Permanent Abstract Number: 4110

PD1 B cell mimotopes with functional PD1-PDL1 blocking capacity and anti-tumor activity: New strategy for a multi-level cancer immunotherapy

Joshua Tobias[1*], C. Battin[1], A. De Sousa Linhares[1], M. Lebens[2], K. Baier[1], K. Ambroz[1], M. Drinic[1], S. Högler[3], A. Inic-Kanada[1], E. Garner-Spitzer[1], M. Preusser[1,4], L. Kenner[3], M. Kundi[1], C. C. Zielinski[1,4], P. Steinberger[1], U. Wiedermann[1*]

Introduction and Aim

The use of immune checkpoint inhibitors (ICI) has become a major focus in cancer immunotherapy, and the application of monoclonal antibodies (mAbs) as ICIs shows impressive results in the therapy against cancers like melanoma, renal cell carcinoma or non-small cell lung cancer. Active immunization with B cell epitopes/mimotopes of ICIs rather than application of the corresponding mAbs for passive immunotherapy may, however, provide advantages such as induction of antibodies by the patient's own immune system and overcoming the costly treatment of the mAbs. By establishing a platform, mimotopes (B cell epitopes) of the anti-human PD1 mAb Nivolumab (as an example) and of anti-mouse PD1 mAb (as a proof of principle for evaluating anti-tumor activity *in vivo*) were identified and characterized.

Aim: To generate ICIs mimotopes with strong capacity in inducing antibody response with anti-tumor activity.

Methods

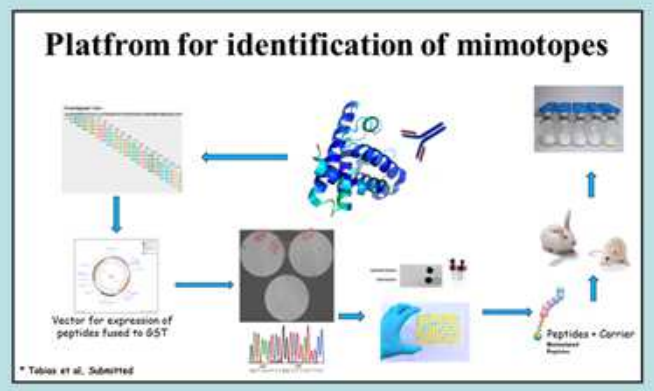
Platfrom for identification of mimotopes
Vector for expression of peptides fused to GST
Peptides + Carrier
* Tobias et al, Submitted

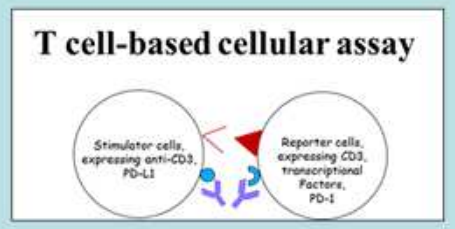
T cell-based cellular assay
Stimulator cells, expressing anti-CD3, PD-L1
Reporter cells, expressing CD3, transcriptional Factors, PD-1

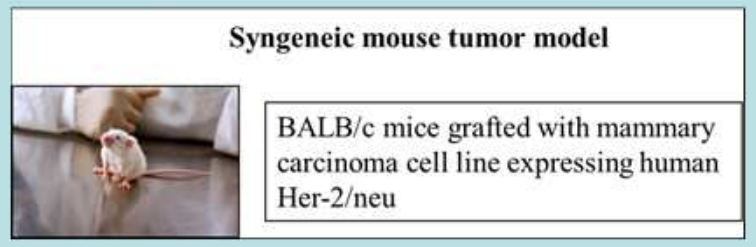
Syngeneic mouse tumor model
BALB/c mice grafted with mammary carcinoma cell line expressing human Her-2/neu

Results

Mimotopes of Nivolumab

Specifically and dose-dependently inhibit binding of Nivolumab to T cells expressing human PD1

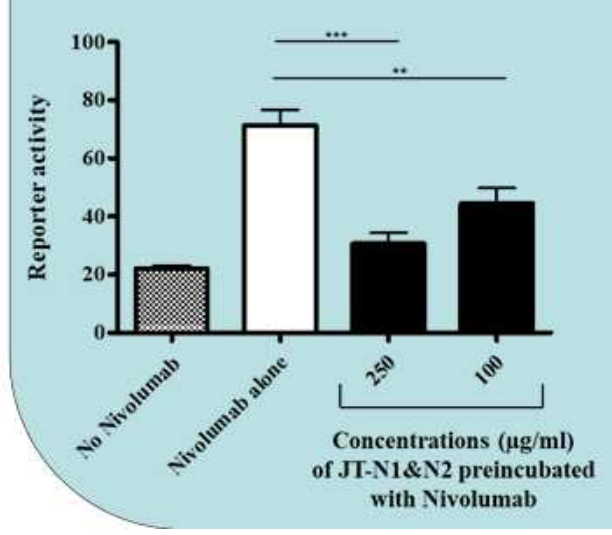
Reporter activity
100
80
60
40
20
0

**
No Nivolumab
Nivolumab alone
250
100
Concentrations (µg/ml) of JT-N1&N2 preincubated with Nivolumab

Induce IgGs with a capacity in blocking the human PD1/PDL1 interaction (*in vitro*)

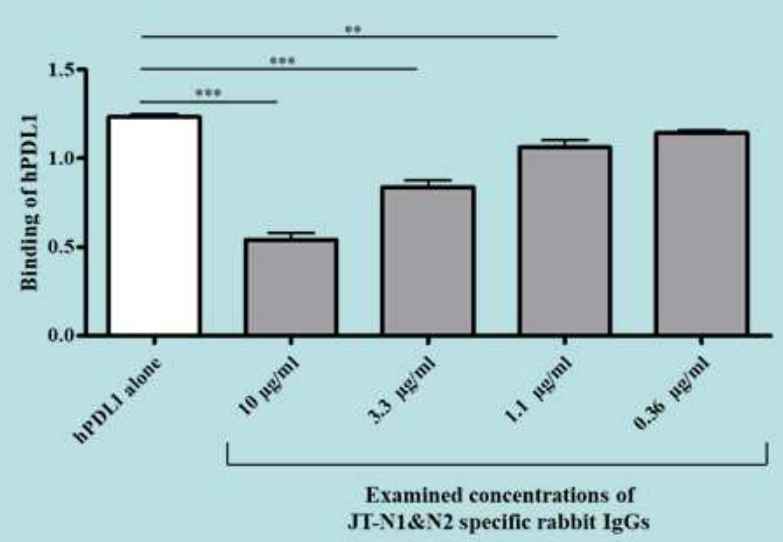
Binding of hPDL1
1.5
1.0
0.5
0.0
**

hPDL1 alone
10 µg/ml
3.3 µg/ml
1.1 µg/ml
0.36 µg/ml
Examined concentrations of JT-N1&N2 specific rabbit IgGs

Mimotope of anti-mouse PD1 mAb
Proof of principle for evaluation of anti-tumor activity *in vivo*, in syngeneic mouse model with tumors expressing human Her-2/neu

Passive administration of mimotope-specific IgG induces reduction of tumor progression

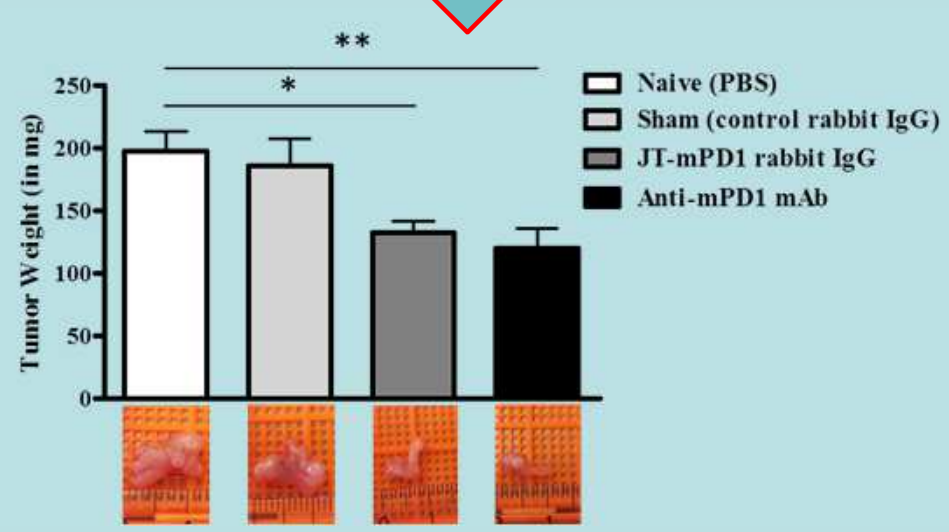
Tumor Weight (in mg)
250
200
150
100
50
0
**
*
Naive (PBS)
Sham (control rabbit IgG)
JT-mPD1 rabbit IgG
Anti-mPD1 mAb

Active immunization with the mimotope induces reduction of tumor progression and increased level of PD1-expressing immune cells in the tumors

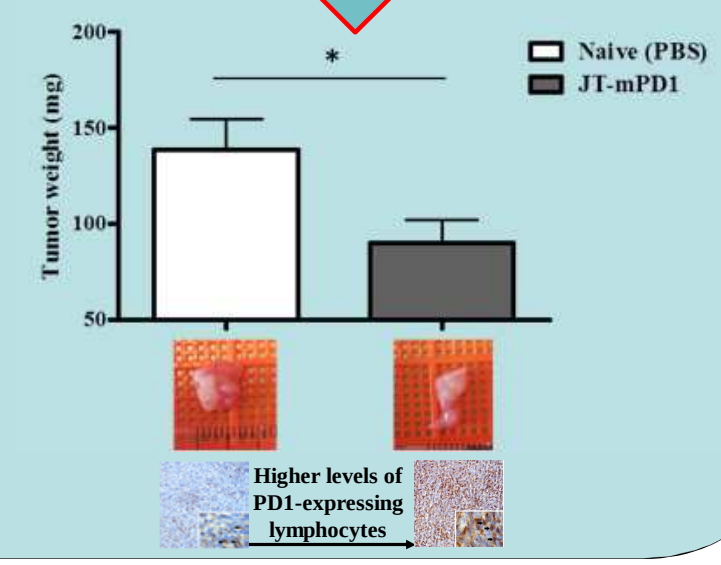
Tumor weight (mg)
200
150
100
50
*
Naive (PBS)
JT-mPD1
Higher levels of PD1-expressing lymphocytes

Summary and conclusion

By applying a syngeneic mouse model with tumors expressing human Her-2/neu, we show:

1. Anti-tumor activity by passive administration of specific IgGs against ICI mimotope, with comparable anti-tumor effect as the corresponding ICI

2. Anti-tumor activity by active immunization with mimotope of ICI

Our results may pave the way for a paradigm change for immunotherapy by active immunization with PD1 or other ICI mimotope-based vaccines against cancer, including Her-2 positive cancers. Furthermore, ICI mimotopes may also serve as adjuvants for cancer peptide-based vaccines, including Her-2/neu, thereby increasing the anti-tumor activity of the cancer vaccine.

[1] Medical University of Vienna, Austria, [2] University of Gothenburg, Sweden, [3] University of Veterinary Medicine, Vienna, Autsria [4] Vienna Cancer Center, Austria, * Contact: **joshua.tobias@meduniwien.ac.at , ursula.wiedermann@meduniwien.ac.at**