IMUGENE
Developing Cancer Immunotherapies

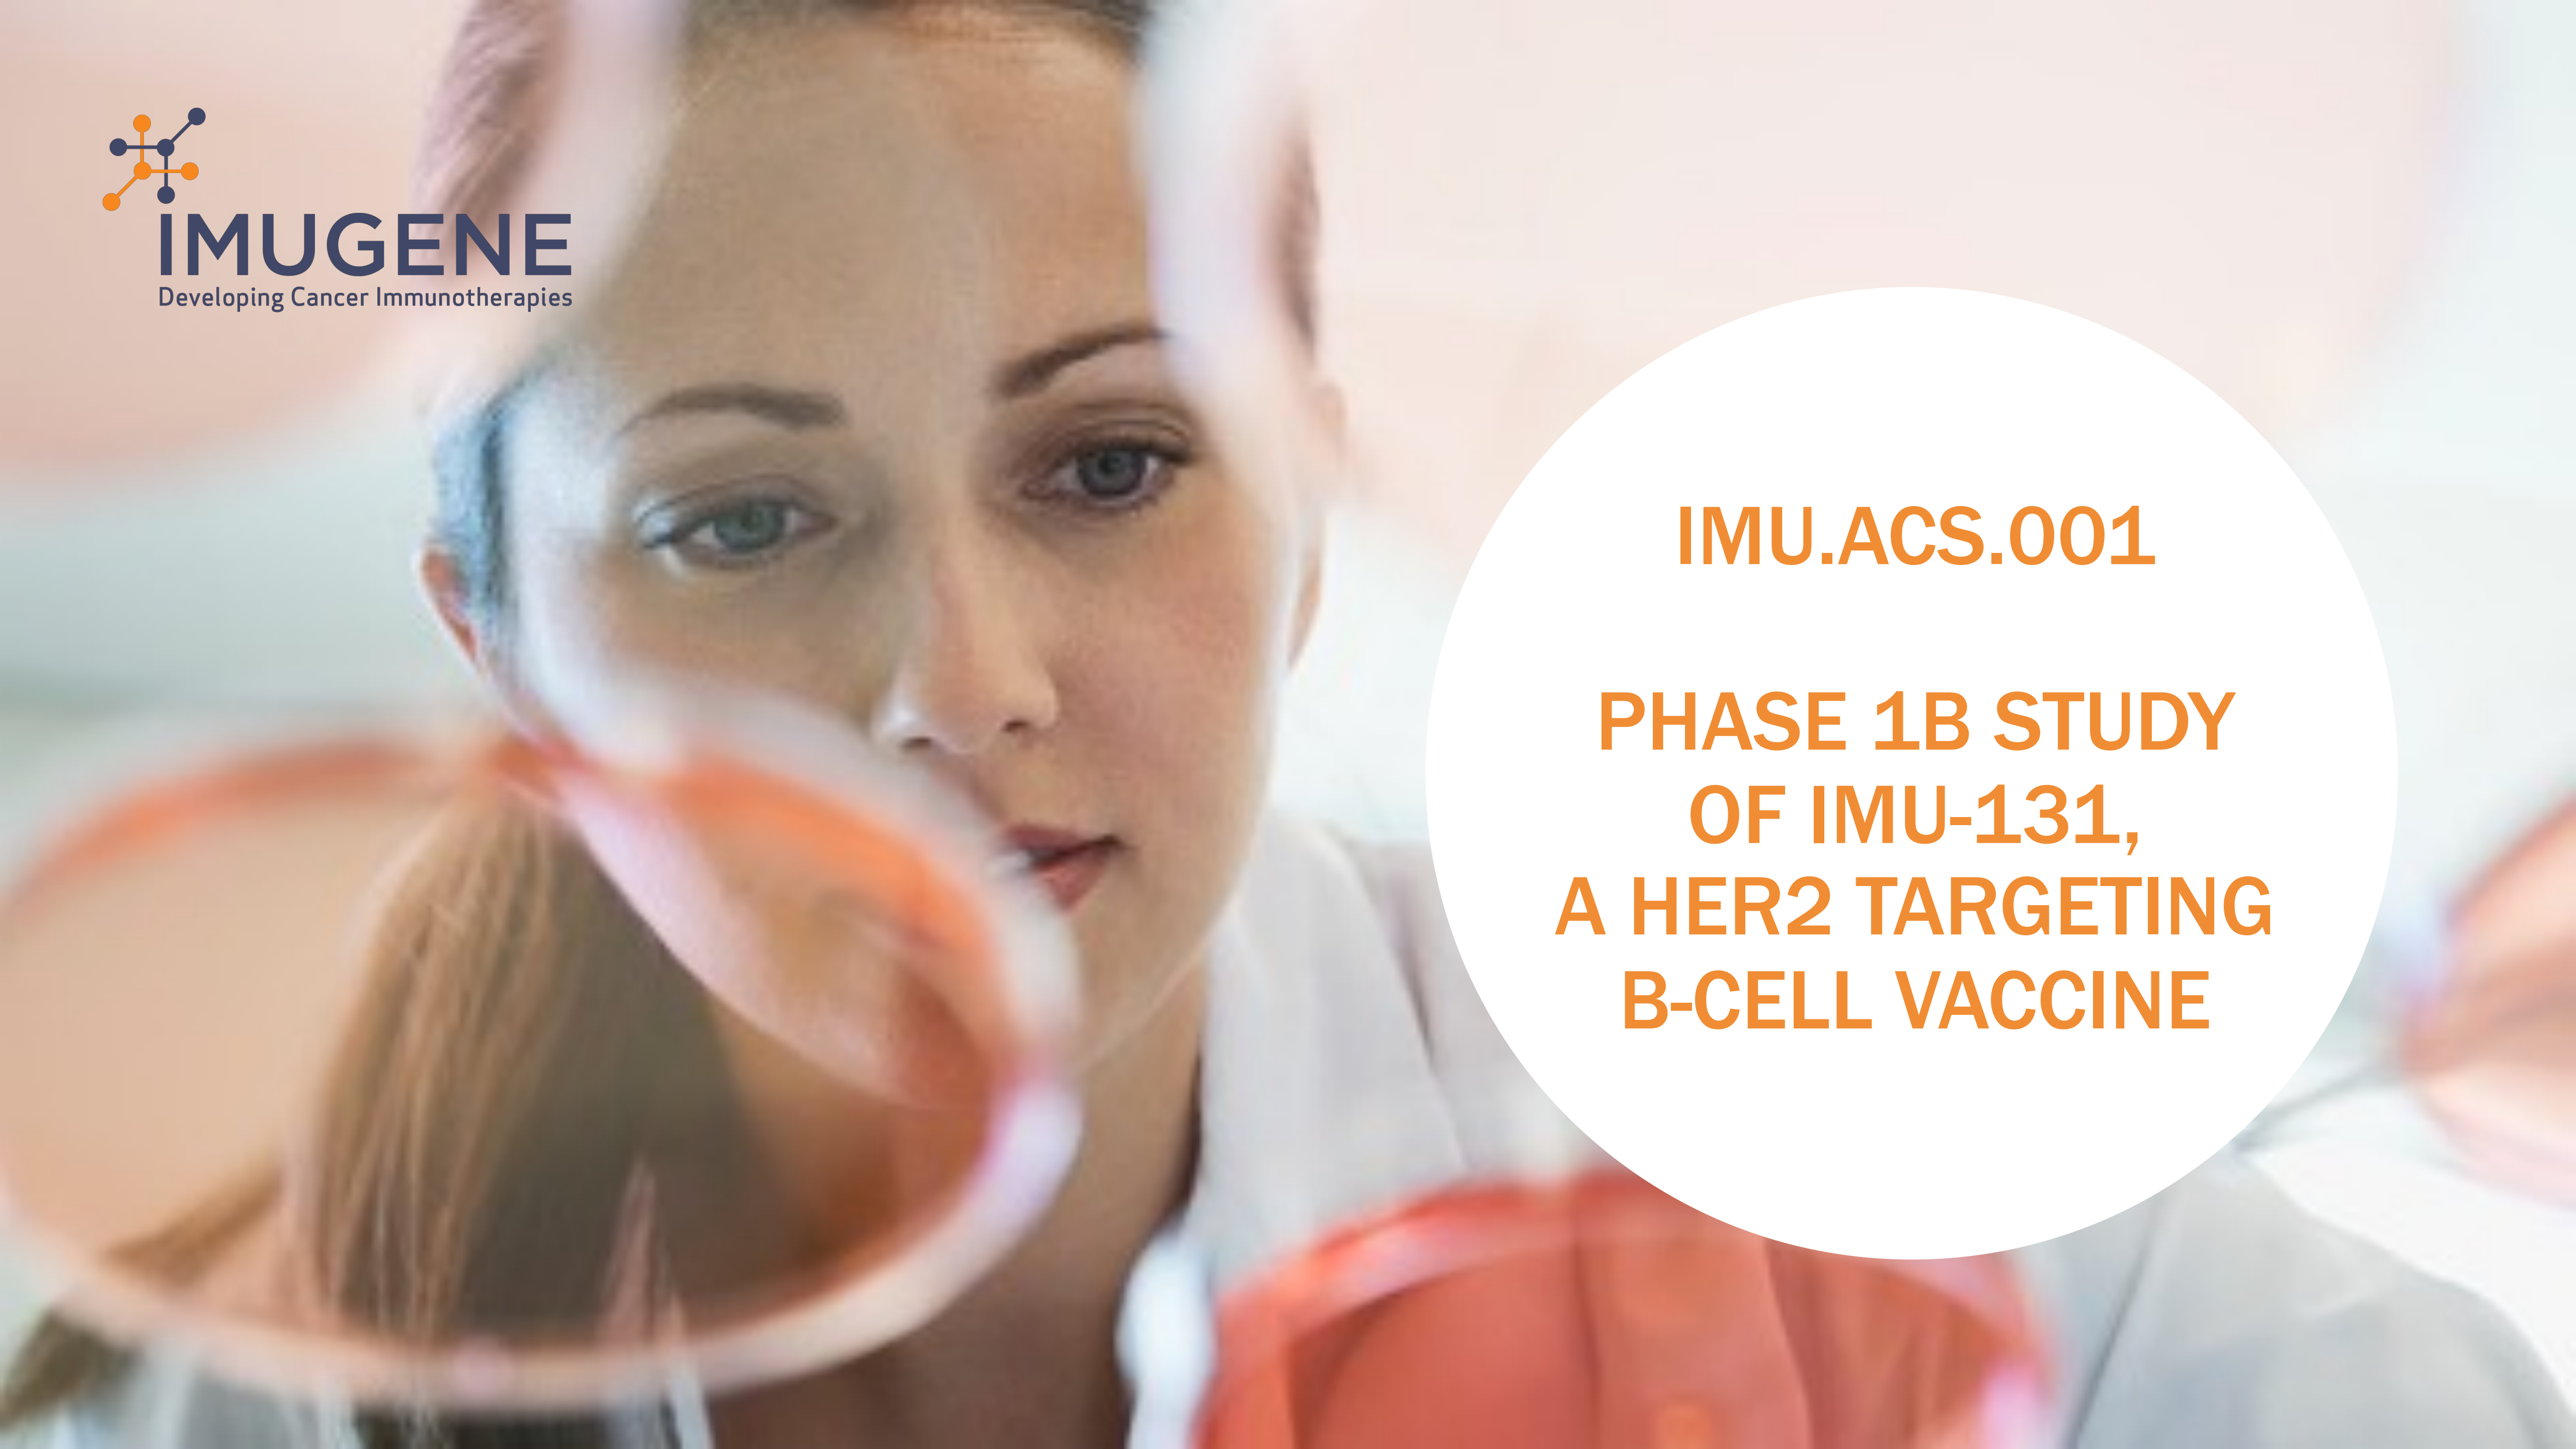

IMU.ACS.001

PHASE 1B STUDY OF IMU-131, A HER2 TARGETING B-CELL VACCINE

MEDICAL UNIVERSITY
OF VIENNA
2

IMU-131 IS A THERAPEUTIC B-CELL PEPTIDE VACCINE COMPOSED OF 3 EPITOPES FROM THE EXTRACELLULAR DOMAIN OF HER2/NEU CONJUGATED TO CRM197, EMULSIFIED WITH MONTANIDE

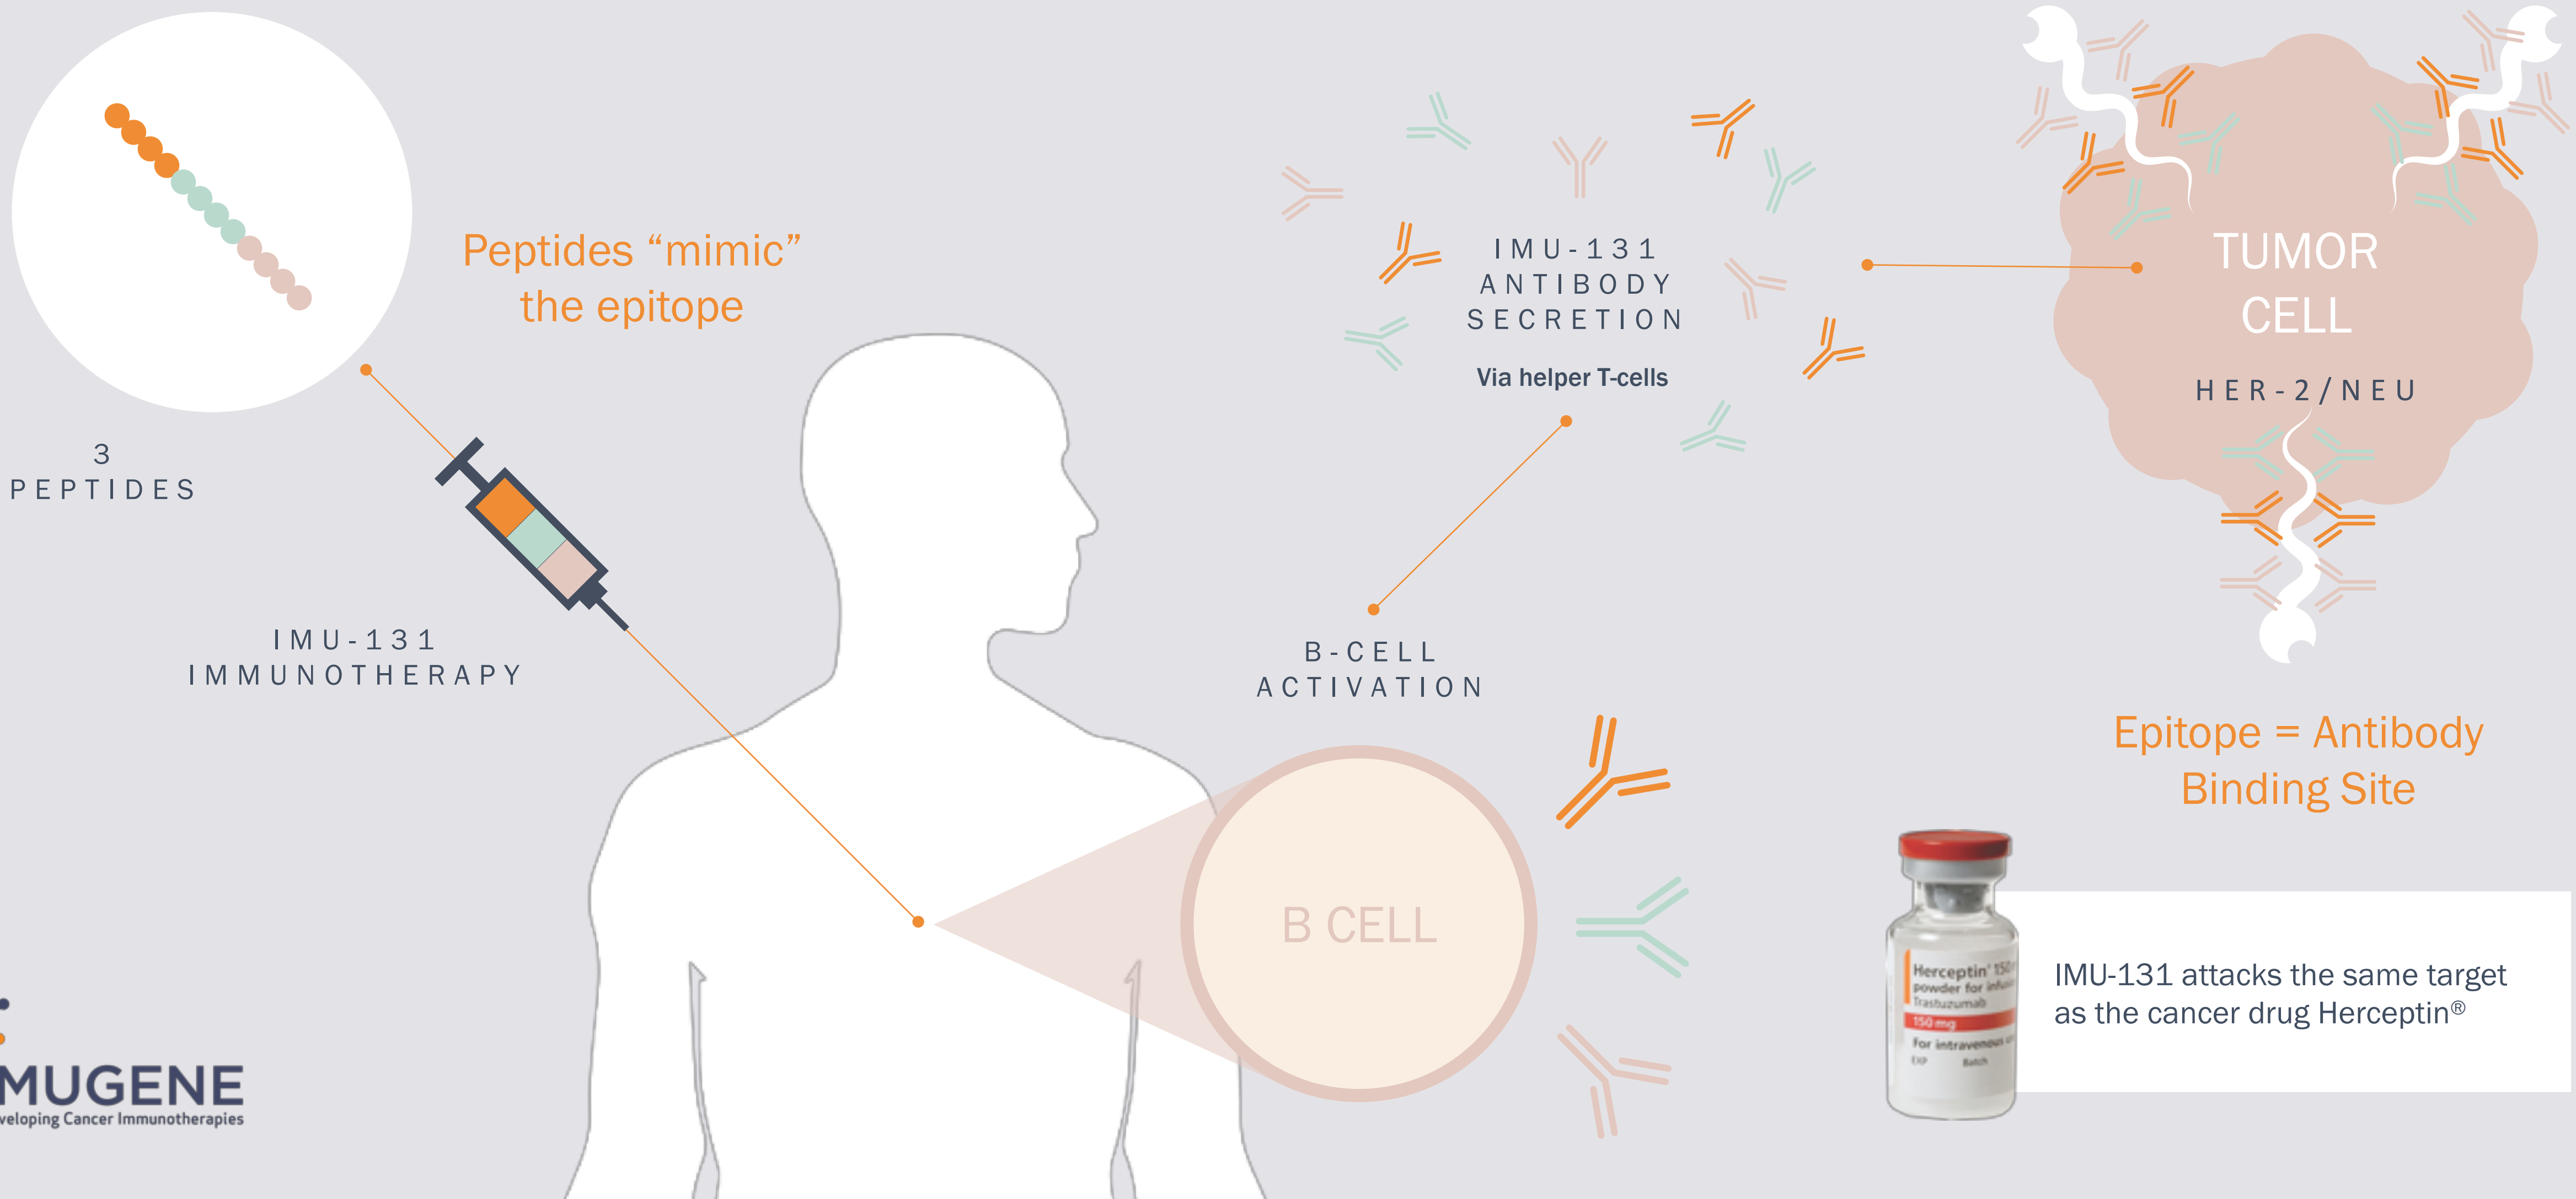
Peptides "mimic" the epitope
3 PEPTIDES
IMU-131 IMMUNOTHERAPY
IMU-131 ANTIBODY SECRETION
Via helper T-cells
B-CELL ACTIVATION
B CELL
TUMOR CELL
HER-2/NEU
Epitope = Antibody Binding Site
Herceptin
Trastuzumab
150 mg
IMU-131 attacks the same target as the cancer drug Herceptin®

IMUGENE
Developing Cancer Immunotherapies

HARNESSING B-CELLS FOR CANCER IMMUNOTHERAPY 3

IMU.ACS.001 PHASE 1B: DESIGN & RESULTS

Trial

- HER2 Gastric or GEJ cancer
- Phase 1b
- Open label
- Dose escalation
- 14 sites in Asia and Eastern Europe

Patients

- Advanced stage IIIb or IV
- 7 HER2+++, 3 HER2++ (FISH positive), 4 HER2++ expressing tumors
- Age 57yo (21 - 79)
- ECOG 1(7) and 0(7)
- 9 Asian, 5 Caucasian
- 5 female, 9 male

Study

- 14 patients in 3 cohorts (10µg (3), 30µg (6) and 50µg (5))
- Dosed on D0, D14, D35
- IMU-131 in combination with chemo: cisplatin and 5FU or capecitabine

Endpoints

- Recommended Phase 2 Dose of IMU-131
- Safety and Toxicity
- Immunogenicity (anti-peptide (P467) and anti-HER-2 antibody titres)

Study Results

- No safety or toxicity issues
- All patients had increased antibody response
- 1 Complete Response
- 5 Partial Response
- 4 Stable Disease
- 1 Progressive Disease
- 50 µg selected as Ph2 dose

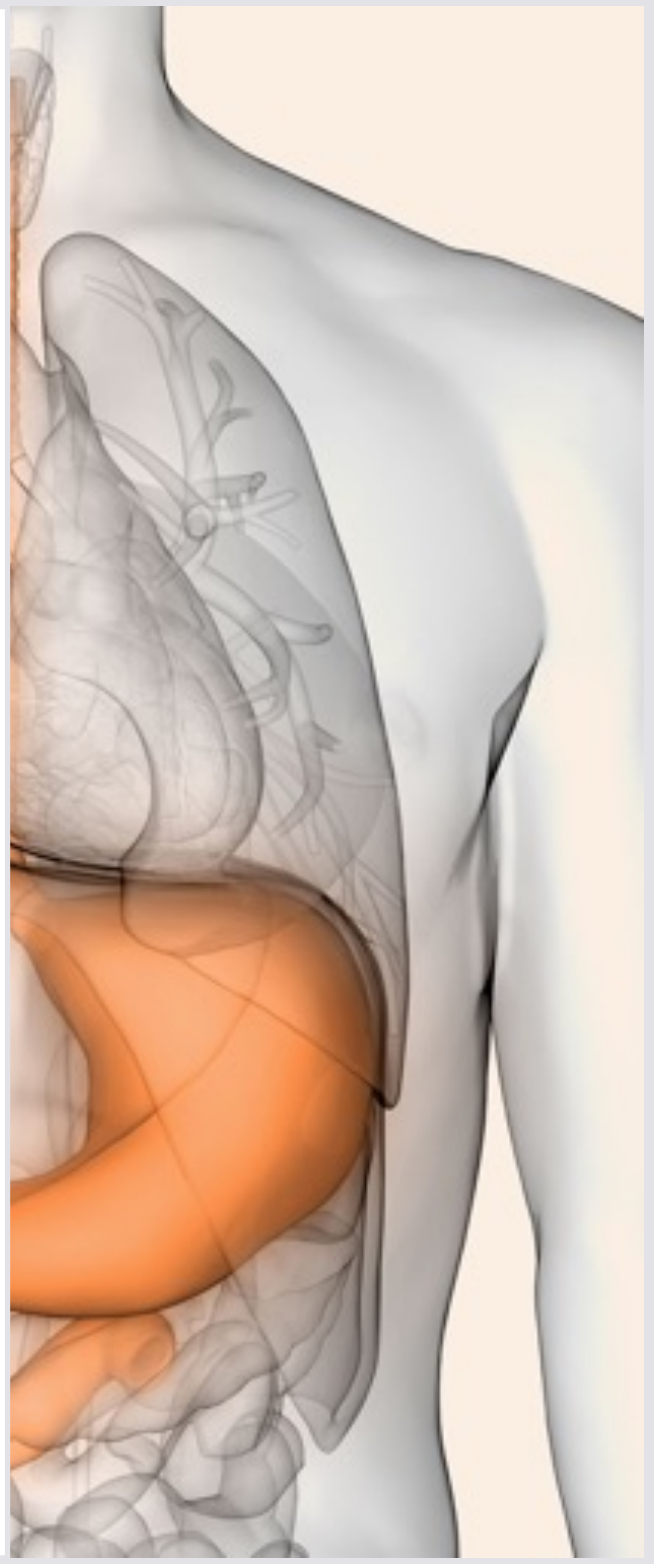

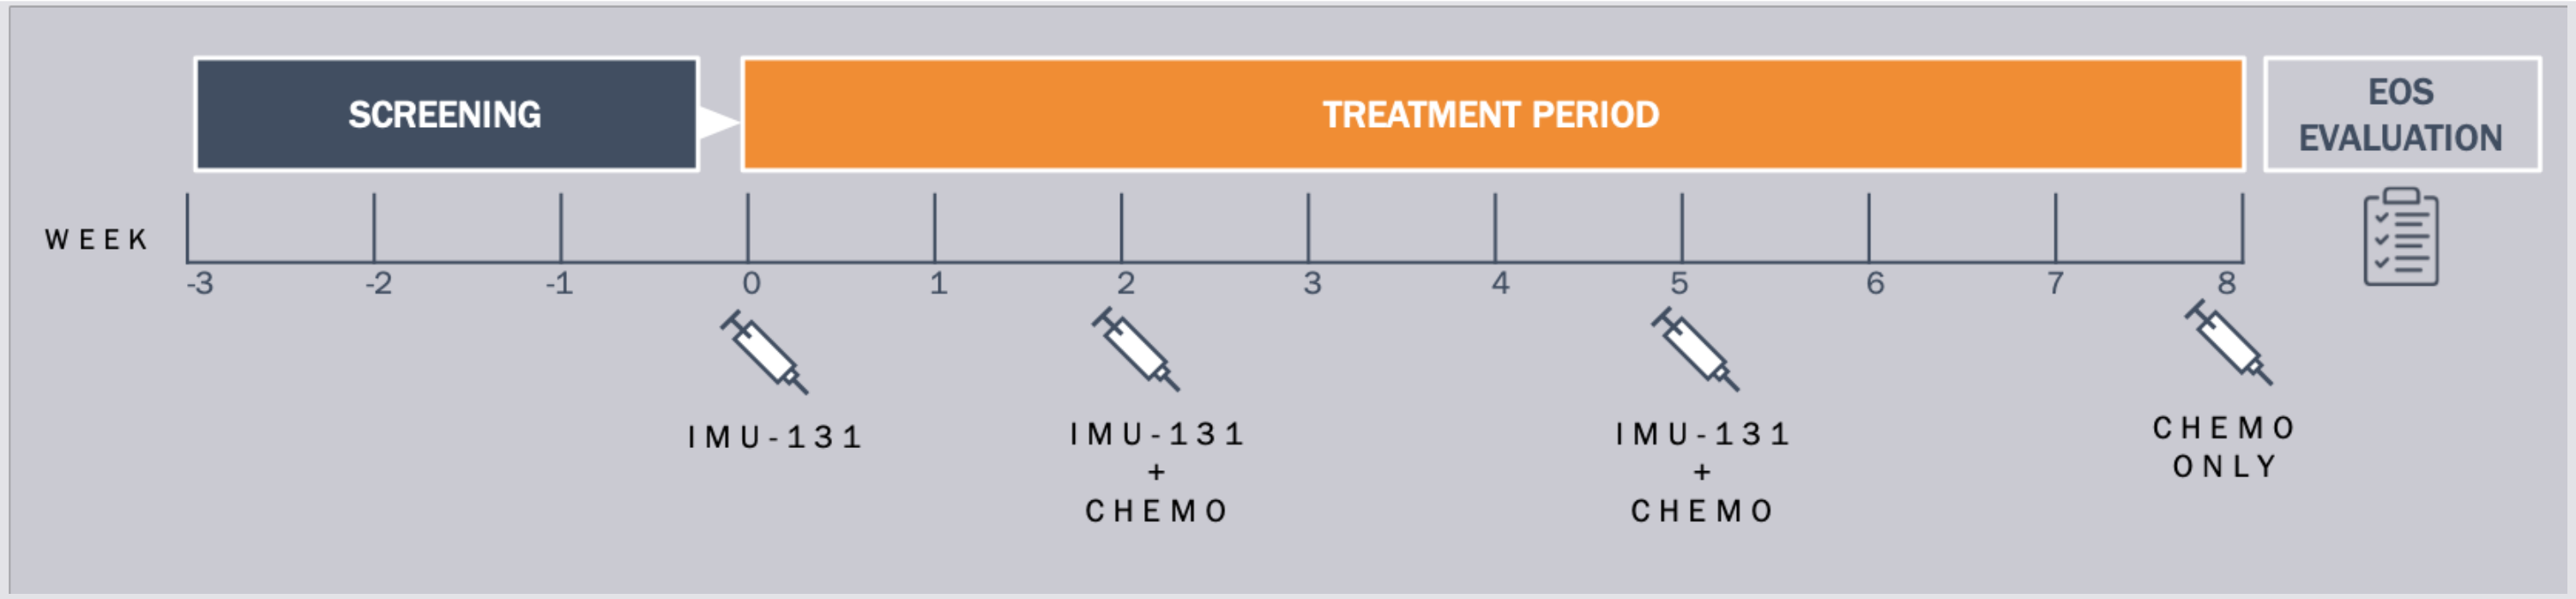
SCREENING
TREATMENT PERIOD
EOS
EVALUATION
WEEK
-3
-2
-1
0
1
2
3
4
5
6
7
8
IMU-131
IMU-131
+
CHEMO
IMU-131
+
CHEMO
CHEMO
ONLY

HARNESSINGB-CELLS FOR CANCER IMMUNOTHERAPY 4

IMU.ACS.001 PHASE 1B: SAFETY & ANTIBODIES

- 10 patients had 18 SAEs
- No SAEs related to IMU-131
- 1 mild injection site reaction
- No significant LVEF changes

- The 50 µg dose produced the most consistent anti-peptide (P467) and anti-HER-2 specific antibodies compared to 10 and 30 µg doses

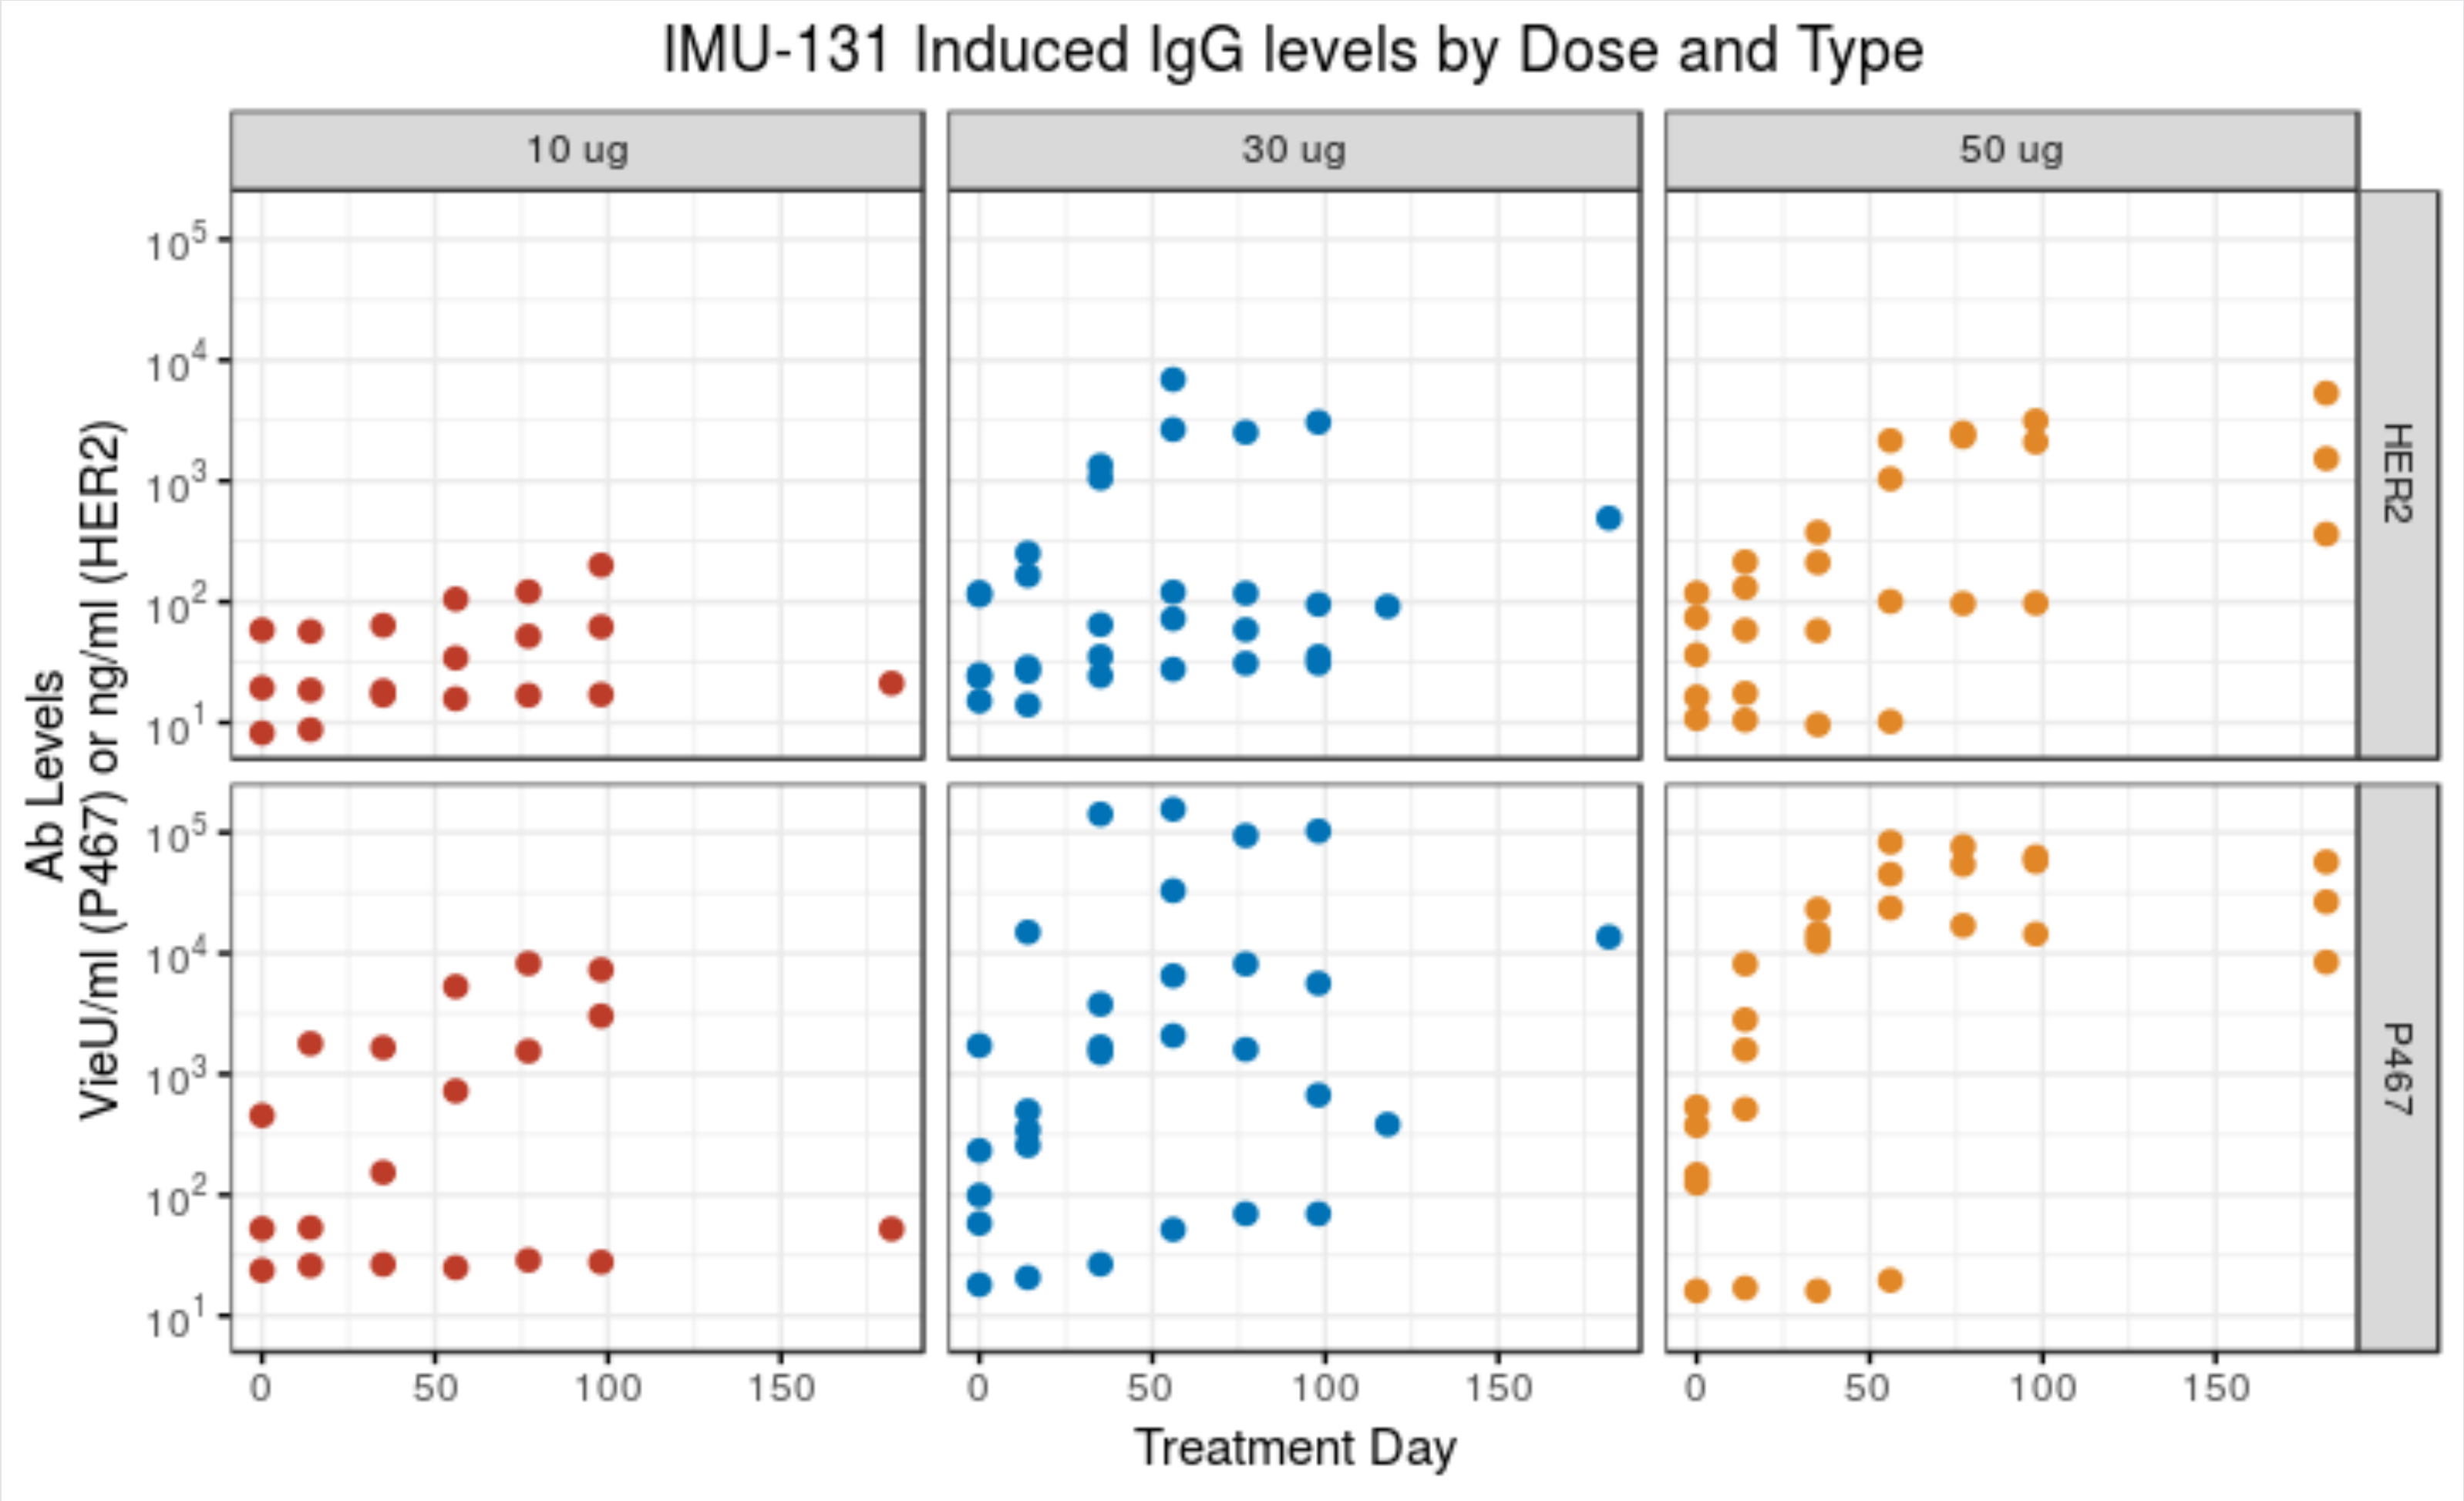
IMU-131 Induced IgG levels by Dose and Type
10 ug
30 ug
50 ug
HER2
P467
Ab Levels
VieU/ml (P467) or ng/ml (HER2)
10^1
10^2
10^3
10^4
10^5
0
50
100
150
Treatment Day

IMUGENE
Developing Cancer Immunotherapies

4

HARNESSING B-CELLS FOR CANCER IMMUNOTHERAPY 5

IMU.ASC.001 PHASE 1B: CLINICAL RESPONSE & CONCLUSIONS

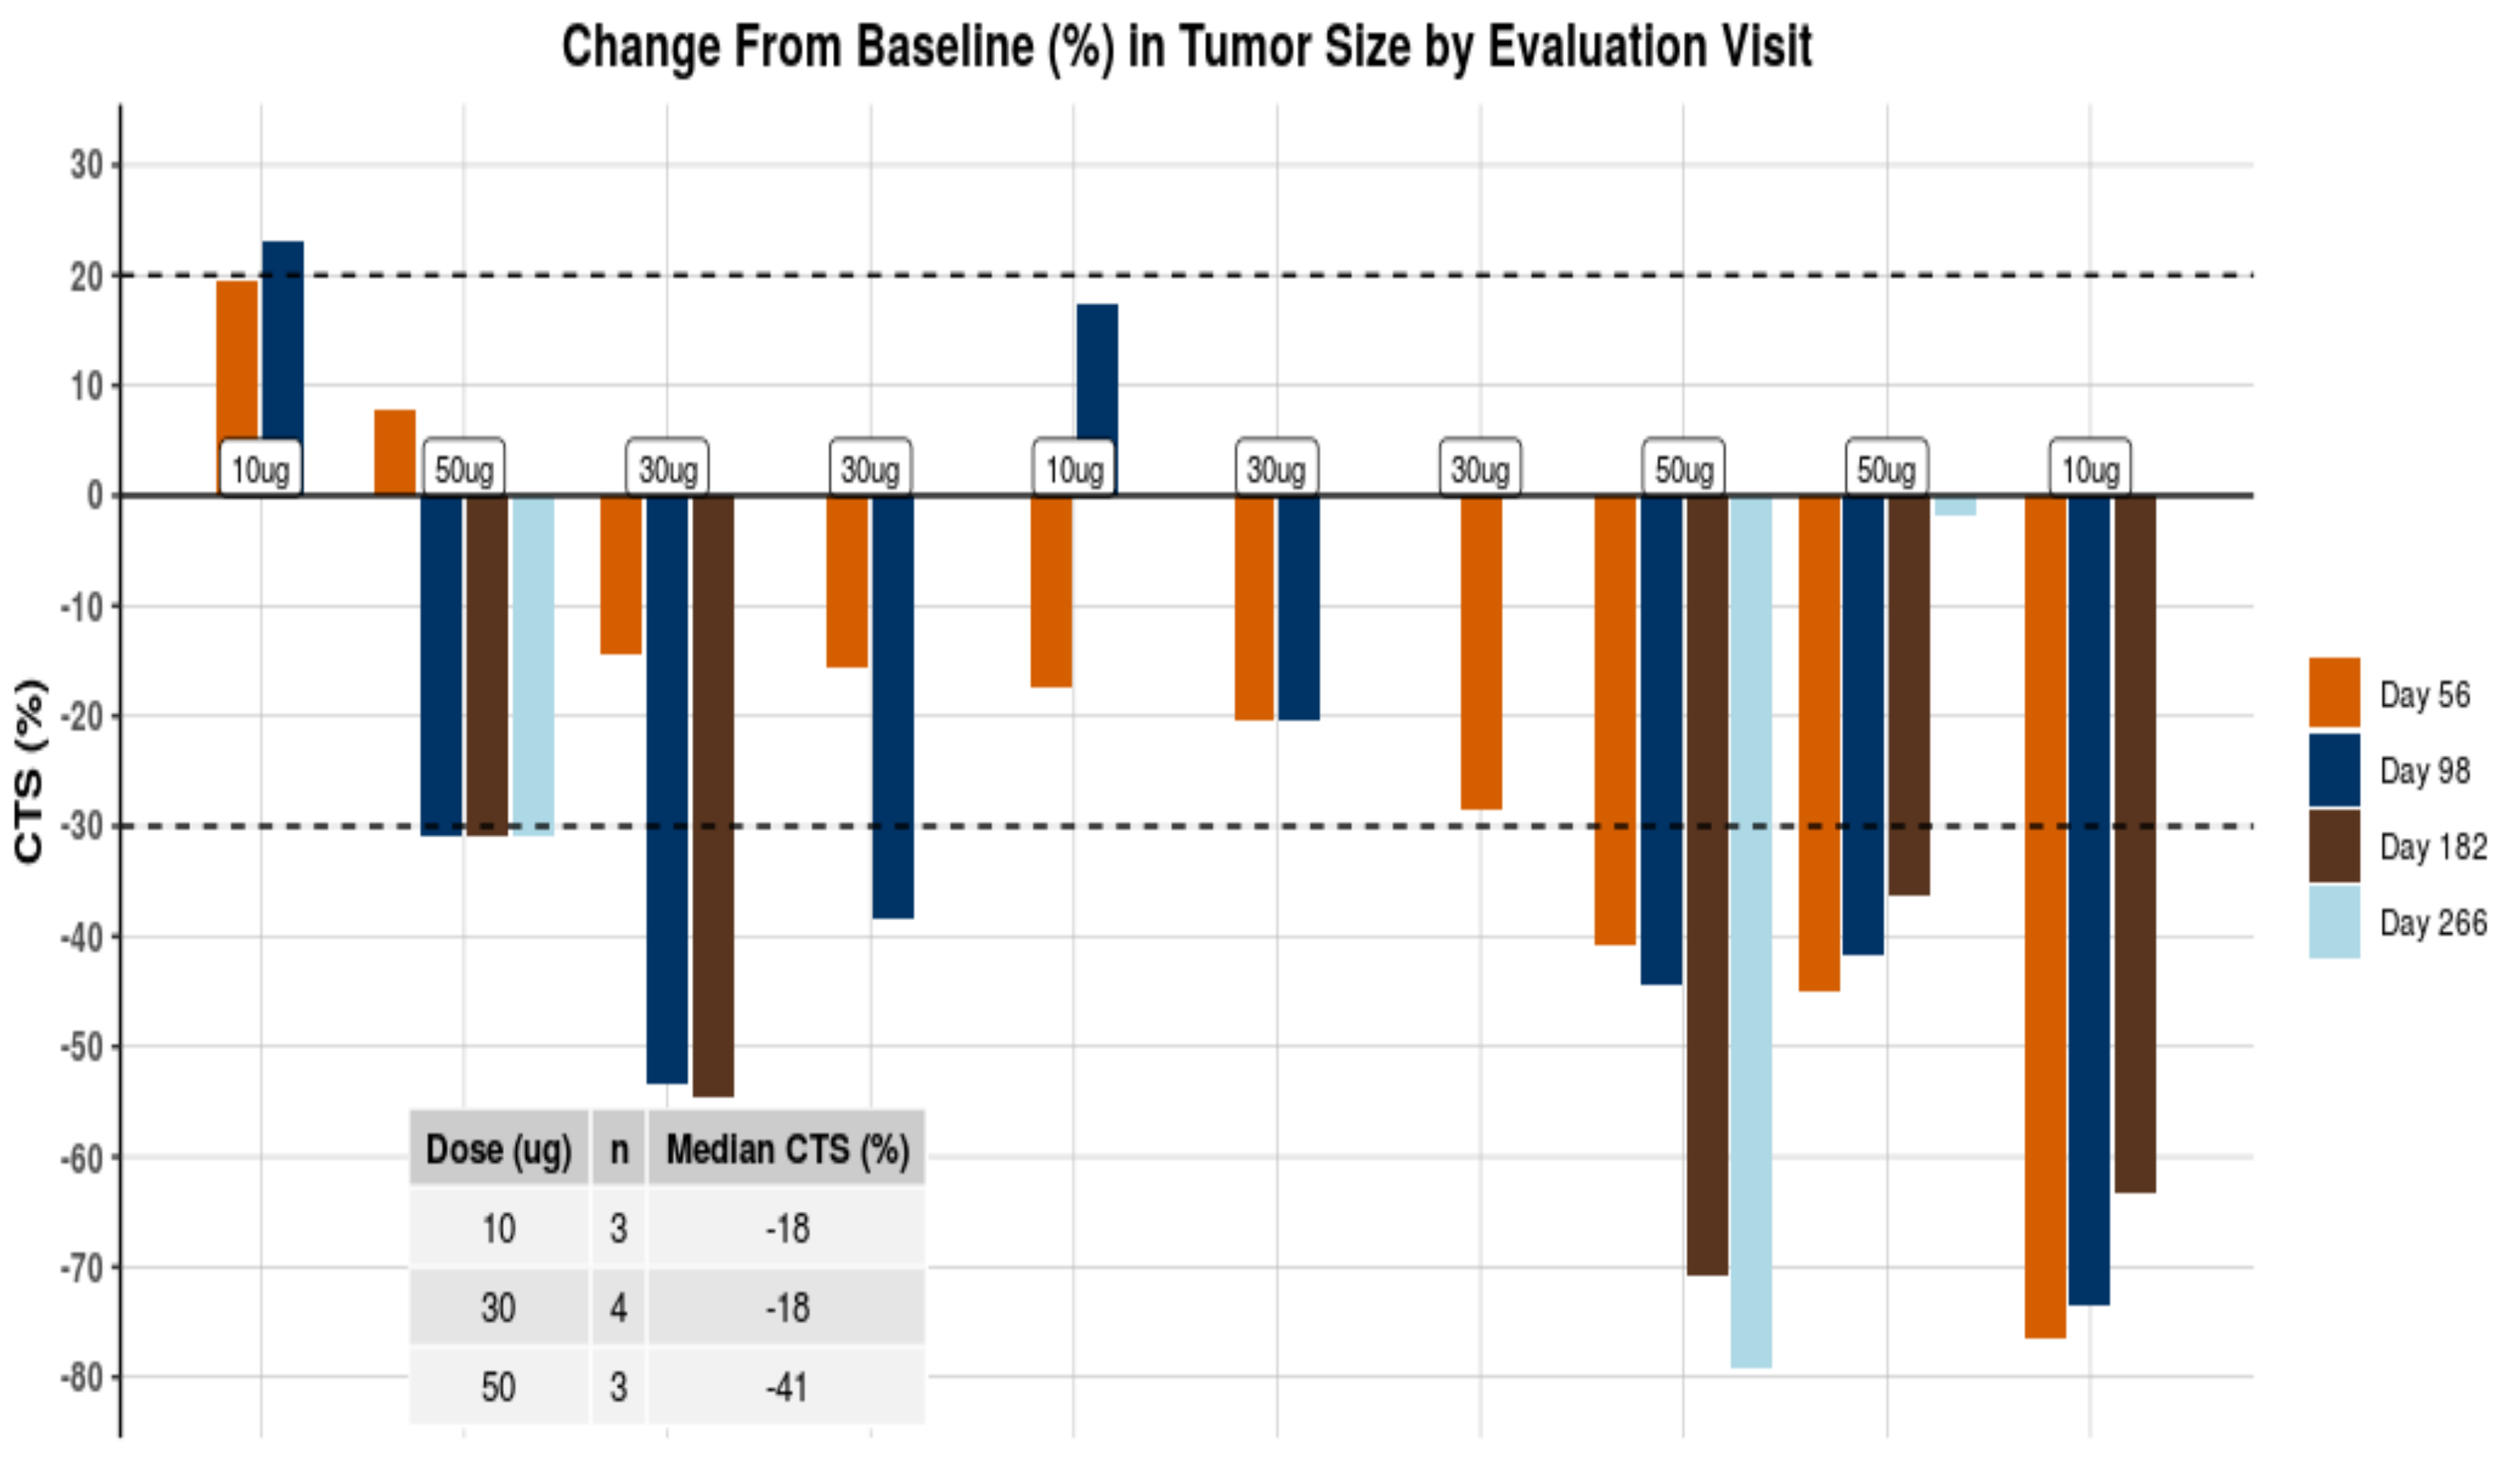
Change From Baseline (%) in Tumor Size by Evaluation Visit
CTS (%)
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
10ug
50ug
30ug
30ug
10ug
30ug
30ug
50ug
50ug
10ug
Day 56
Day 98
Day 182
Day 266

Dose (ug)	n	Median CTS (%)
10	3	-18
30	4	-18
50	3	-41

- The preliminary immunology and clinical response data are promising.
- Safety data indicate that IMU-131 is well-tolerated with no significant local or systemic reactions.
- There were no dose-limiting toxicities observed, no significant injection site reactions and no IMU-131 related SAEs.
- Preliminary response data demonstrates 50 μg of IMU-131 was associated with tumor size reduction.
- The 50 μg dose of IMU-131 is being used in a phase 2 study.

IMUGENE
Developing Cancer Immunotherapies