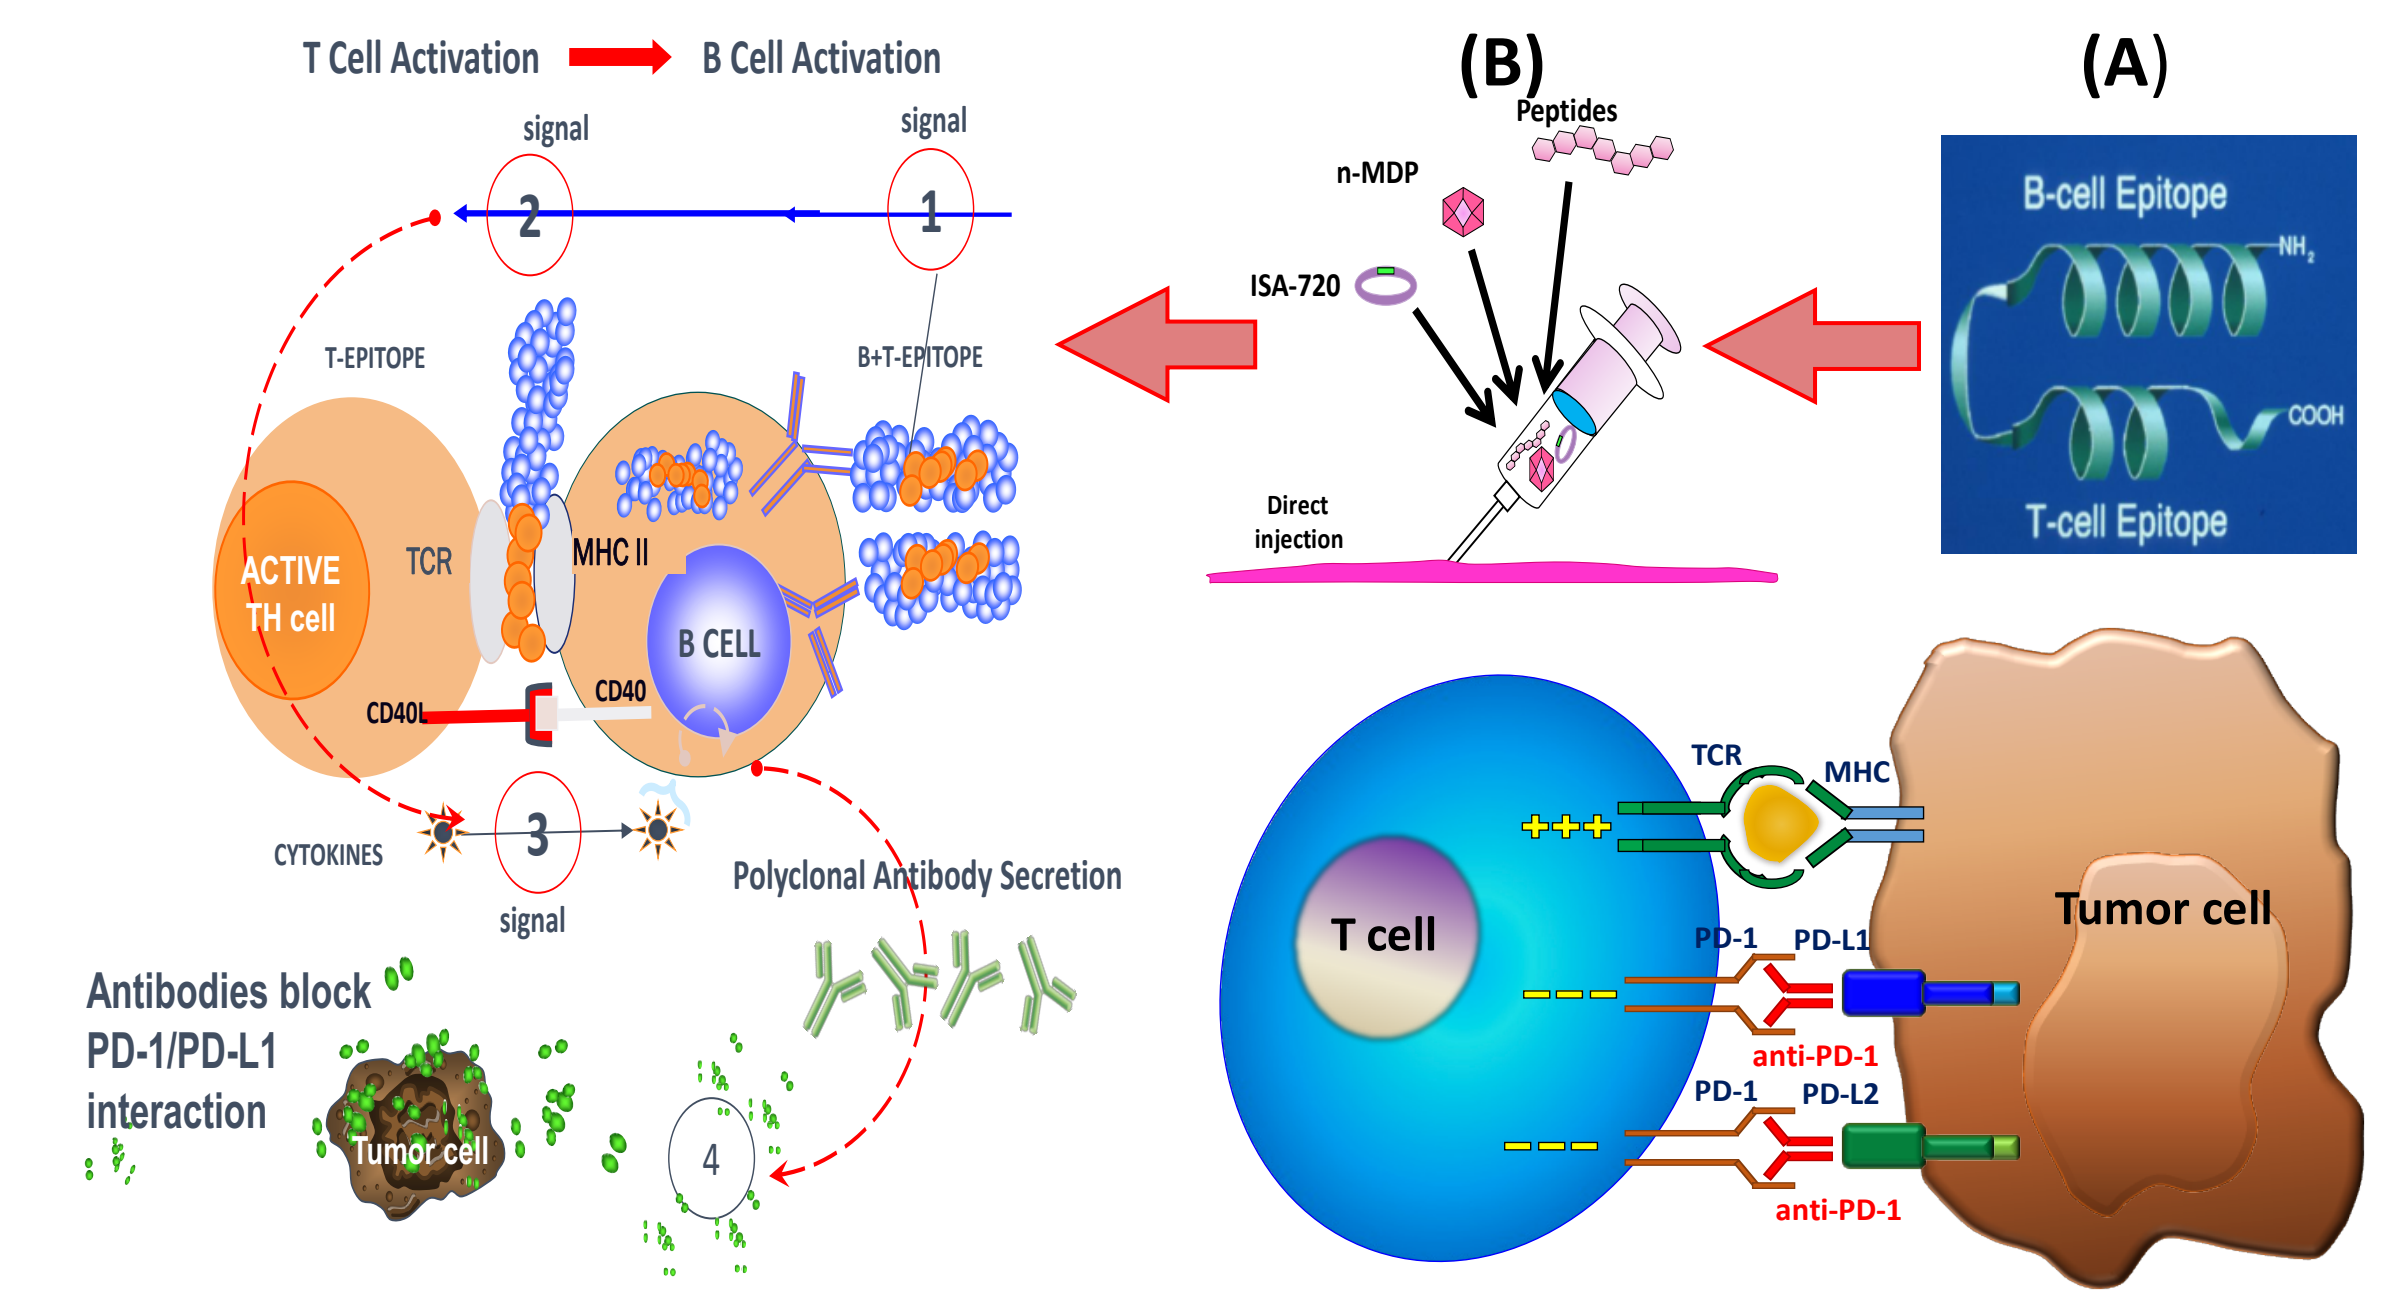
T Cell Activation
B Cell Activation
signal
T-EPITOPE
B+T-EPITOPE
ACTIVE TH cell
TCR
MHC II
B CELL
CD40L
CD40
CYTOKINES
Polyclonal Antibody Secretion
Antibodies block PD-1/PD-L1 interaction
Tumor cell
(B)
Peptides
n-MDP
ISA-720
Direct injection
(A)
B-cell Epitope
T-cell Epitope
T cell
TCR
MHC
PD-1
PD-L1
PD-L2
anti-PD-1
Tumor cell

Anti-PD-1(92-110) and anti- HER-2 antibodies

Anti-PD-1(92-110): Key-Vaxx inhibit

Chimeric PD-1 or HER-2 peptides incorporating an Measles virus fusion protein epitope (MVF) "promiscuous" T cell epitope **(A)** when emulsified with nor-MDP adjuvant in ISA720 vehicle **(B)** elicit polyclonal antibodies. The B & T epitope is presented to antigen presenting cells (APCs) or B cells **(1)** without processing, the T cell epitope binds MHC class II and activates the T cell **(2)**. Cytokines are liberated **(3)** to help B cells make antipeptide polyclonal antibodies. Both B-Vaxx and Key-Vaxx follow similar pathway. The Her-Vaxx antibodies binds and eliminate the tumor cells overexpressing HER-2. The Key-Vax antibodies inhibiting interaction with PD-L1 on tumor cell and promotes T cell activation.

PD-1 Vaccines are safe and do not exhibit toxicity or autoimmunity

- All mice vaccinated over a period of 9 weeks showed no signs of scruffiness, lesions, and lethargy
- Organs (spleen, liver, heart, lung, kidney, and tumor) from the Balb/c mice vaccinated with combination peptides (HER-2 and PD-1) were collected from mice and submitted for analysis at the Comparative Pathology & Mouse Phenotyping Core facility:
 - No significant lesions were noted in any of the organs
 - There were also no overt biochemical abnormalities noted.
 - All mice had hepatocellular vacuolation consistent with glycogenosis. Glycogen accumulation in the liver is interpreted to be a normal finding, and varies depending on the physiological state of the animal. Glycogen accumulation can also be observed as a manifestation of toxicity or with glycogen storage diseases.

Results and Conclusions

- Robust HER-2 and PD-1 antibody responses in all vaccinated mice.
- Combined vaccination is effective in reducing tumor growth in a Balb/c syngeneic model of colon carcinoma
- Triple vaccination is more effective than PD-1 vaccine alone
- Triple Vaccination efficacy greater than the positive control gold standard anti-mouse PD-1 monoclonal antibody (29F.1A12).
- The vaccine combination was found to be safe and did not appear to exhibit toxicity or autoimmunity.
- Triple combination vaccine provides synergistic inhibition of tumor growth with no evidence of toxicity or autoimmunity.
- A Phase 1b human clinical trial with the PD-1 vaccine is under planning **(IMUGENE).**

References

1: Kaumaya PTP. Hum ***Vaccin Immunother***. 2015;11(6):1368-86.

2: Bekaii-Saab,T and Kaumaya PTP et al., Online First on February 25, 2019; DOI: 10.1158/1078-0432. ***Clinical Cancer Research*** (CCR-18-3997)

Acknowledgements

Funding NIH R01 CA CA84356 & NIH R21 CA13508 to PTPK