Active immunization with immune checkpoint inhibitors-mimotope elicits strong *in vivo* anti-tumor effect against Her-2/neu-expressing tumors

Joshua Tobias[1*], C. Battin[1], A. De Sousa Linhares[1], M. Lebens[2], K. Baier[1], K. Ambroz[1], M. Drinic[1], S. Högler[3], A. Inic-Kanada[1], E. Garner-Spitzer[1], M. Preusser[1,4], L. Kenner[3], M. Kundi[1], C. C. Zielinski[1,4], P. Steinberger[1], U. Wiedermann[1*]

Introduction and Aim

Immunotherapy based on the application of monoclonal antibodies (mAbs) as immune checkpoint inhibitors (ICIs) has demonstrated remarkable clinical activity in various malignancies. The cost intensiveness as a possible limiting factor of such approach, however, may be overcome by direct immunization with mimotopes (B cell epitopes) of the corresponding ICIs. By establishing a platform, involving overlapping peptides spanning the extracellular domain of PD1, T cell-based cellular assay and a syngeneic mouse model with tumor expressing Her-2/neu, mimotopes of the anti-human PD1 mAb Nivolumab (as an example) and of anti-mouse PD1 mAb (for evaluating anti-tumor activity *in vivo*) were identified and characterized.

Aim: To generate ICIs mimotopes with strong capacity in inducing antibody response with anti-tumor activity.

In vitro results: Specificity of the identified mimotopes

Nivolumab mimotope (JT-N1) specifically and dose-dependently inhibits the interaction between human PD1 and PD-L1 (in a T cell-based assay)

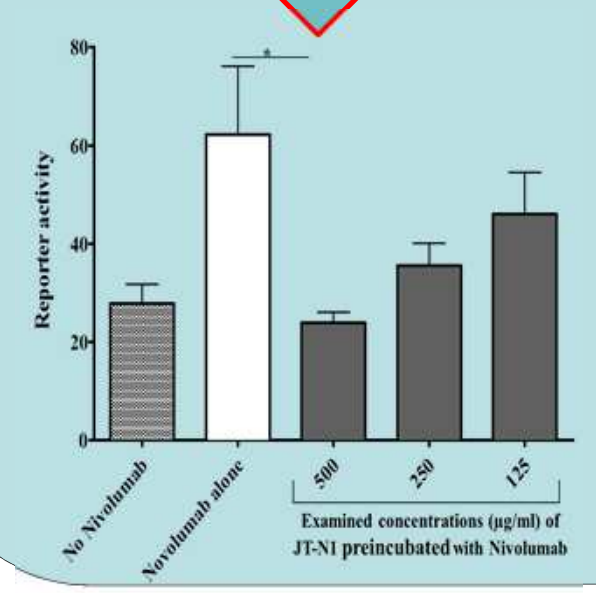
Reporter activity
80
60
40
20
0
*
No Nivolumab
Novolumab alone
500
250
125
Examined concentrations (µg/ml) of JT-N1 preincubated with Nivolumab

Specific IgG against mouse PD1-derived mimotope (JT-mPD1) inhibits the interaction between mouse PD1 and PD-L1 (in an ELISA assay)

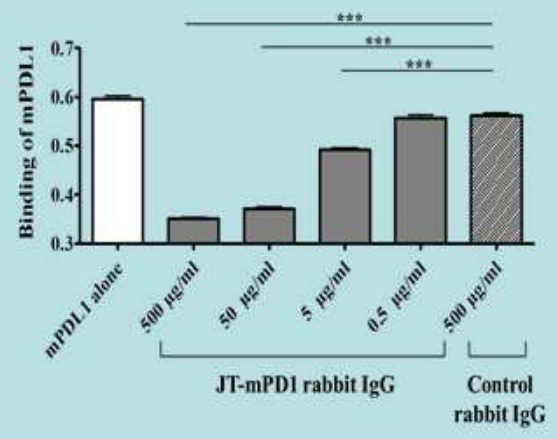
Binding of mPDL1
0.7
0.6
0.5
0.4
0.3

mPDL1 alone
500 µg/ml
50 µg/ml
5 µg/ml
0.5 µg/ml
500 µg/ml
JT-mPD1 rabbit IgG
Control rabbit IgG

In vivo results: Anti-tumor evaluation in syngeneic mouse model with tumors expressing human Her-2/neu

Passive and active immunization with mouse PD1-derived mimotope (JT-mPD1)

Passive immunization with specific IgG against the mimotope induces reduction of tumor progression

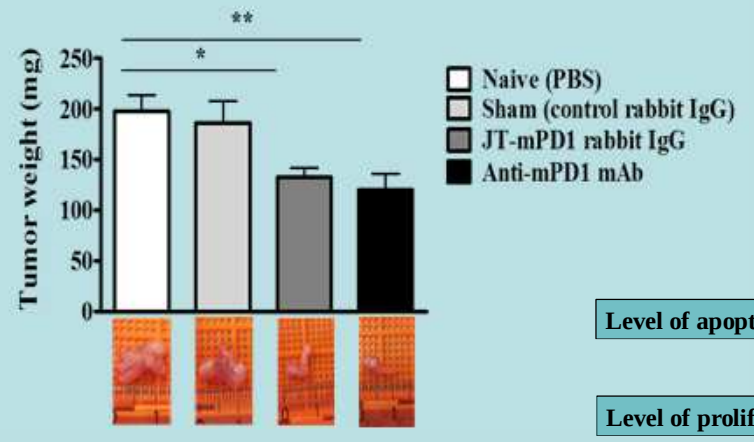
Tumor weight (mg)
250
200
150
100
50
0
**
*
Naive (PBS)
Sham (control rabbit IgG)
JT-mPD1 rabbit IgG
Anti-mPD1 mAb

Active immunization with the mimotope induces reduction of tumor progression, as well as apoptotic and anti-proliferative effect

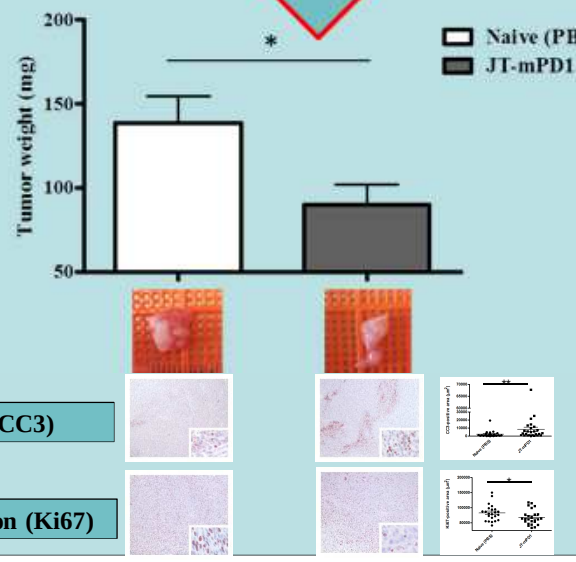
Tumor weight (mg)
200
150
100
50
*
Naive (PBS)
JT-mPD1
Level of apoptosis (CC3)
Level of proliferation (Ki67)

Combination therapy

Pre-liminary results:
Active immunization with PD1-derived mimotope increases the anti-tumor activity of P467* (Her-Vaxx; multipeptide-based anti-Her-2/neu vaccine)

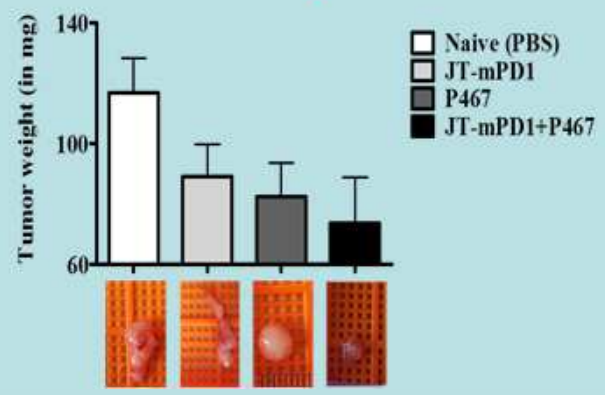
Tumor weight (in mg)
140
100
60
Naive (PBS)
JT-mPD1
P467
JT-mPD1+P467

* Tobias et al, 2017, BMC Cancer.

Summary and conclusion

Our results show:

1. Identification of PD1-derived mimotopes

2. Immunization with PD1-derived mimotope induces antibodies with anti-tumor activity (passive immunization) comparable to the corresponding ICI.

3. Active immunization with PD1-derived mimotope induces anti-tumor activity in vivo with increased apoptotic and anti-proliferative effect in the tumors.

4. Active immunization with PD1-derived mimotope combined with the second generation of our anti-Her-2/neu vaccine (Her-Vaxx) increases the anti-tumor effect of the vaccine compared to each antigen alone.

Our results imply active immunization with mimotopes of ICIs as future treatment strategies with the corresponding ICIs, and their use either as monovalent vaccines or in combination with tumor specific antigens for enhancing the efficacy of the vaccinations against different malignancies.

[1] Medical University of Vienna, Austria, [2] University of Gothenburg, Sweden, [3] University of Veterinary Medicine, Vienna, Autsria [4] Vienna Cancer Center, Austria.

[*] Conflict of interests: MP, CCZ, PS and UW declare potential conflicts of interests. [**] The study was supported by a research grant from Imugene (Australia) to the Medical University of Vienna.

[***] **Contact: joshua.tobias@meduniwien.ac.at, ursula.wiedermann@meduniwien.ac.at**