MEDICAL UNIVERSITY OF VIENNA #

CT059

A Phase Ib open label multicenter study with a HER2/neu peptide vaccine administered with cisplatin and 5-fluorouracil or capecitabine chemotherapy shows safety, immunogenicity and clinical response in patients with HER2/neu overexpressing advanced cancer of the stomach

IMUGENE Developing Cancer Immunotherapies ##

Ursula Wiedermann[1], Erika Garner-Spitzer[1], Yee Chao[2], Iurie Bulat[3], Arunee Dechaphunkul[4], Wichit Arpornwirat[5], Chaiyut Charoentum[6], Chia-Jui Yen[7], Thomas Cheung Yau[8], Marina Maglakelidze[9], Suebpong Tanasanvimon[10], Jedzada Maneechavakajorn[11], Aumkhae Sookprasert,[12], Li-Yuan Bai[13], Wen-Chi Chou[14], Teerapat Ungtrakul[15], Mirjana Drinic[1], Joshua Tobias[1], Christoph C. Zielinski[16], Leslie Chong[17], Nick Ede[17], Anthony J Good[17]

[1]Medical University, Vienna, Austria; [2]Department of Oncology, Taipei Veterans General Hospital, Taipei, Taiwan; [3]ARENSIA Exploratory Medicine Research Unit, Institute of Oncology, Chisinau, Republic of Moldova; [4]University of Alberta Cross Cancer Institute, Edmonton, AB, Canada; [5]National Cancer Institute, Bangkok, Thailand; [6]Maharaj Nakorn Chiang Mai Hospital, Mueang Chiang Mai District, Thailand; [7]National Cheng Kung University, Tainan, Taiwan; [8]Department of Medicine, The University of Hong Kong, Queen Mary Hospital, Hong Kong, Hong Kong; [9]ARENSIA Exploratory Medicine LLC, Tbilisi, Georgia; [10]King Chulalongkorn Memorial Hospital, Bangkok, Thailand; [11]Rajavithi Hospital, Bangkok, Thailand; [12]Srinagarind Hospital, Khon Kaen, Thailand; [13]China Medical University Hospital, Taichung City, Taiwan; [14]Linkou Chang Gung Memorial Hospital, Taoyuan, Taiwan; [15]Faculty of Medicine and Public Health, HRH Princess Chulabhorn College of Medical Science, Bangkok, Thailand; [16]Central European Cooperative Oncology Group, Vienna, Austria; [17]Imugene, Sydney, Australia

Background:
HER2/neu is overexpressed in 15-25% of gastric cancer patients and associated with poor prognosis. Recombinant monoclonal antibodies (mAbs) against HER2/neu have been shown to be effective in clinical application but alternative treatments are needed due to cost and global availability issues of mAbs.

Thus a **B-cell peptide vaccine (IMU-131)** was developed at the Medical University of Vienna # (1), consisting of three fused B-cell epitopes (p467) from the HER2/neu extracellular domain coupled to CRM197 and administered with the adjuvant Montanide.

The goal of the study was to evaluate the optimal and safe vaccine dose which leads to immunogenicity and clinical responses.

Material & Methods:
An **open label multicenter Phase 1b trial** was performed in SE-Asia (Taiwan, Thailand) and Eastern Europe (Georgia, Moldova).

Fourteen patients with HER2/neu overexpressing gastric or gastro-esophageal junction adenocarcinoma were enrolled.

Dose escalation with 10, 30 and 50 µg of IMU-131 was performed in 3 cohorts, each including 3 to 5 patients.

Immune responsiveness to **IMU-131** was evaluated according to antibody levels against p467 peptide and HER2 and by cellular responses.

HER2- and p467-specific IgG levels were determined by ELISA, cytokine levels were measured by Luminex Technology in PBMC cultures after 48h of HER2-stimulation and lymphocyte subsets were analyzed by FACS.

Timeline of interventions:

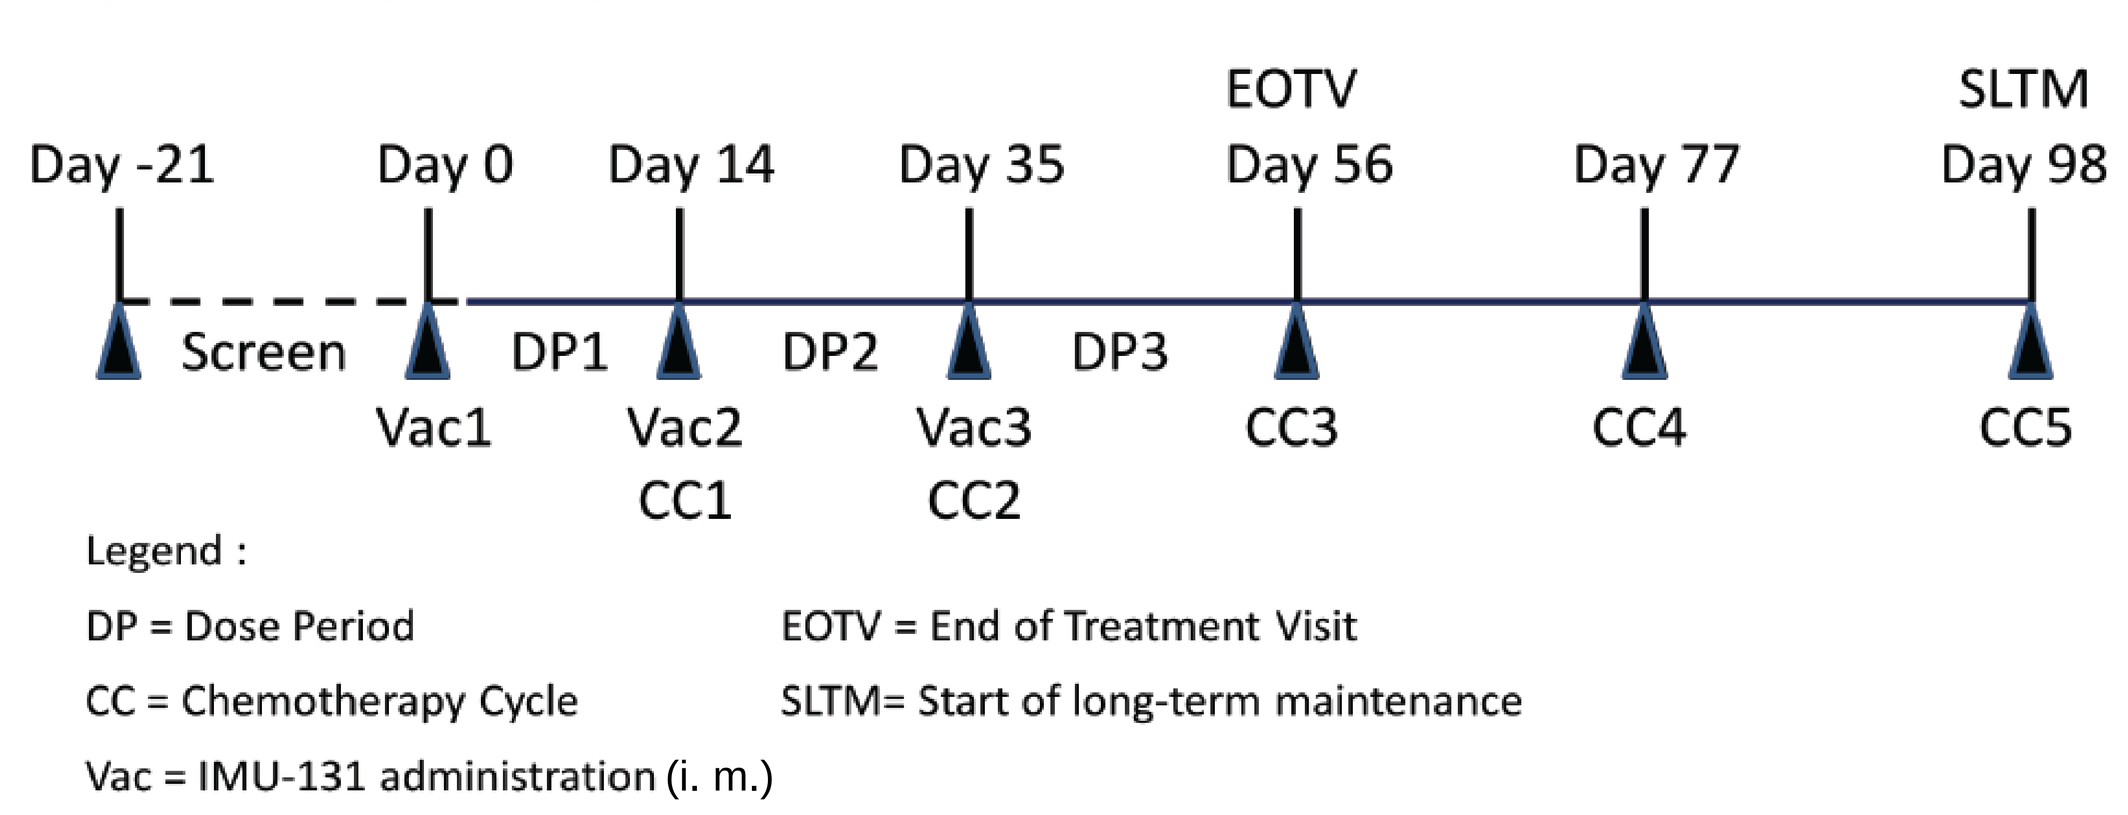

EOTV
SLTM
Day -21
Day 0
Day 14
Day 35
Day 56
Day 77
Day 98
Screen
DP1
DP2
DP3
Vac1
Vac2
CC1
Vac3
CC2
CC3
CC4
CC5
Legend :
DP = Dose Period
EOTV = End of Treatment Visit
CC = Chemotherapy Cycle
SLTM= Start of long-term maintenance
Vac = IMU-131 administration (i. m.)

Results:
Ph1b study data indicate IMU-131 is safe and tolerated with no significant local or systemic reactions, and no need for pre-treatment or for modification to the dose or treatment schedule due to safety. No SAEs related to administration of IMU-131 were reported.

Eleven of fourteen patients were evaluable for vaccine-specific immune responses and tumor response assessment (Table 1, Fig. 1).

Higher HER2-specific IgG levels were observed in cohort 2 (30 µg/dose) compared to cohort 1 (10 µg/dose). Three of five patients in cohort 2 displayed moderate or little increase in antibody titers. In contrast, all patients in cohort 3 (50µg/dose) showed a moderate or high increase of HER2-specific Ab levels upon vaccination (Fig. 2).

Response rate was an exploratory endpoint of this clinical trial and of 11 evaluable patients one showed complete response, 5 partial response and 4 stable disease (RECIST response, Tables 1 & 2).

In cohort 3, high antibody levels correlated with clinical responses /changes in tumor size (Fig. 1, 2 & 3). The Abs present in serum of C3 patient TW02_004 at d56, which were correlated with strong tumor reduction, showed capacity to inhibit HER2 phosphorylation (Fig. 4). Cohort 2 showed only moderate correlation between humoral and clinical responses. However, the C2 patients with marked tumor reduction despite weak Ab increases showed high IFNg/IL-10 or TNFα/IL-10 ratios (Fig. 5). Also the complete response present in Cohort 1 did not correlate with Ab levels, but rather with an increased TNFα/IL-10 ratio (Fig. 5).

Moreover, in patients with marked decrease in tumor size, the percentage of T-regulatory cells declined (Fig. 6).

	Patient	gender	age	Dose (µg)	HER2 Status	SOD d 0 (mm)	SOD d 56 (mm)	SOD d 98 (mm)	SOD d 182 (mm)	SAE (not IMU-131 related)
C1	TH02 001	m	41	10	2+/Fish-	97	80 (SD)	114 (PD)	Withdrawn	none
C1	TH03 001	f	33	10	2+/Fish-	268	320 (SD)	330 (PD)	Deceased	Convulsion
C1	TW02 002	f	79	10	3+	38	9 (CR)	10 (PR)	14 (PD)	Neutropenia
C2	TH05 013	f	54	30	2+/Fish+	49	39 (SD)	39 (SD)	Deceased	Hyponatraemia
C2	TH05 015	m	55	30	3+	78	66 (SD)	36 (PR)	35 (PR)	none
C2	GE01 004	m	58	30	3+	122	103 (SD)	75 (PR)	Deceased	Pneumonia
C2	TW01 001	f	80	30	3+	56	40 (PD)*	Withdrawn	Withdrawn	Kidney failure
C2	MD01 003	m	64	30	3+	NTL**	NTL (SD)	NTL (PD)	Withdrawn	none
C3	TW02 003	m	75	50	2+/Fish+	66	36 (PR)	39 (PR)	TBD	Gastritis
C3	MD01 007	m	66	50	2+/Fish -	26	28 (SD)	18 (PR)	TBD	none
C3	TW02 004	f	21	50	3+	177	105 (PR)	99 (PR)	TBD	Leucocytosis

Table 1: Patient characteristics; SOD sum of diameters

* due to 2 new lesions; ** NTL non-target lesions

Best Overall Response	IMU-131 10 µg (N=3)	IMU-131 30 µg (N=6)	IMU-131 50 µg (N=5)
Complete Response (CR)	1 (33.3%)	0 (0.0%)	0 (0.0%)
Partial Response (PR)	0 (0.0%)	2 (33.3%)	3 (60.0%)
Stable Disease (SD)	2 (66.7%)	2 (33.3%)	0 (0.0%)
Progressive Disease (PD)	0 (0.0%)	1 (16.7%)	0 (0.0%)
Not Evaluable (NE)	0 (0.0%)	0 (0.0%)	1 (20.0%)
Not Applicable (NA)	0 (0.0%)	1 (16.7%)	1 (20.0%)
Objective Response (CR+PR)	1 (33.3%)	2 (33.3%)	3 (60.0%) *
Disease Control Rate (CR+PR+SD)	3 (100.0%)	4 (66.7%)	3 (60.0%)

Table 2: Best overall response

* In cohort 3, the 3 patients evaluated at Day 98 after completed IMU-131 vaccinations showed 100% objective response (CR+PR), while 2 patients dropped out early due to incorrect enrolment or SAE (non-vaccine related).

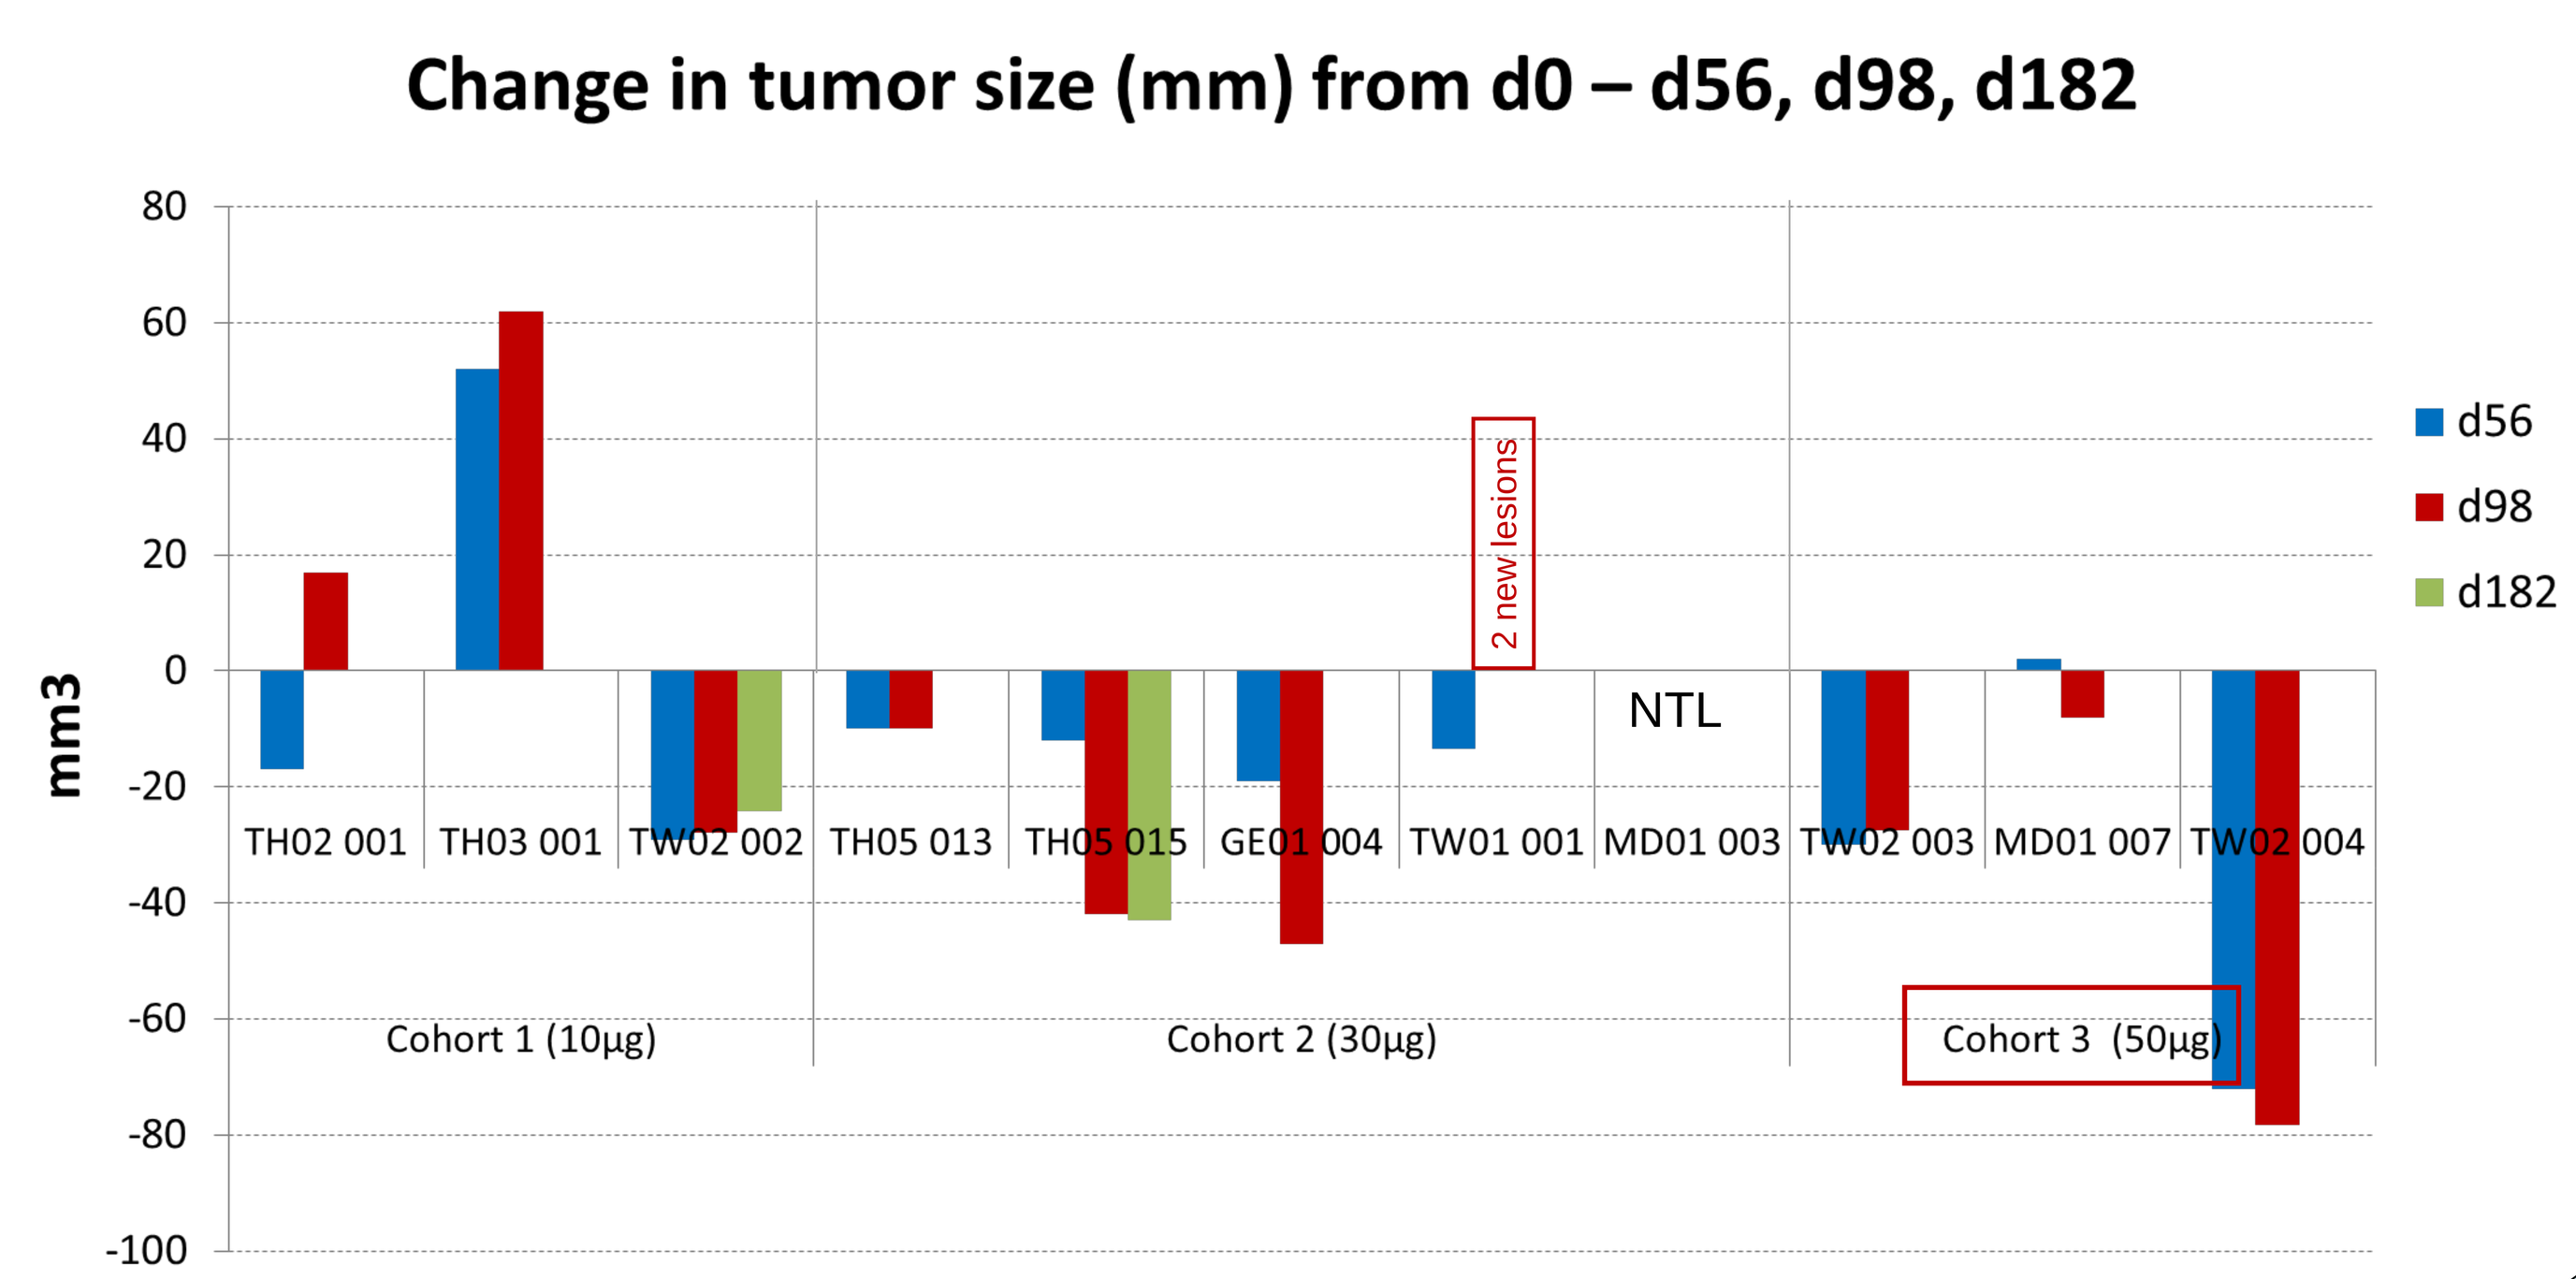

Change in tumor size (mm) from d0 – d56, d98, d182
mm3
d56
d98
d182
2 new lesions
NTL
TH02 001
TH03 001
TW02 002
TH05 013
TH05 015
GE01 004
TW01 001
MD01 003
TW02 003
MD01 007
TW02 004
Cohort 1 (10µg)
Cohort 2 (30µg)
Cohort 3 (50µg)

Fig. 1: Change in tumor size (in mm) from Day 0 to Day 56, 98 and 182.

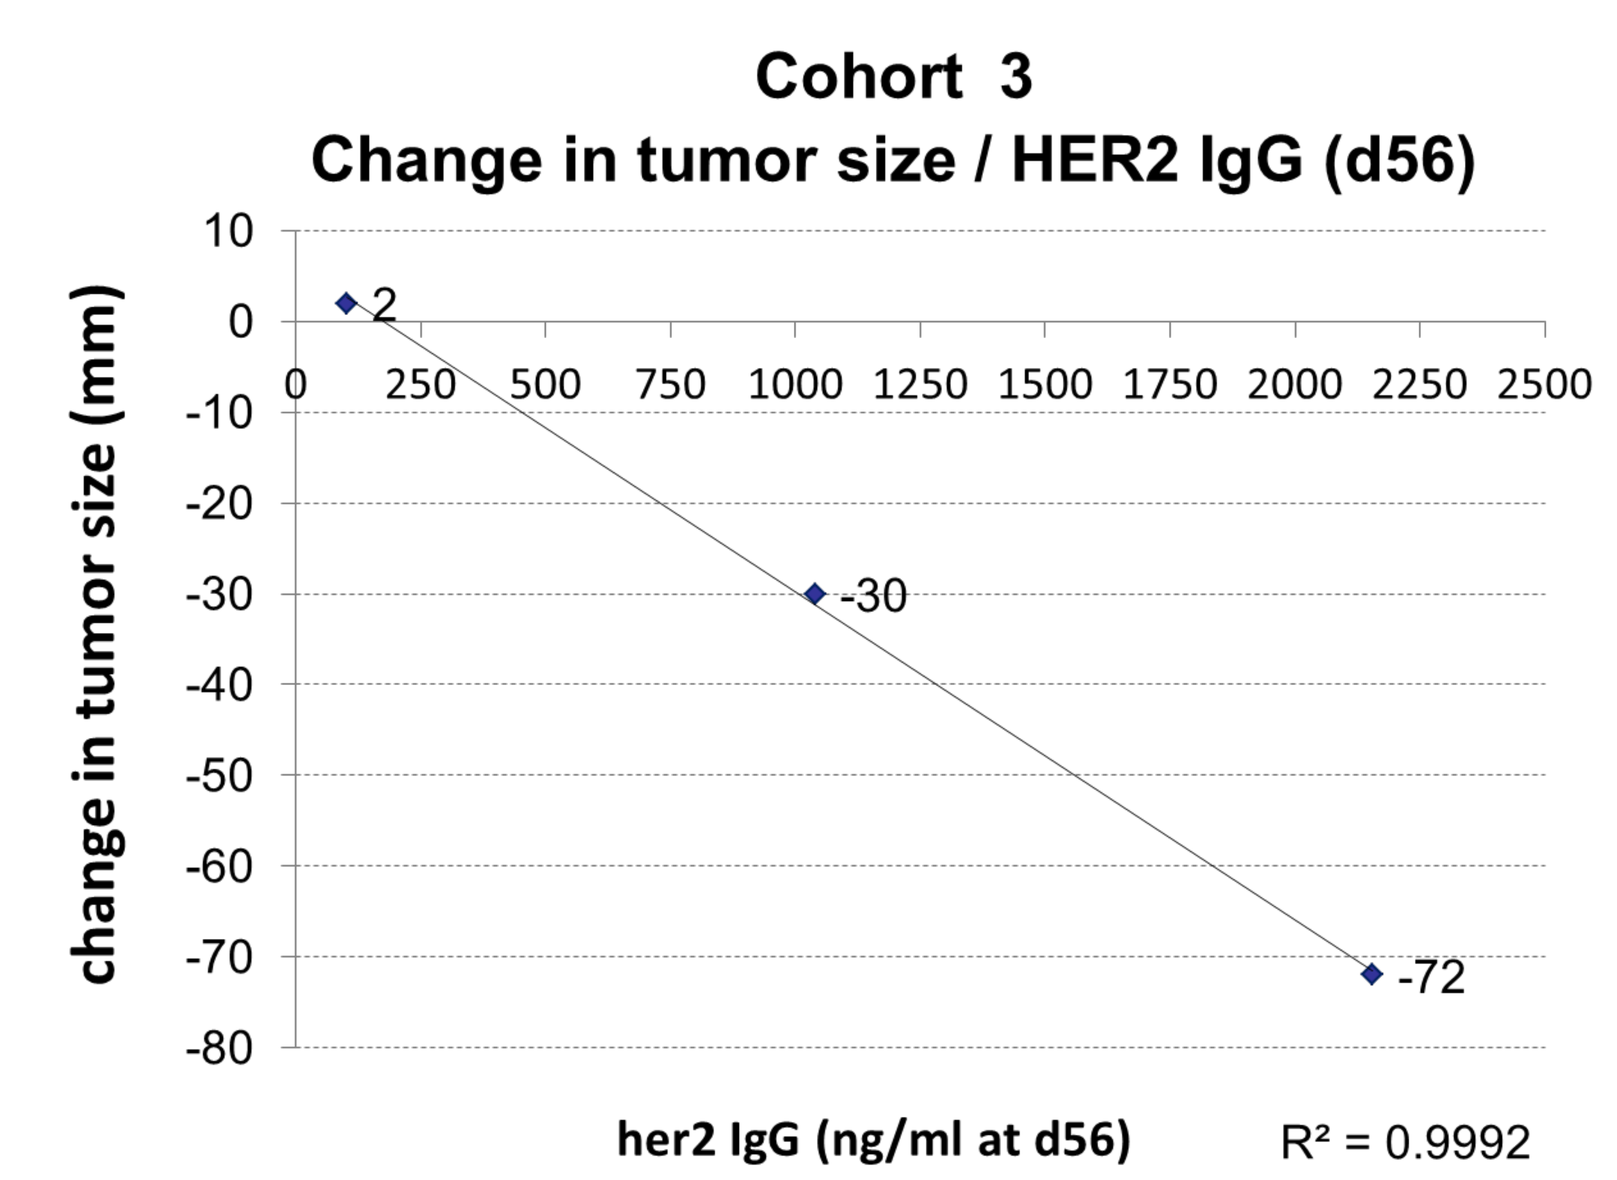

Cohort 3
Change in tumor size / HER2 IgG (d56)
change in tumor size (mm)
her2 IgG (ng/ml at d56)
R² = 0.9992
2
-30
-72

Fig. 3: Correlation of change in tumor size from Day 0 to Day 56 and HER2-specific IgG levels at Day 56 in Cohort 3.

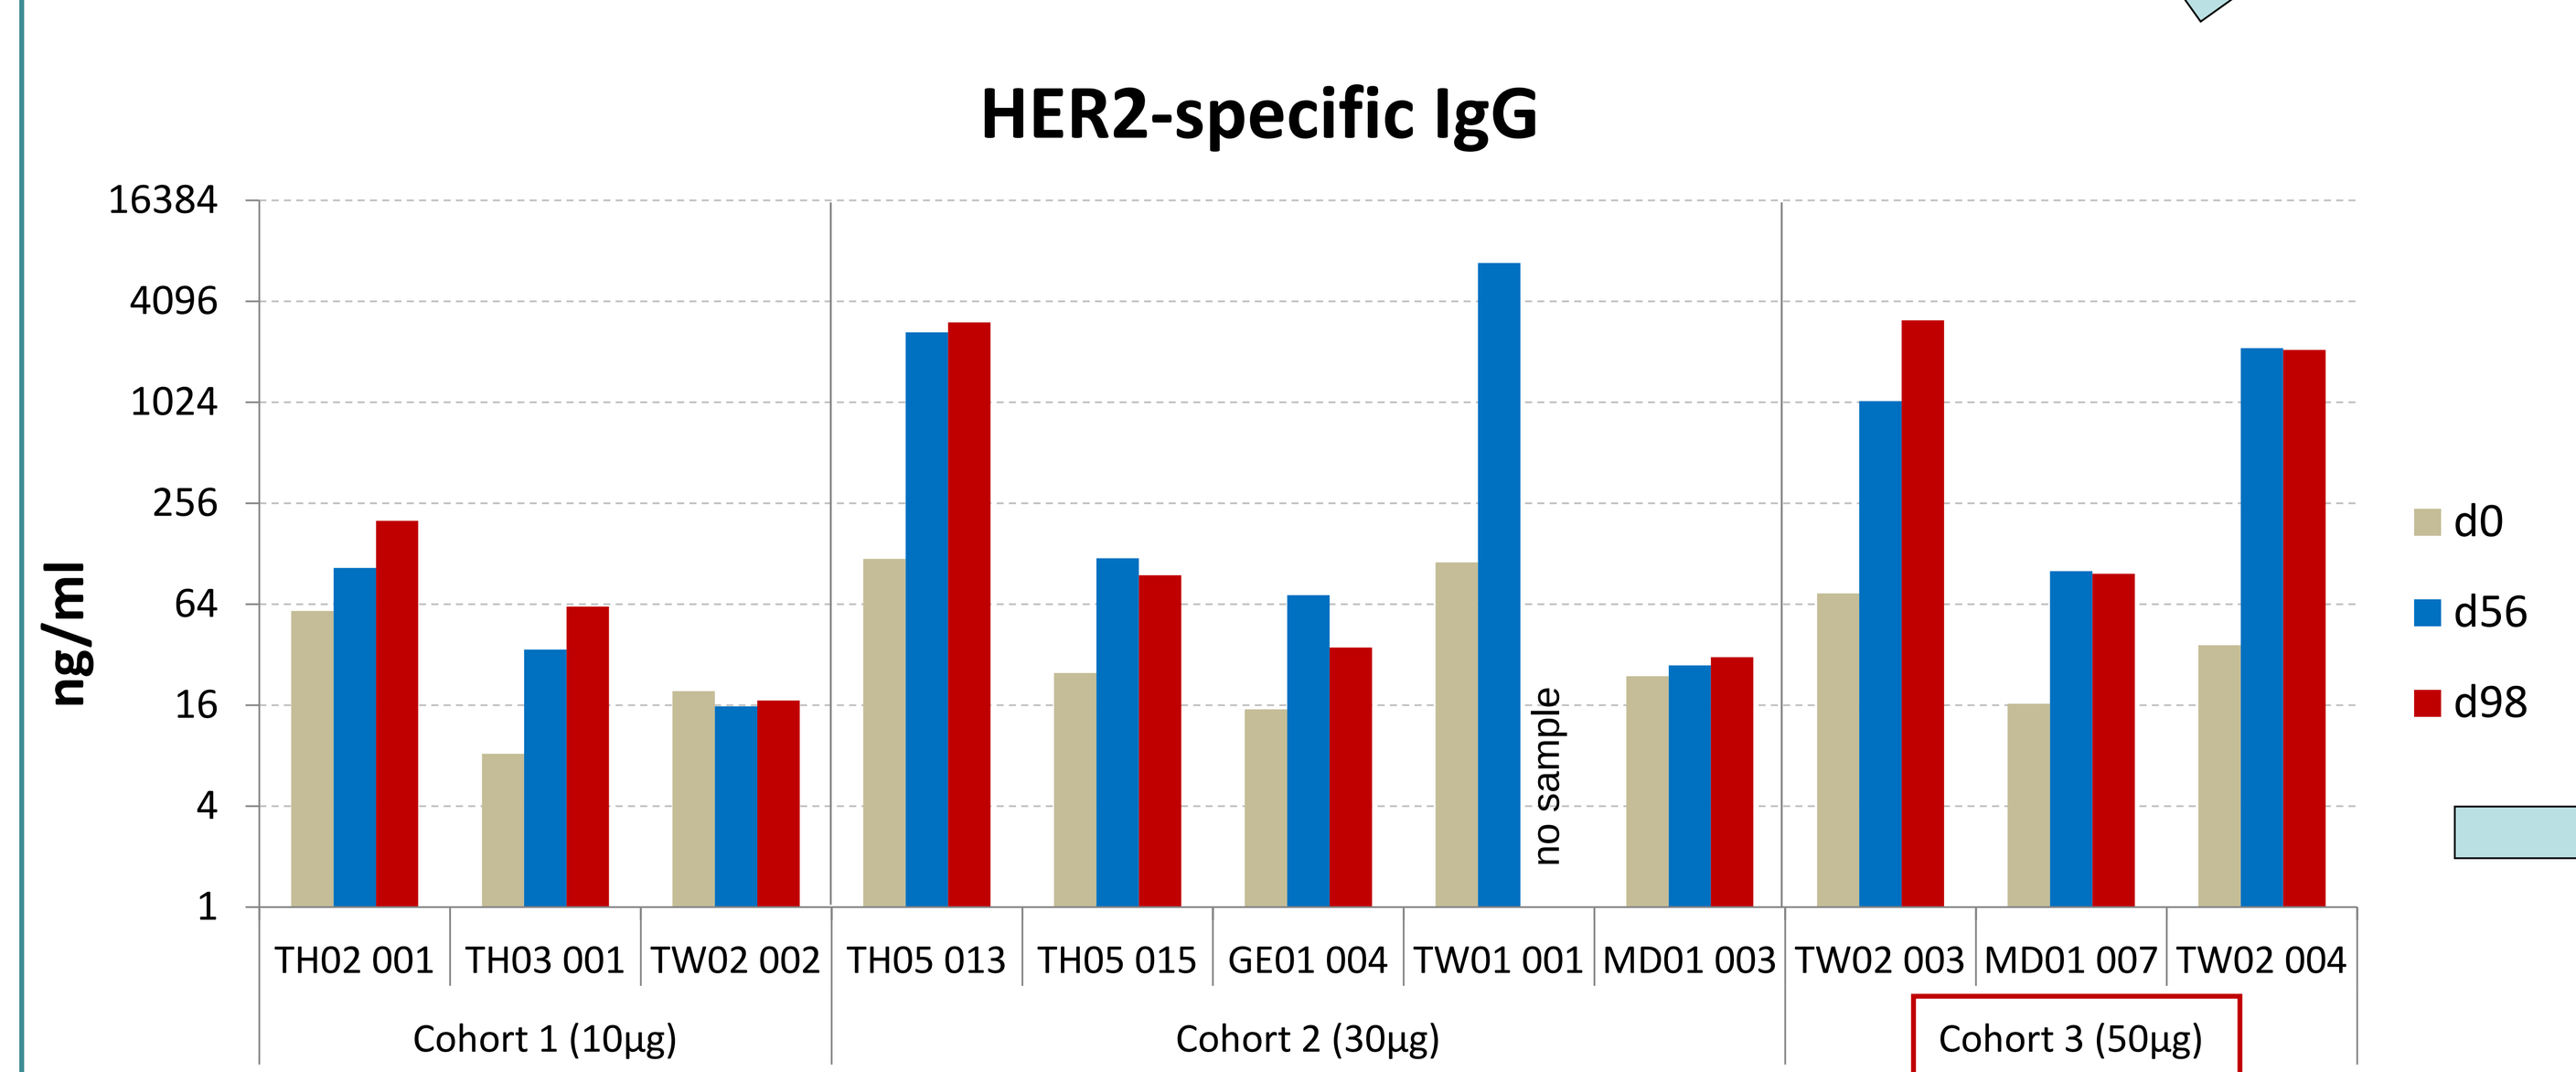

HER2-specific IgG
ng/ml
d0
d56
d98
no sample
TH02 001
TH03 001
TW02 002
TH05 013
TH05 015
GE01 004
TW01 001
MD01 003
TW02 003
MD01 007
TW02 004
Cohort 1 (10µg)
Cohort 2 (30µg)
Cohort 3 (50µg)

Fig. 2: HER2-specific IgG Abs in Cohort 1, 2, and 3 measured in sera obtained at Day 0, 56 and 98. Concentrations in ng/ml calculated from a Herceptin Standard Curve Dilution.

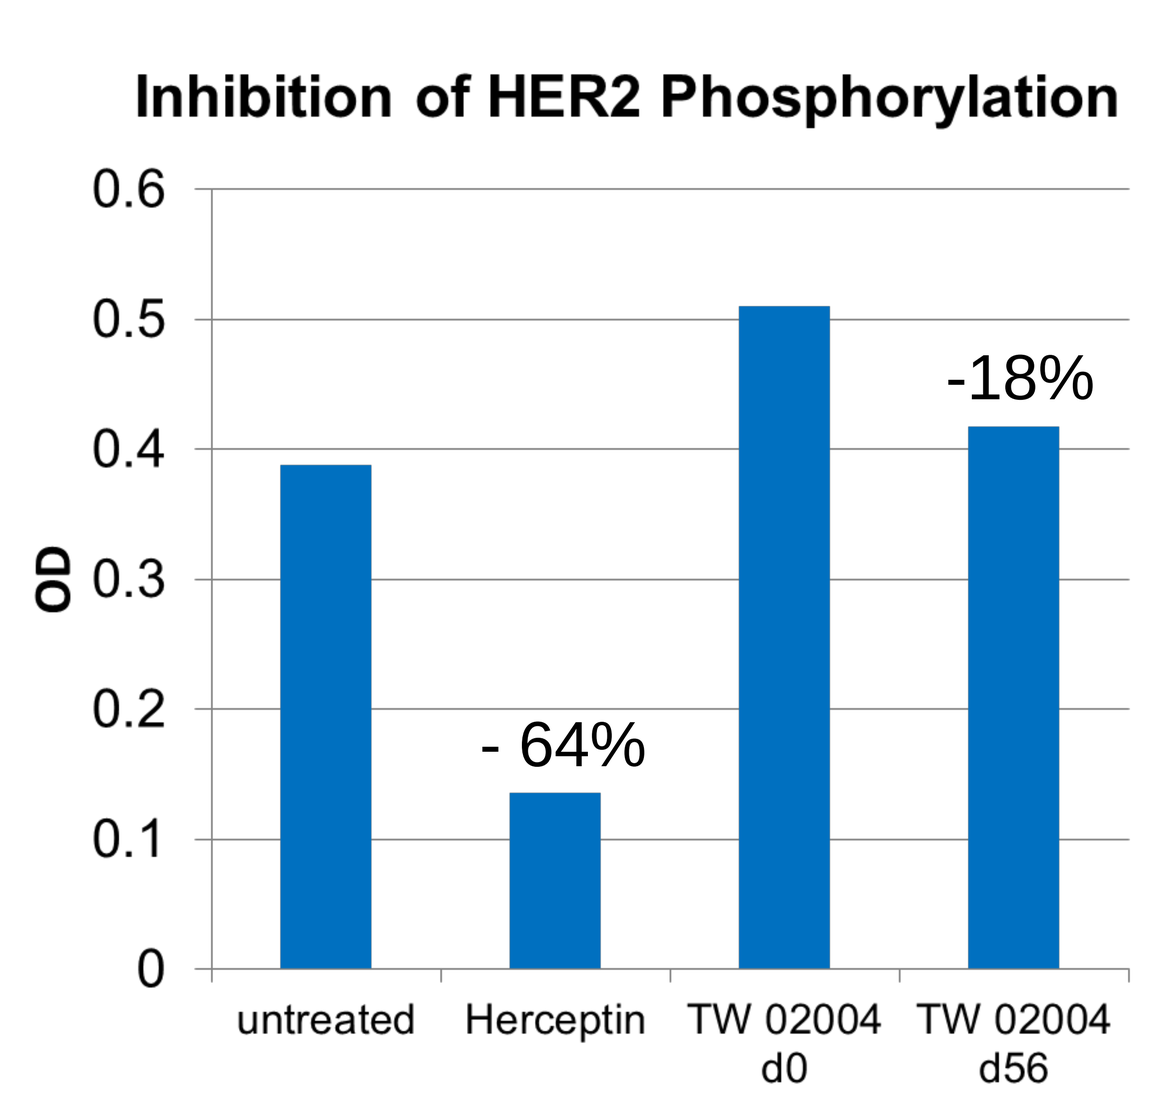

Inhibition of HER2 Phosphorylation
OD
- 64%
-18%
untreated
Herceptin
TW 02004 d0
TW 02004 d56

Fig. 4:: HER2-expressing gastric cancer cells (N87) were incubated with patient serum, Herceptin or left untreated and the respective cell lysates were tested with capture ELISA for phosphorylated HER2.

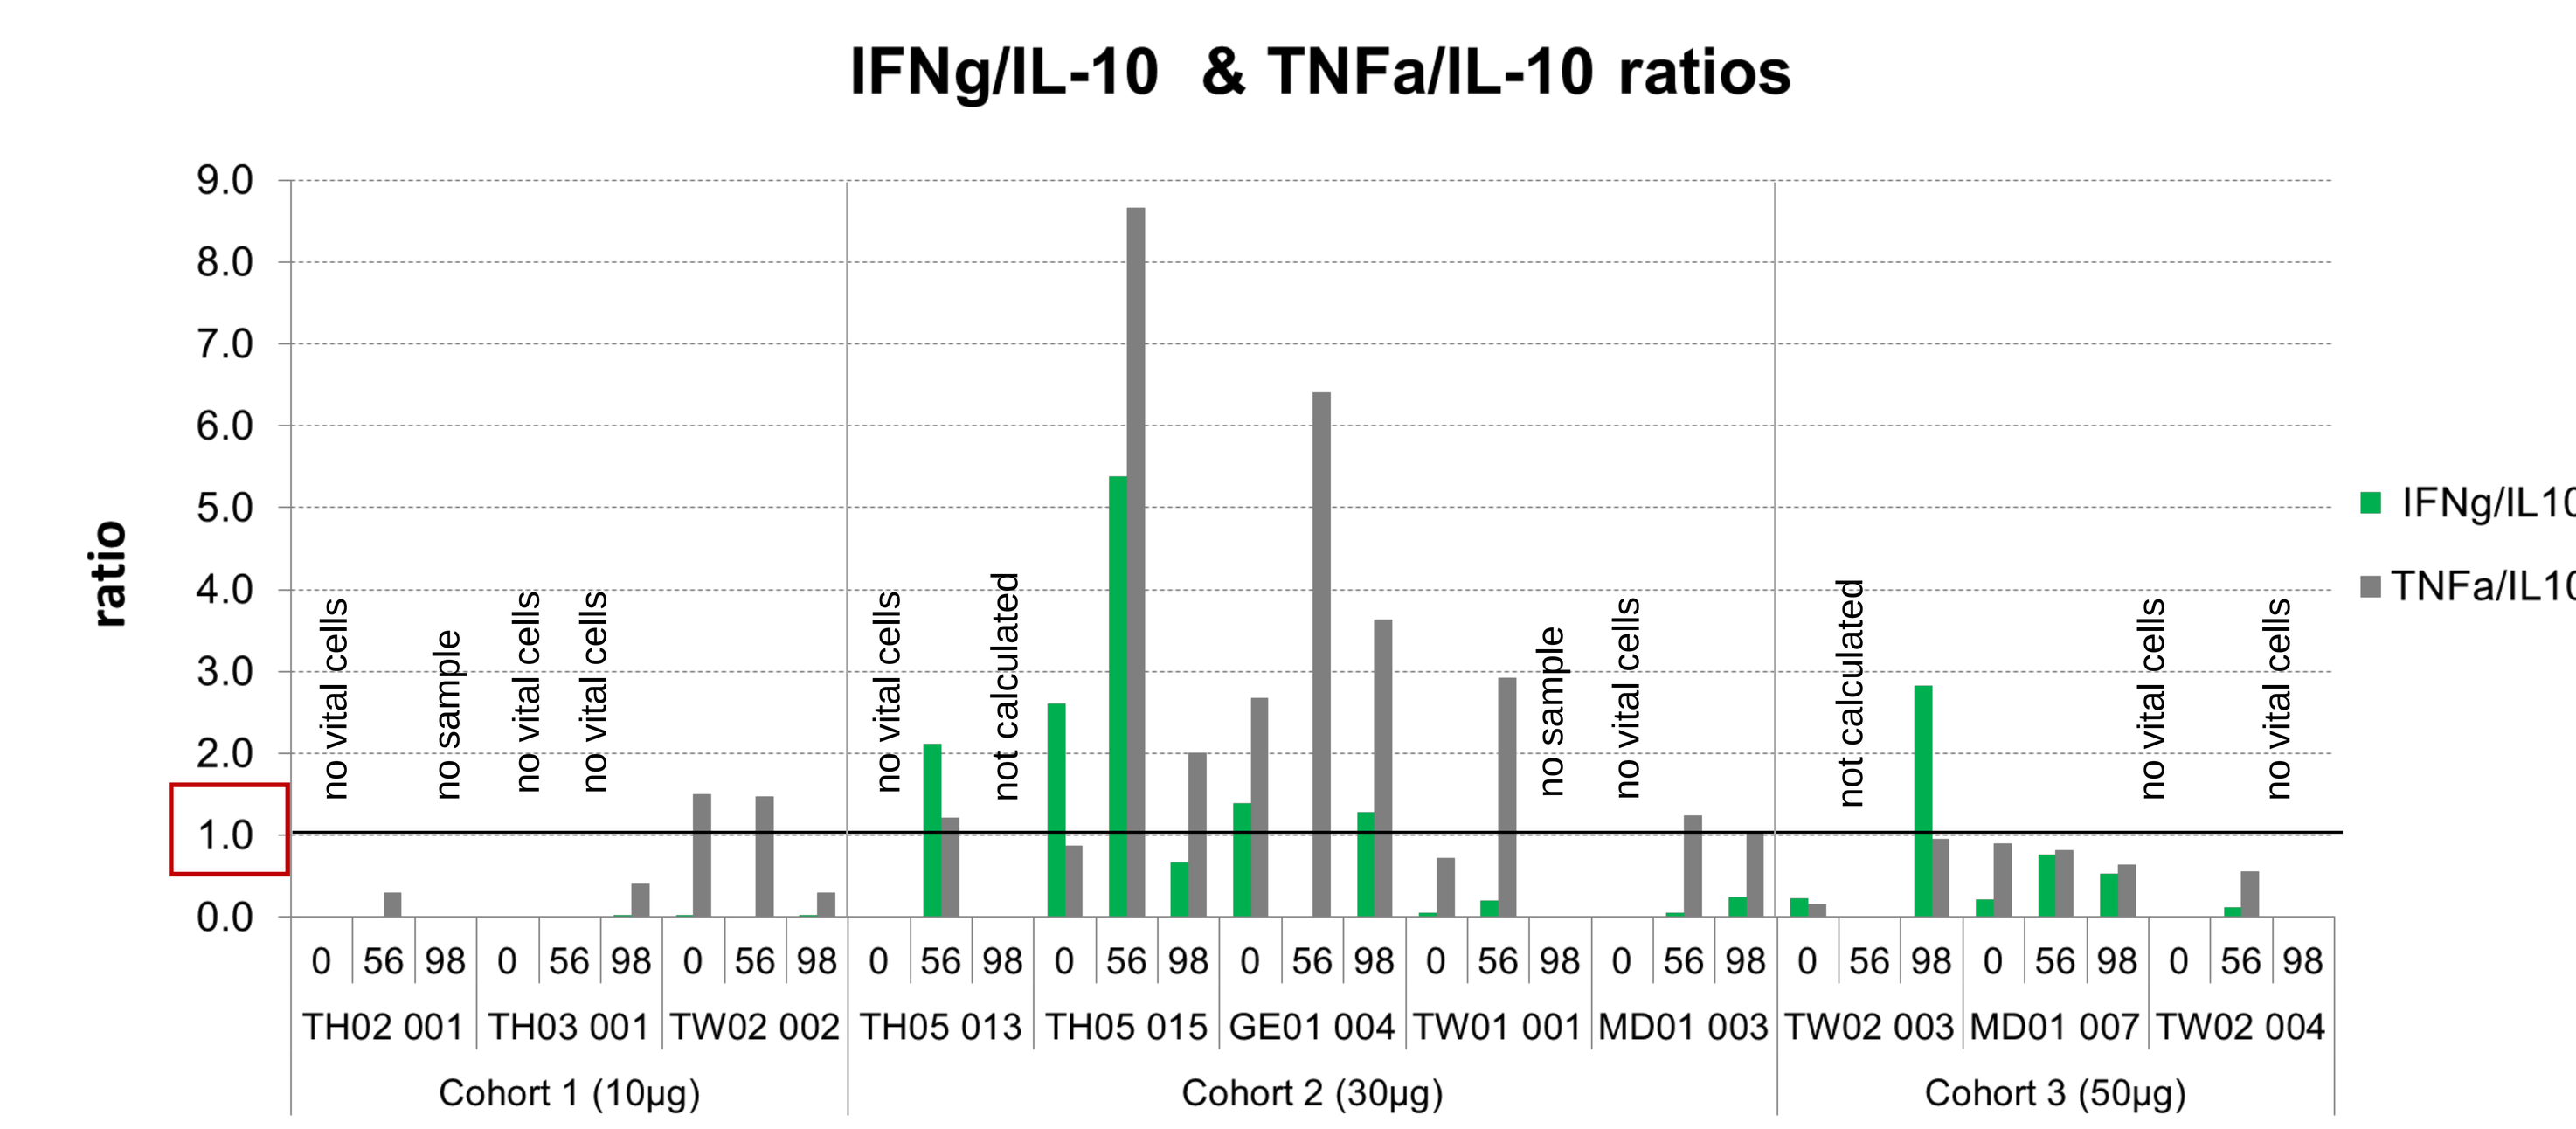

IFNg/IL-10 & TNFa/IL-10 ratios
ratio
IFNg/IL10
TNFa/IL10
no vital cells
no sample
not calculated
TH02 001
TH03 001
TW02 002
TH05 013
TH05 015
GE01 004
TW01 001
MD01 003
TW02 003
MD01 007
TW02 004
Cohort 1 (10µg)
Cohort 2 (30µg)
Cohort 3 (50µg)

Fig. 5: Concentrations of cytokines IFNg, TNFa and IL-10 were measured in supernatants of HER2 re-stimulated cultures of PBMC, which were obtained at Day 0, Day 56 and Day 98. Ratios of IFNg/IL-10 and TNFa/IL-10 were calculated.

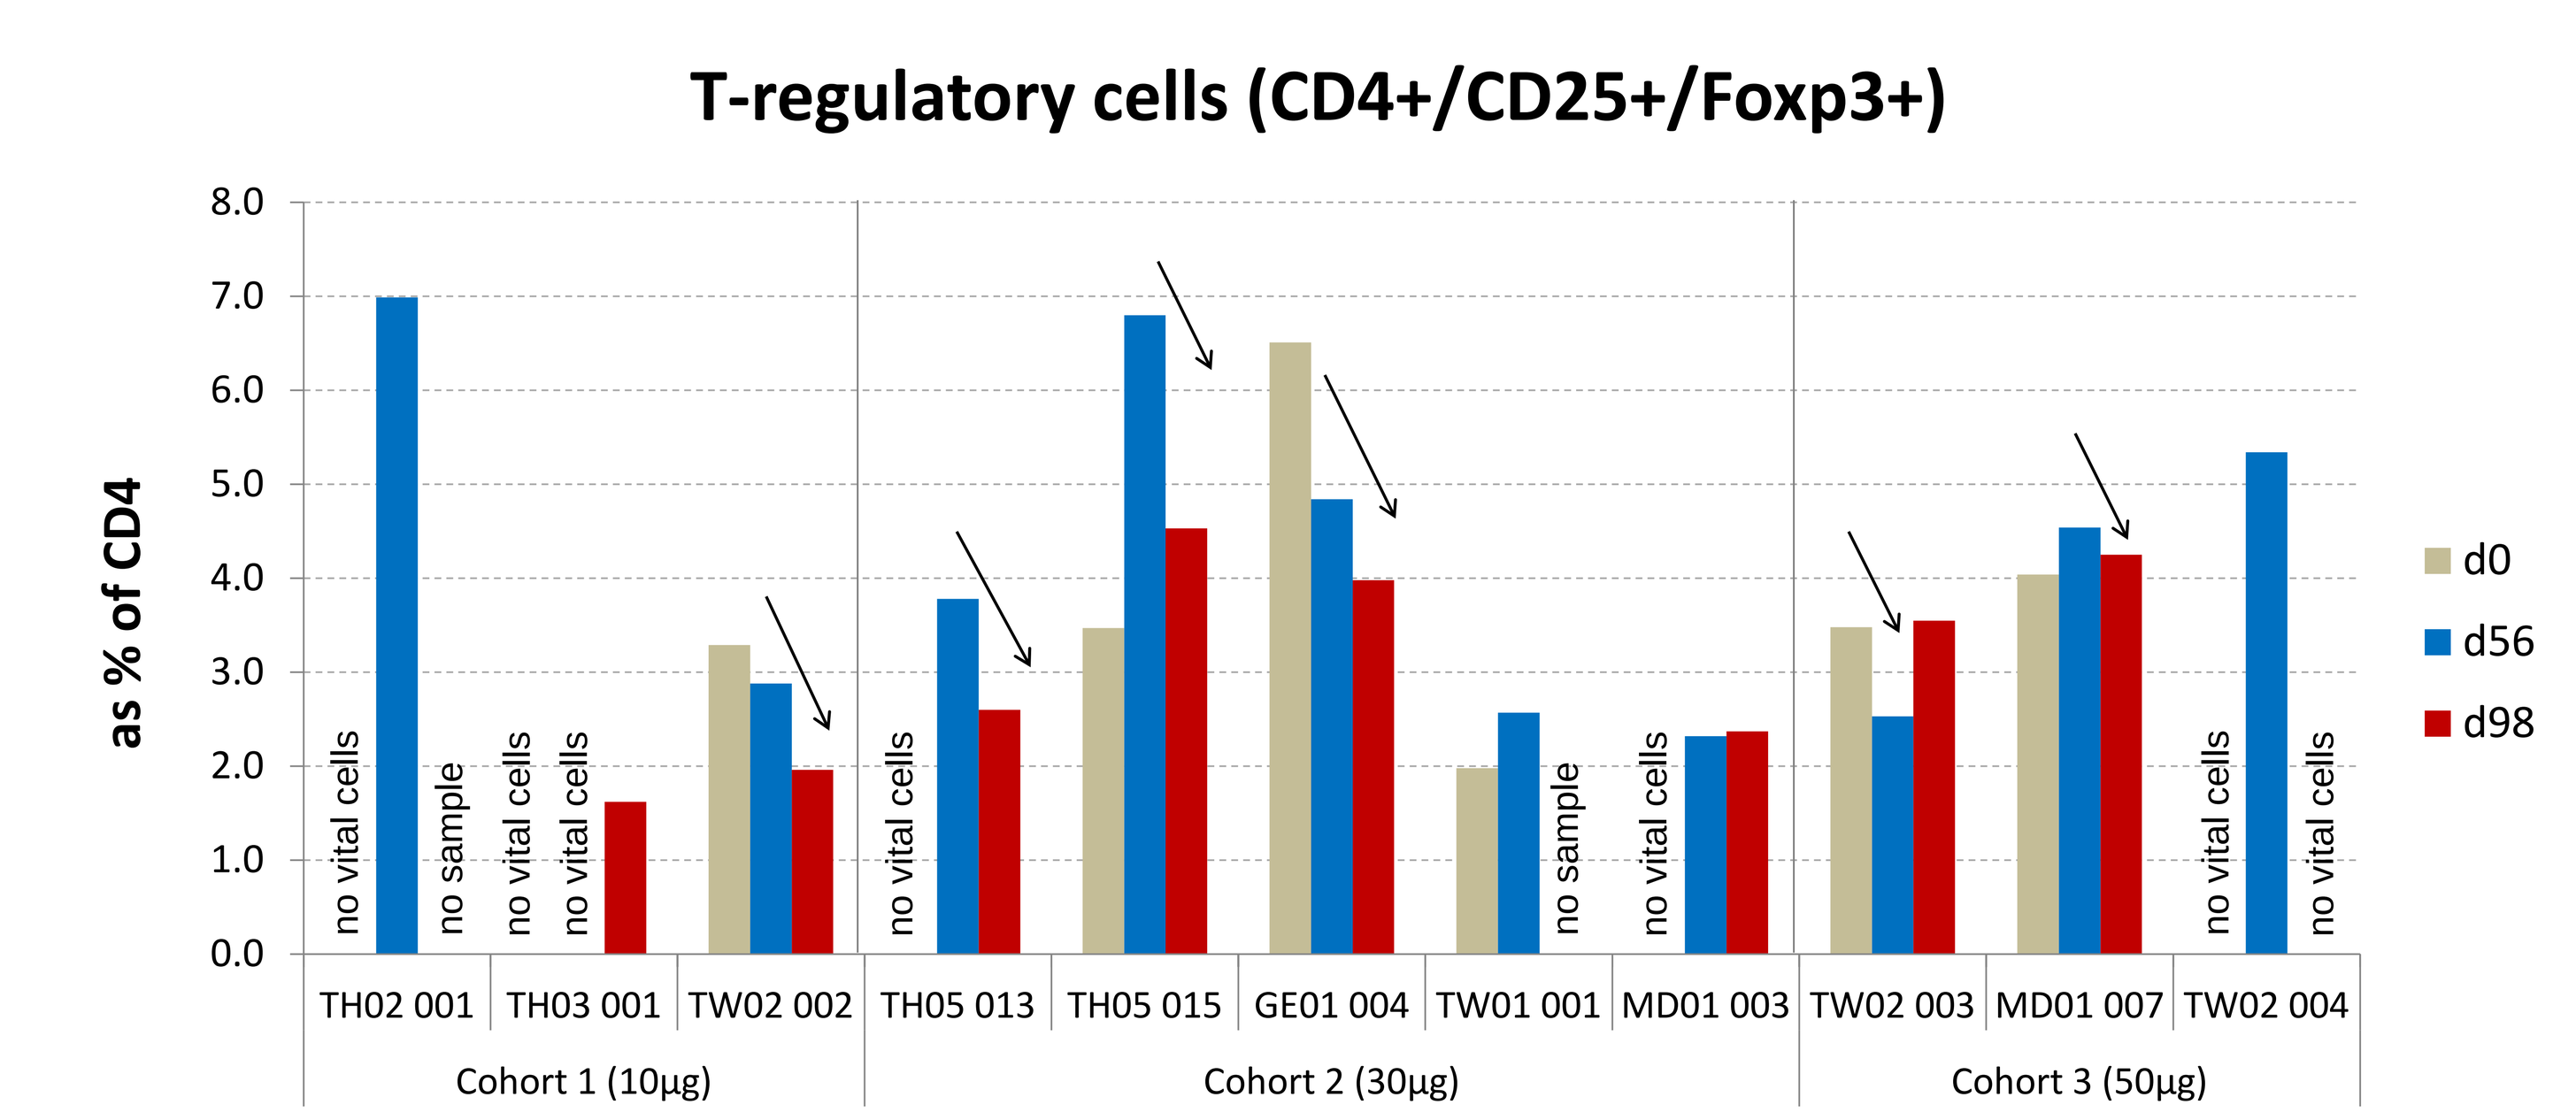

T-regulatory cells (CD4+/CD25+/Foxp3+)
as % of CD4
d0
d56
d98
no vital cells
no sample
TH02 001
TH03 001
TW02 002
TH05 013
TH05 015
GE01 004
TW01 001
MD01 003
TW02 003
MD01 007
TW02 004
Cohort 1 (10µg)
Cohort 2 (30µg)
Cohort 3 (50µg)

Fig. 6: T-regulatory cells (CD4+/CD25+/Foxp3+) were quantified as % of CD4+ T-helper cells. PBMC from Day 0, Day 56 and Day 98 were stained with the respective fluorochrome-labelled Abs and measured by Flow Cytometry.

Conclusions:
The vaccine was well tolerated and safe, with antibody responses at the highest dose (50 µg) showing a strong correlation with clinical responses. Thus, a dose of 50 µg was recommended for further evaluation in a Phase II trial, featuring two arms of either IMU-131 plus chemotherapy (CT) or CT alone, which has been initiated (first patient in).

Proposed mechanisms of action:
Direct effect of Abs on phosphorylation status of HER2, HER2-specific Th1-biased cytokine profile and decrease of T-regulatory cells.

Correspondence: ursula.wiedermann@meduniwien.ac.at
erika.garner-spitzer@meduniwien.ac.at

The study was financed by Imugene Limited, Australia ##

(1) Tobias J, Jasinska J, Baier K, Kundi M, Ede N, Zielinski C, Wiedermann U. Enhanced and long term immunogenicity of a Her-2/neu multi-epitope vaccine conjugated to the carrier CRM19 7in conjunction with the adjuvant Montanide. BMC Cancer. 2017 Feb 9;17(1):118.