CF33-CD19T ONCOLYTIC VIRUS (onCARlytics) IN COMBINATION WITH OFF-THE-SHELF ALLOGENEIC CYCART-19 T-CELLS TARGETING DE NOVO CD19T EXPRESSING TUMORS

Anthony K. Park[1], Isabel Monroy[1], Colin Cook[2], Shuyang He[3], Kathy Karasiewicz[3], Monil Shah[4], Leslie M.O. Chong[4], Nimali P. Withana[4], Robert Hariri[3], Yuman Fong[2], and Saul J. Priceman[1]

[1] Department of Hematology and Hematopoietic Cell Transplantation, Beckman Research Institute, City of Hope National Medical Center, Duarte, CA 91010 USA
[2] Department of Surgery, Division of Surgical Oncology, City of Hope National Medical Center, Duarte, CA 91010 USA
[3] Celularity Inc., Florham Park, NJ 07932
[4] Imugene Limited, Sydney, Australia

IMUGENE
Developing Cancer Immunotherapies

celularity® City of Hope®

Introduction

Autologous chimeric antigen receptor (CAR) T Cell therapy has shown impressive clinical responses against CD19+ B-Cell hematological malignancies and is being actively explored in the treatment of solid tumors. However, several barriers have precluded therapeutic responses in solid tumors, including limited tumor-restricted CAR targets and the immunosuppressive tumor microenvironment. We have recently reported the successful combination immunotherapy using a novel chimeric vaccinia-based oncolytic virus (OV), called onCARlytics (Imugene Limited), that is engineered to express a non-signaling, truncated CD19 (CD19t) antigen for tumor-selective delivery, enabling de novo targeting of tumor cells by autologous CD19-CAR T Cell. One of the field's unanswered questions is whether treatment-naïve allogeneic CAR T Cell are superior to cancer patient-derived T-Cells for product manufacturing to improve overall responses against solid tumors.

Here, we evaluated this combination strategy using two allogeneic CAR T Cell products generated from peripheral blood mononuclear cells (PBMC) and placental T-Cells, respectively. PBMC-derived CAR T Cell were manufactured from normal, healthy donors. CYCART-19 (Celularity®, Inc.) Cells were derived from postpartum human placental T-Cells that are genetically modified to express the CD19-CAR followed by CRISPR-Cas9- mediated knockout of the endogenous TCR and expanded to produce multiple doses of allogeneic "off the shelf" treatment.

CYCART-19 T-Cells induced potent cytolytic activity against solid tumor cells infected with onCARlytics. Interestingly, while we observed comparable anti-tumor activity between PBMC-derived CD19-CAR T Cells and CYCART-19, significant differences in cytokine secretion were detected. This warrants the possibility that the placental-derived CAR T product may elicit reduced CRS potential in patients with maintained or improved efficacy. This combination approach demonstrated impressive in vivo anti-tumor response in human tumor xenograft models. In summary, our results have demonstrated that further development of this combination immunotherapy for the potential treatment of a wide array of solid tumors is warranted.

Figure 1

Delivering truncated CD19t (CD19t) to tumor cells using oncolytic virus (OV) as a target for CD19-CAR T Cell.

onCARlytics selectively infect solid tumor cells and deliver truncated CD19 (CD19t) as a target for CD19-CAR T Cell.

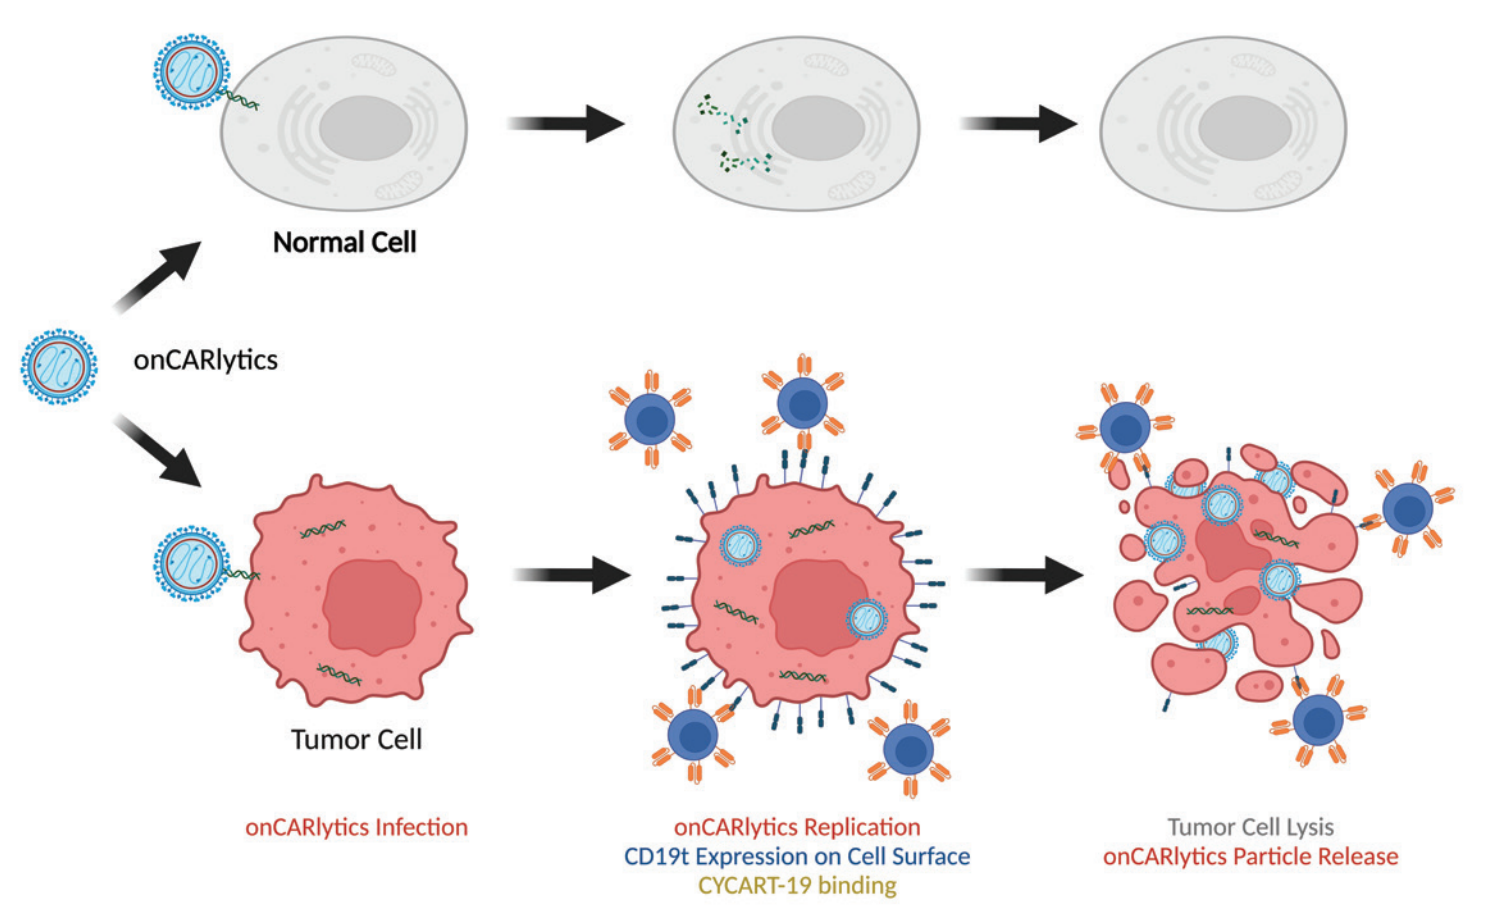

Normal Cell
onCARlytics
Tumor Cell
onCARlytics Infection
onCARlytics Replication
CD19t Expression on Cell Surface
CYCART-19 binding
Tumor Cell Lysis
onCARlytics Particle Release

Figure 2

Postpartum human placental derived allogeneic T-Cells expressing CAR–CYCART-19

Celularity® has developed an allogeneic placental T-Cell with knockout of endogenous T-Cell receptors, derived from postpartum human placenta expressing CD19-CAR called CYCART-19. Placental-derived T-Cells are mostly naïve (CD45RA+ CCR7+), expand readily ex vivo, express markers of stem cell memory, and have lower expression of effector or exhaustion markers, which has been associated with greater stemness, enhanced proliferative capacity, and increased persistence in vivo.

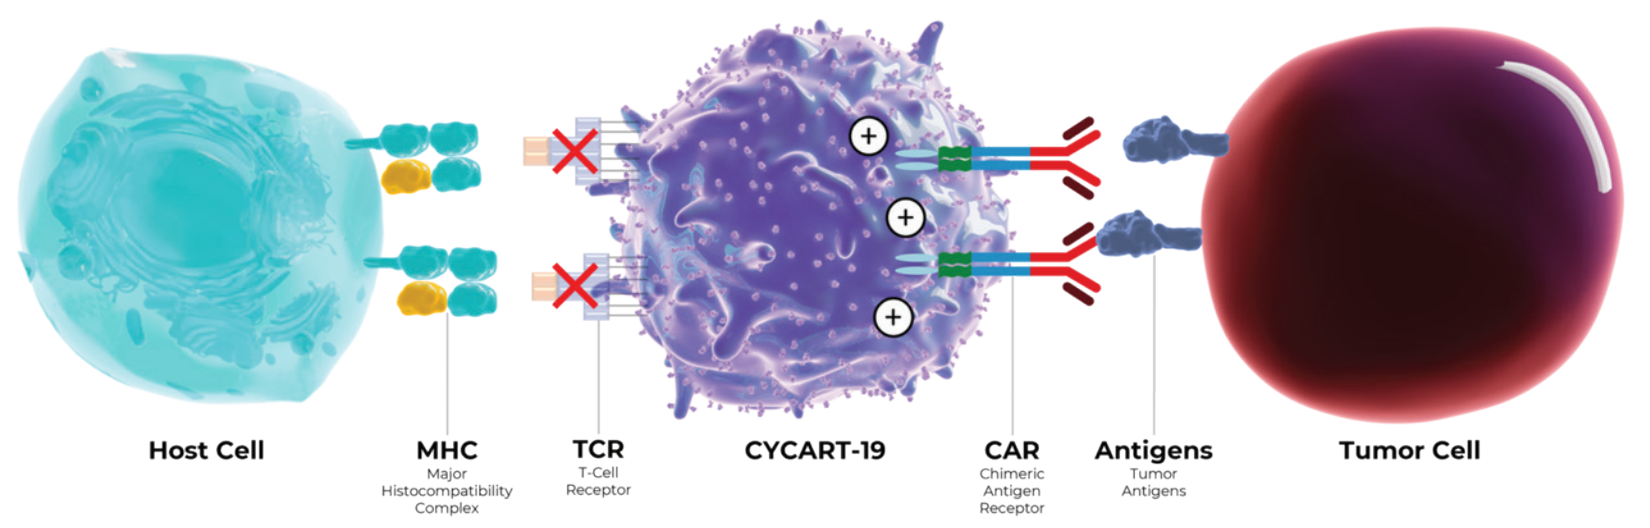

Host Cell
MHC
Major Histocompatibility Complex
TCR
T-Cell Receptor
CYCART-19
CAR
Chimeric Antigen Receptor
Antigens
Tumor Antigens
Tumor Cell

Figure 3

Specific CYCART-19 tumor cell killing following onCARlytics infection

A Bright-field microscopy (10X magnification) of MDA-MB-468 tumor cells at 24h following onCARlytics infection or MDA-MB-468-CD19t (positive control lentivirally transduced to stably express CD19t) in the presence of untransduced (NT) or CYCART-19 T-Cells. B In vitro killing assay at 24h and C 48h of MDA-MB-468 tumor cells infected with onCARlytics and treated with untransduced autologous T-Cells, autologous CD19-CAR T Cell, NT (1 donor), or CYCART-19 (3 donors) T-Cells. Graph on the left represents tumor killing, and in the middle represents CD19t expression on tumor cells. Graph on the right represents tumor count against MDA-MB-468-CD19t treated with untransduced autologous T-Cells, autologous CD19-CAR T Cell, NT (1 donor), or CYCART-19 (3 donors) T-Cells.

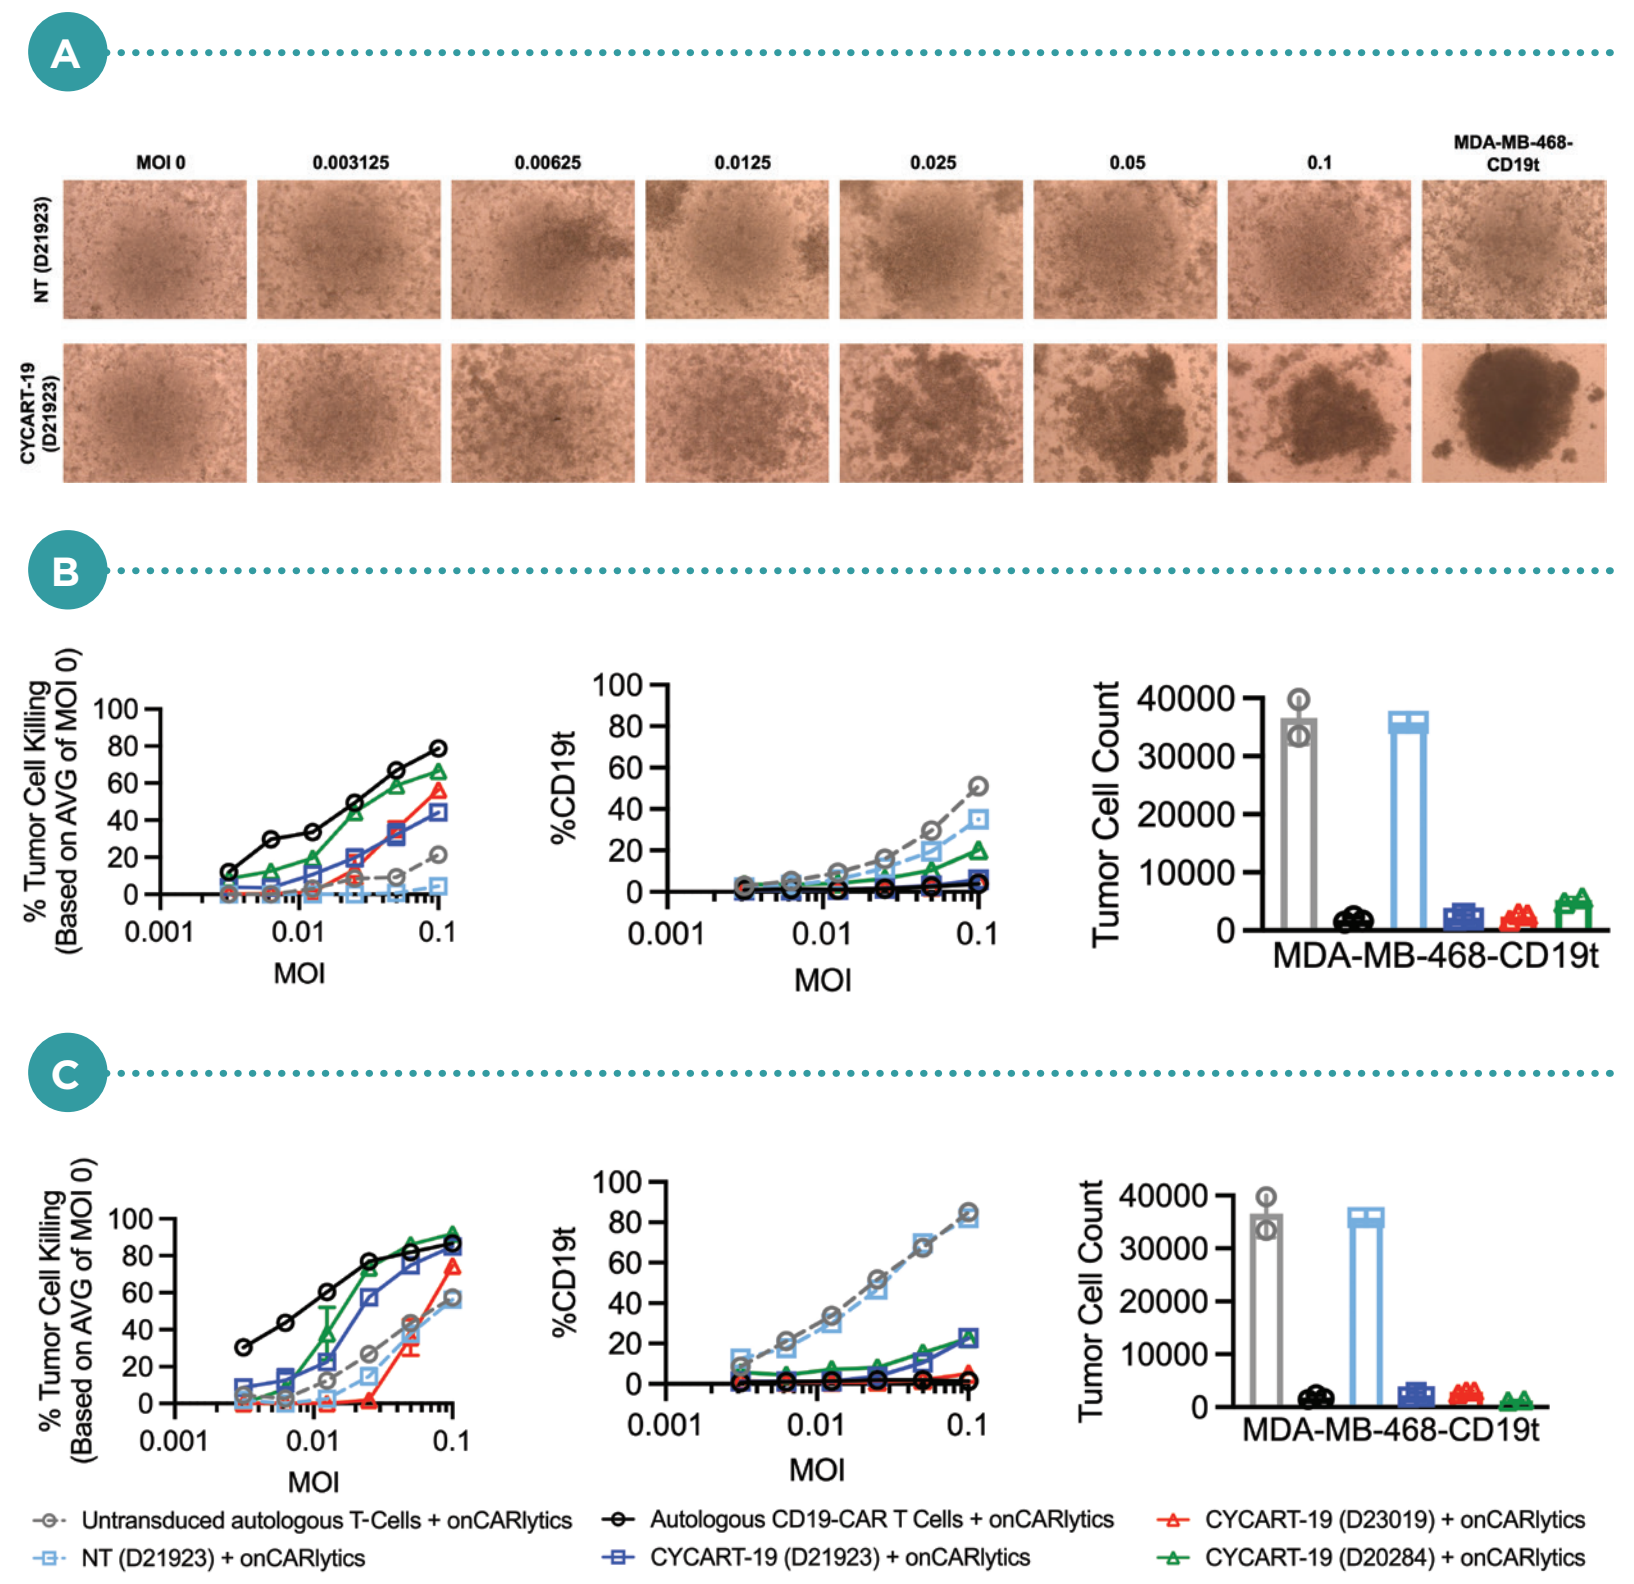

A
MOI 0
0.003125
0.00625
0.0125
0.025
0.05
0.1
MDA-MB-468-CD19t
NT (D21923)
CYCART-19 (D21923)
B
% Tumor Cell Killing (Based on AVG of MOI 0)
%CD19t
Tumor Cell Count
MOI
MDA-MB-468-CD19t
C
Untransduced autologous T-Cells + onCARlytics
NT (D21923) + onCARlytics
Autologous CD19-CAR T Cells + onCARlytics
CYCART-19 (D21923) + onCARlytics
CYCART-19 (D23019) + onCARlytics
CYCART-19 (D20284) + onCARlytics

Figure 4

Activation of CYCART-19 by targeting of tumor cells expressing CD19t following onCARlytics infection

A Expression of activation marker (CD137) on untransduced autologous T-Cells, autologous CD19-CAR T Cell, untransduced [NT] (1 donor), or CYCART-19 (3 donors) T-Cells following 24h (left) and 48h (right) in vitro co-culture with MDA-MB-468 tumor cells infected with onCARlytics. B IFNγ and C IL-2 production following in vitro infection of MDA-MB-468 tumor cells with onCARlytics in the presence of autologous untransduced, autologous CD19-CAR, NT, or CYCART-19 T-Cell measured at 24h (left) and 48h (right) by ELISA. D IFNγ and E IL-2 production following in vitro co-culture of MDA-MB-468-CD19t with autologous untransduced, autologous CD19-CAR, NT (1 donor), or CYCART-19 (1 donors) T-Cells measured at 24h (left) and 48h (right) by ELISA.

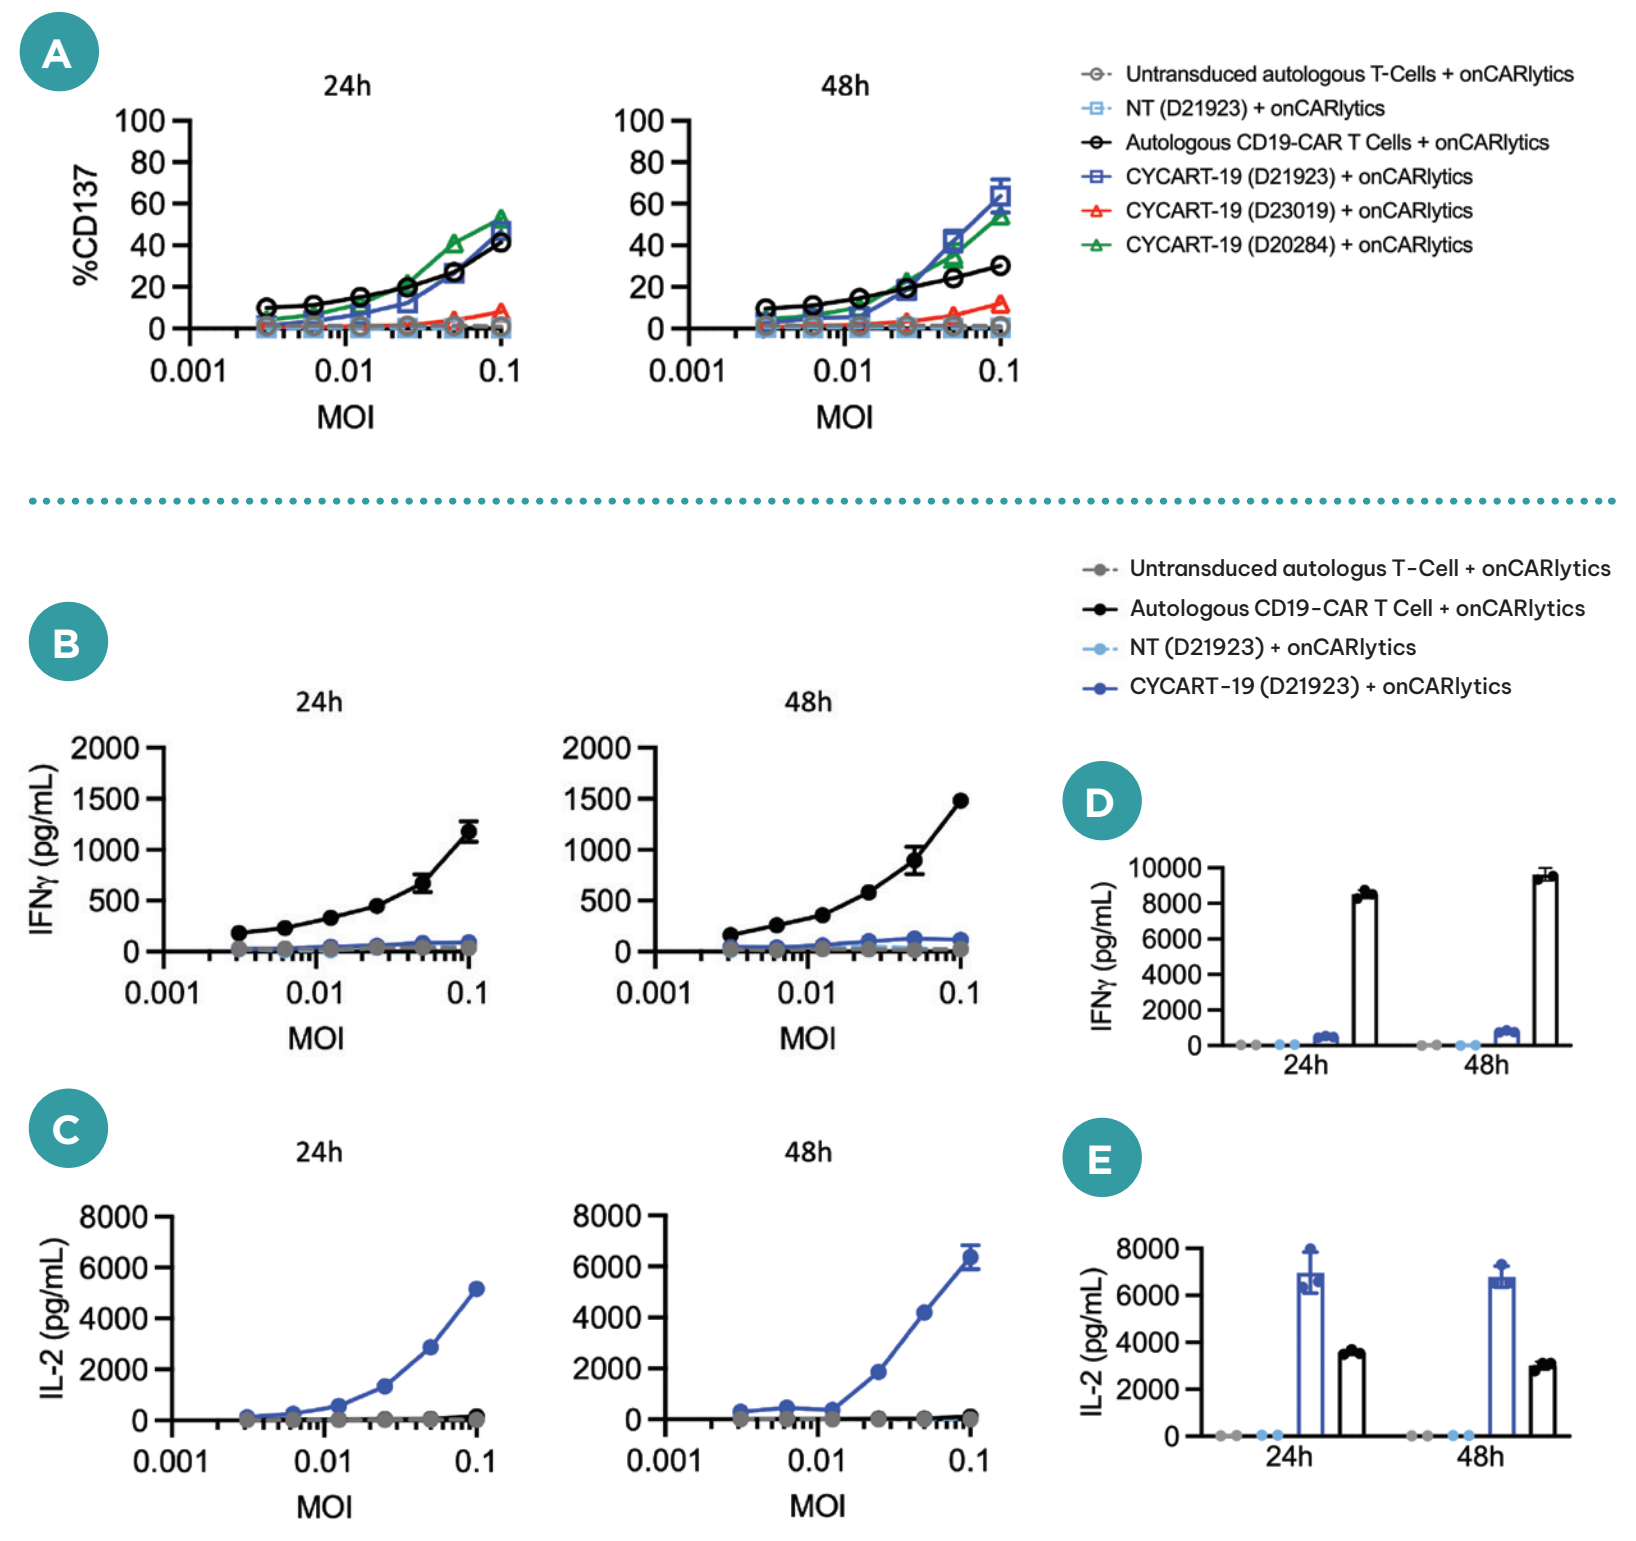

A
24h
48h
%CD137
MOI
Untransduced autologous T-Cells + onCARlytics
NT (D21923) + onCARlytics
Autologous CD19-CAR T Cells + onCARlytics
CYCART-19 (D21923) + onCARlytics
CYCART-19 (D23019) + onCARlytics
CYCART-19 (D20284) + onCARlytics
B
IFNγ (pg/mL)
Untransduced autologus T-Cell + onCARlytics
Autologous CD19-CAR T Cell + onCARlytics
NT (D21923) + onCARlytics
CYCART-19 (D21923) + onCARlytics
C
IL-2 (pg/mL)
D
E

Figure 5

CD19t expression in tumor cells following onCARlytics infection in vivo

Subcutaneously engrafted MDA-MB-468 tumors were collected 3, 7, or 10 days from NSG mice following onCARlytics infection at three indicated virus pfu per mouse and analyzed via flow cytometry for the expression of CD19t. MDA-MB-468 lentivirally transduced to stably express CD19t were used as a positive control (+ctrl).

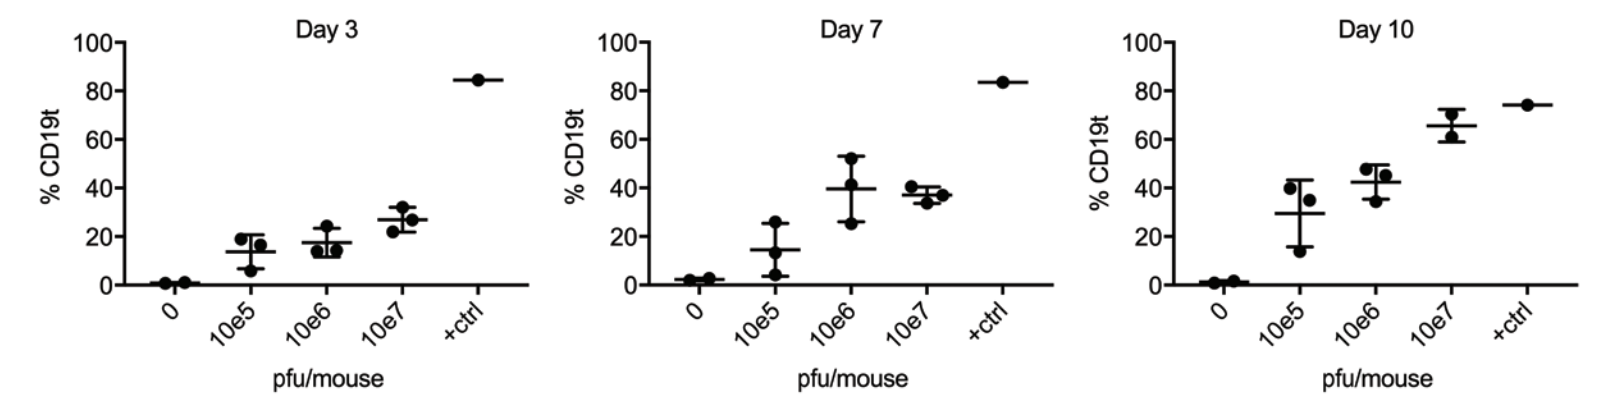

Day 3
Day 7
Day 10
% CD19t
0
10e5
10e6
10e7
+ctrl
pfu/mouse

Figure 6

Schema of in vivo studies testing onCARlytics in combination with CYCART-19

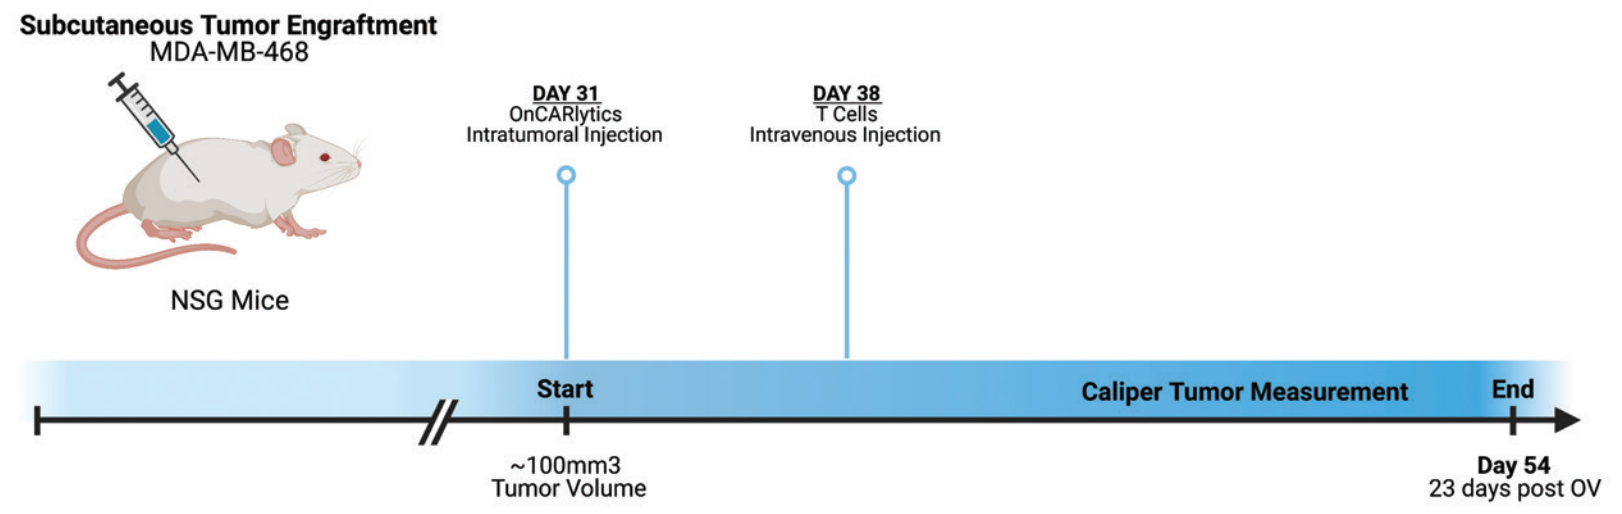

Subcutaneous Tumor Engraftment
MDA-MB-468
NSG Mice
DAY 31
OnCARlytics
Intratumoral Injection
DAY 38
T Cells
Intravenous Injection
Start
Caliper Tumor Measurement
End
~100mm3
Tumor Volume
Day 54
23 days post OV

Figure 7

Comparing anti-tumor activity of CYCART-19 against autologous CD19-CAR T Cell in MDA-MB-468-CD19t bearing NSG mice

Mice were engrafted with subcutaneous MDA-MB-468-CD19t ($5x10^6$ cells) and were intravenously treated with untransduced autologous, autologous CD19-CAR, NT, CYCART-19 (2 donors) T-Cells ($5x10^6$ cells). Tumors were measured to determine T-Cell efficacy against a positive control tumor cell line in vivo.

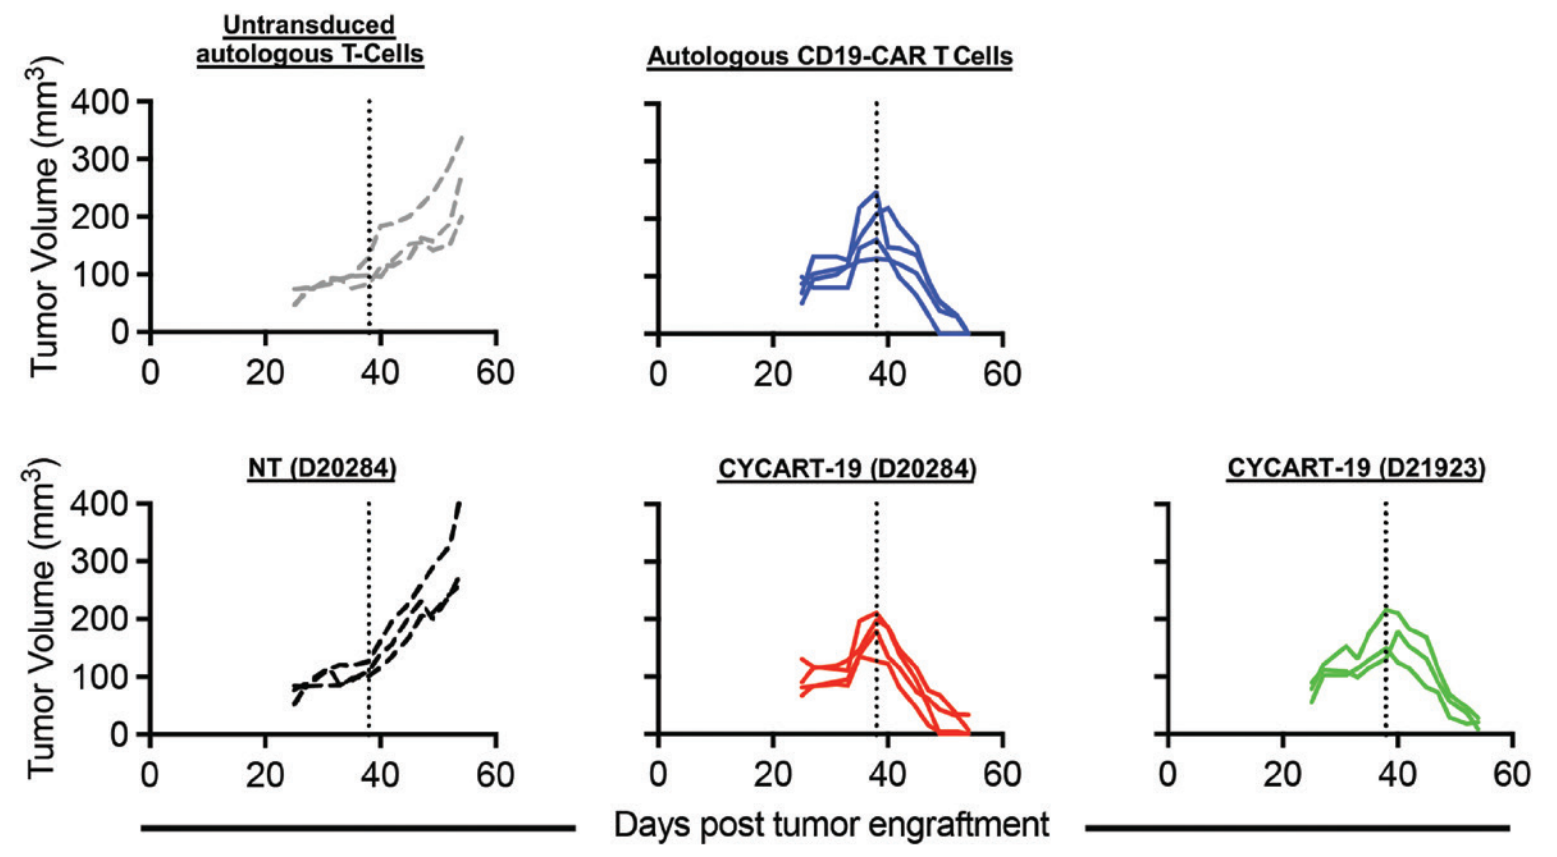

Untransduced autologous T-Cells
Autologous CD19-CAR T Cells
NT (D20284)
CYCART-19 (D20284)
CYCART-19 (D21923)
Tumor Volume (mm³)
Days post tumor engraftment

Figure 8

Anti-tumor activity of CYCART-19 in combination with onCARlytics in human xenograft triple negative breast cancer tumor model

Mice were engrafted with subcutaneous MDA-MB-468 ($5x10^6$ cells) and were intratumorally treated with 0 or 10^6 pfu of onCARlytics per mouse. Mice were intravenously treated with untransduced autologous, autologous CD19-CAR, NT, CYCART-19 (2 donors) T-Cells ($5x10^6$ cells). A Lines represent tumor volumes of individual mice per group (n=5-10) and B average of each group.

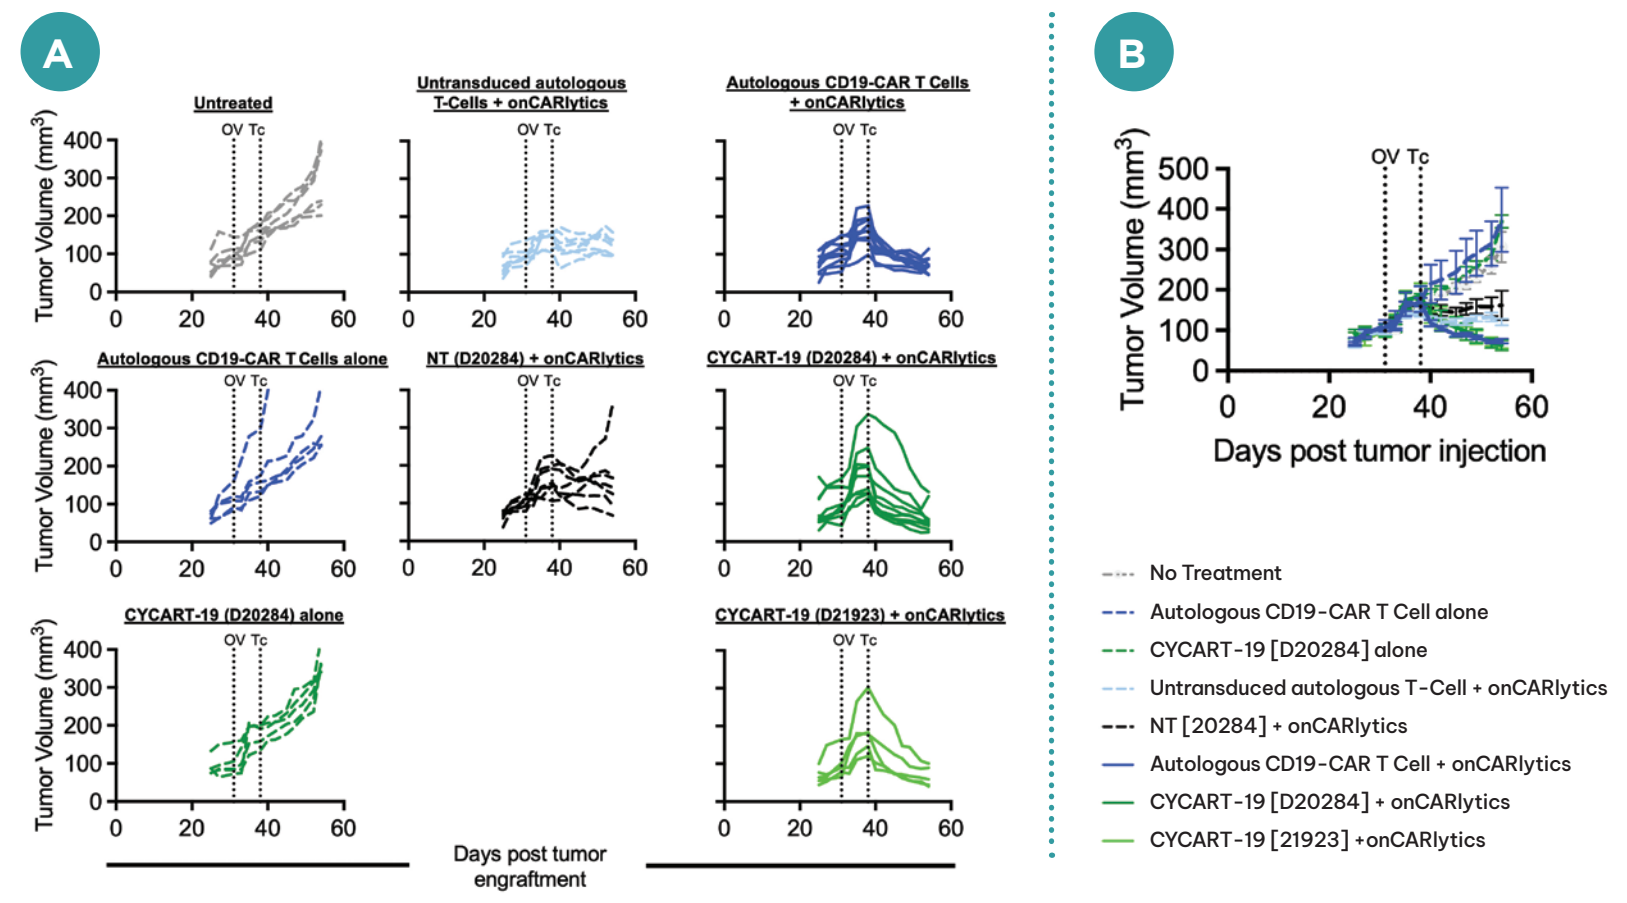

A
Untreated
Untransduced autologous T-Cells + onCARlytics
Autologous CD19-CAR T Cells + onCARlytics
Autologous CD19-CAR T Cells alone
NT (D20284) + onCARlytics
CYCART-19 (D20284) + onCARlytics
CYCART-19 (D20284) alone
CYCART-19 (D21923) + onCARlytics
Tumor Volume (mm³)
Days post tumor engraftment
B
Days post tumor injection
No Treatment
Autologous CD19-CAR T Cell alone
CYCART-19 [D20284] alone
Untransduced autologous T-Cell + onCARlytics
NT [20284] + onCARlytics
Autologous CD19-CAR T Cell + onCARlytics
CYCART-19 [D20284] + onCARlytics
CYCART-19 [21923] +onCARlytics

Summary

- onCARlytics can target triple negative breast cancer cell line MDA-MB-468 to express CD19t as a CAR T Cell target in an MOI-dependent manner.
- CYCART-19 demonstrated efficacy against MDA-MB-468 expressing CD19t following onCARlytics infection.
- There is an increasing trend in CYCART-19 activation and IL-2 production in an MOI-dependent manner.
- Allogeneic CYCART-19 T-Cell produced significantly less IFNγ compared to autologous CD19-CAR T Cell after CD19t expressing tumor killing.
- CD19t expression was detected in tumors following onCARlytics infection in vivo.
- CYCART-19 treatment 7 days post onCARlytics infection shows significant tumor regression compared to onCARlytics or T-Cells alone in a xenograft model of triple negative breast cancer.

References

1. Park AK, et al. Effective combination immunotherapy using oncolytic viruses to deliver CAR targets to solid tumors. Sci Transl Med. 2020. **2.** McCart JA, Ward JM, Lee J, Hu Y, Alexander HR, Libutti SK, Moss B, Bartlett DL. Systemic cancer therapy with a tumor-selective vaccinia virus mutant lacking thymidine kinase and vaccinia growth factor genes. Cancer Res. 2001. **3.** Chaurasiya S, et al. A chimeric poxvirus with J2R (thymidine kinase) deletion shows safety and anti-tumor activity in lung cancer models. Cancer Gene Ther. 2020. **4.** Gattinoni L, et al. Paths to stemness: building the ultimate antitumour T-Cell. Nature Reviews Cancer. 2012. **5.** Busch D, et al. Role of memory T-Cell subsets for adoptive immunotherapy. Seminars in Immunology. 2016. **6.** Sadeline M, et al. Therapeutic T-Cell engineering. Nature. 2017. **7.** Rafiq S, et al. Engineering strategies to overcome the current roadblocks in CAR T Cell therapy. Nature 2020.